Discovery and Preclinical Characterization of RGLS4326 for the Treatment of Autosomal Dominant Polycystic Kidney Disease (ADPKD)

Edmund C. Lee[1, *], Tania Valencia[1], Annelie Schairer[1], Kara Kersjes[1], Jian Li[1], Andrea Flaten[2], Vishal Patel[2]

[1]Regulus Therapeutics Inc., San Diego, California. *Presenting Author

[2]Department of Internal Medicine and Division of Nephrology, University of Texas Southwestern Medical Center, Dallas, Texas.

Abstract

Background

Autosomal dominant polycystic kidney disease (ADPKD), caused by mutations in the *PKD1* or *PKD2* genes, is among the most common human monogenetic disorders and a leading genetic cause of end-stage renal disease (ESRD). MicroRNAs (miRs) are non-coding RNAs that play central roles in cell differentiation, proliferation and survival by binding to complementary target mRNAs, resulting in repression of translation and eventual degradation of the targeted mRNAs. We have previously demonstrated that miR-17 play important roles in ADPKD, and that miR-17 is a promising drug target for the treatment of the disease.

Methods:

RGLS4326 was discovered through screening a chemically-diverse library of >100 oligonucleotides for their ability to inhibit miR-17 in a miR-17 luciferase sensor assay. RGLS4326 was extensively profiled in multiple safety assays, including biochemical, *ex-vivo* tissues slices and *in vivo* studies. Preclinical efficacy of RGLS4326 was studied in both *Pkd2*KO and *Pcy* mouse models of PKD

Results:

In preclinical studies, RGLS4326 potently inhibited miR-17 activity, displaced miR-17 from the translationally active high molecular weight polysomes, and de-repressed multiple miR-17 target genes in different mouse kidney cell lines. RGLS4326 shows favorable pharmacokinetic profiles in both normal and PKD mouse models, where preferential distribution to kidney compared to other tissues was evident. Most importantly, we have demonstrated that RGLS4326 confers efficacy in two mouse models of PKD following subcutaneous administrations.

Conclusion:

Our preclinical data support the clinical development of RGLS4326 for the treatment of ADPKD.

Characteristics of Autosomal Dominant Polycystic Kidney Disease (ADPKD)

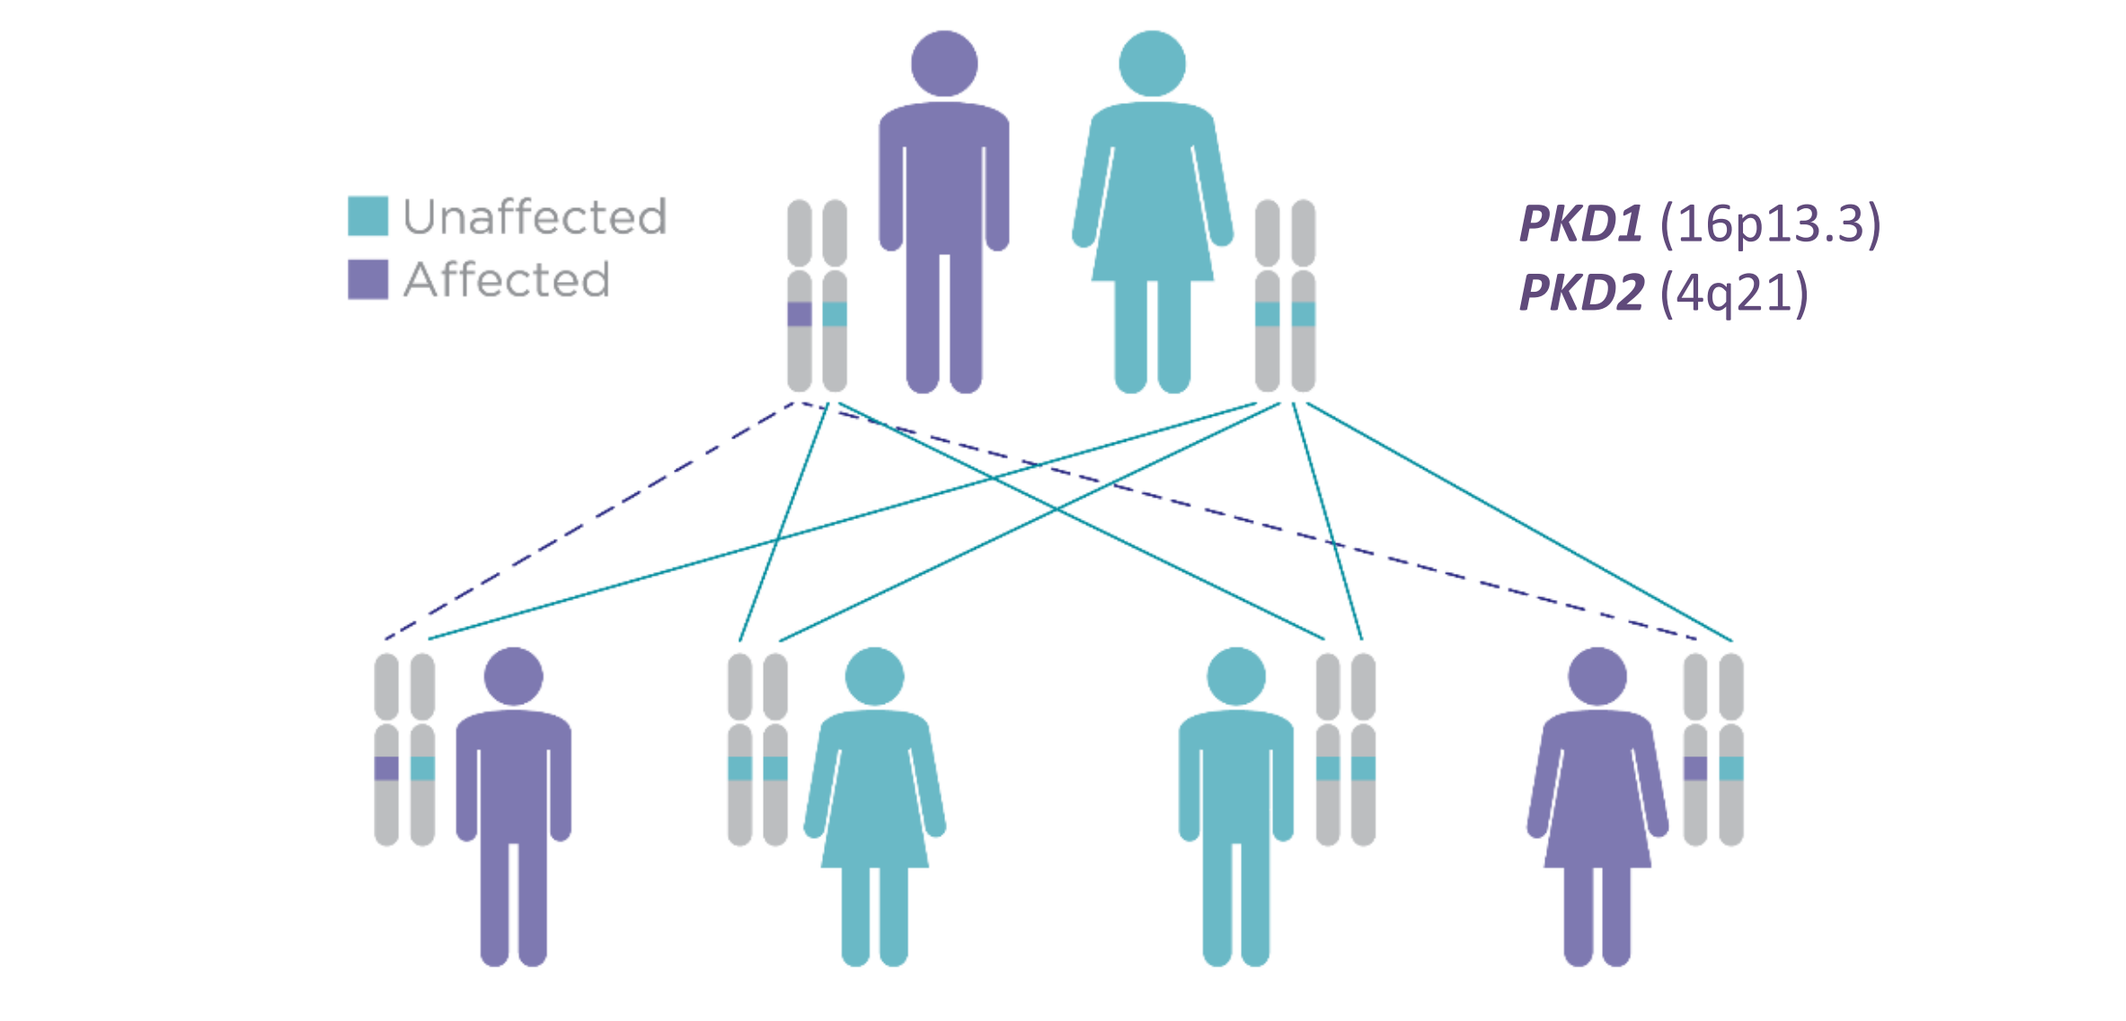
Unaffected
Affected
PKD1 (16p13.3)
PKD2 (4q21)

ADPKD is a monogenetic disorder caused by mutations in either PKD1 (85%) or PKD2 genes (15%), which encode the proteins polycystin-1 (PC1) and polycystin-2 (PC2), respectively.

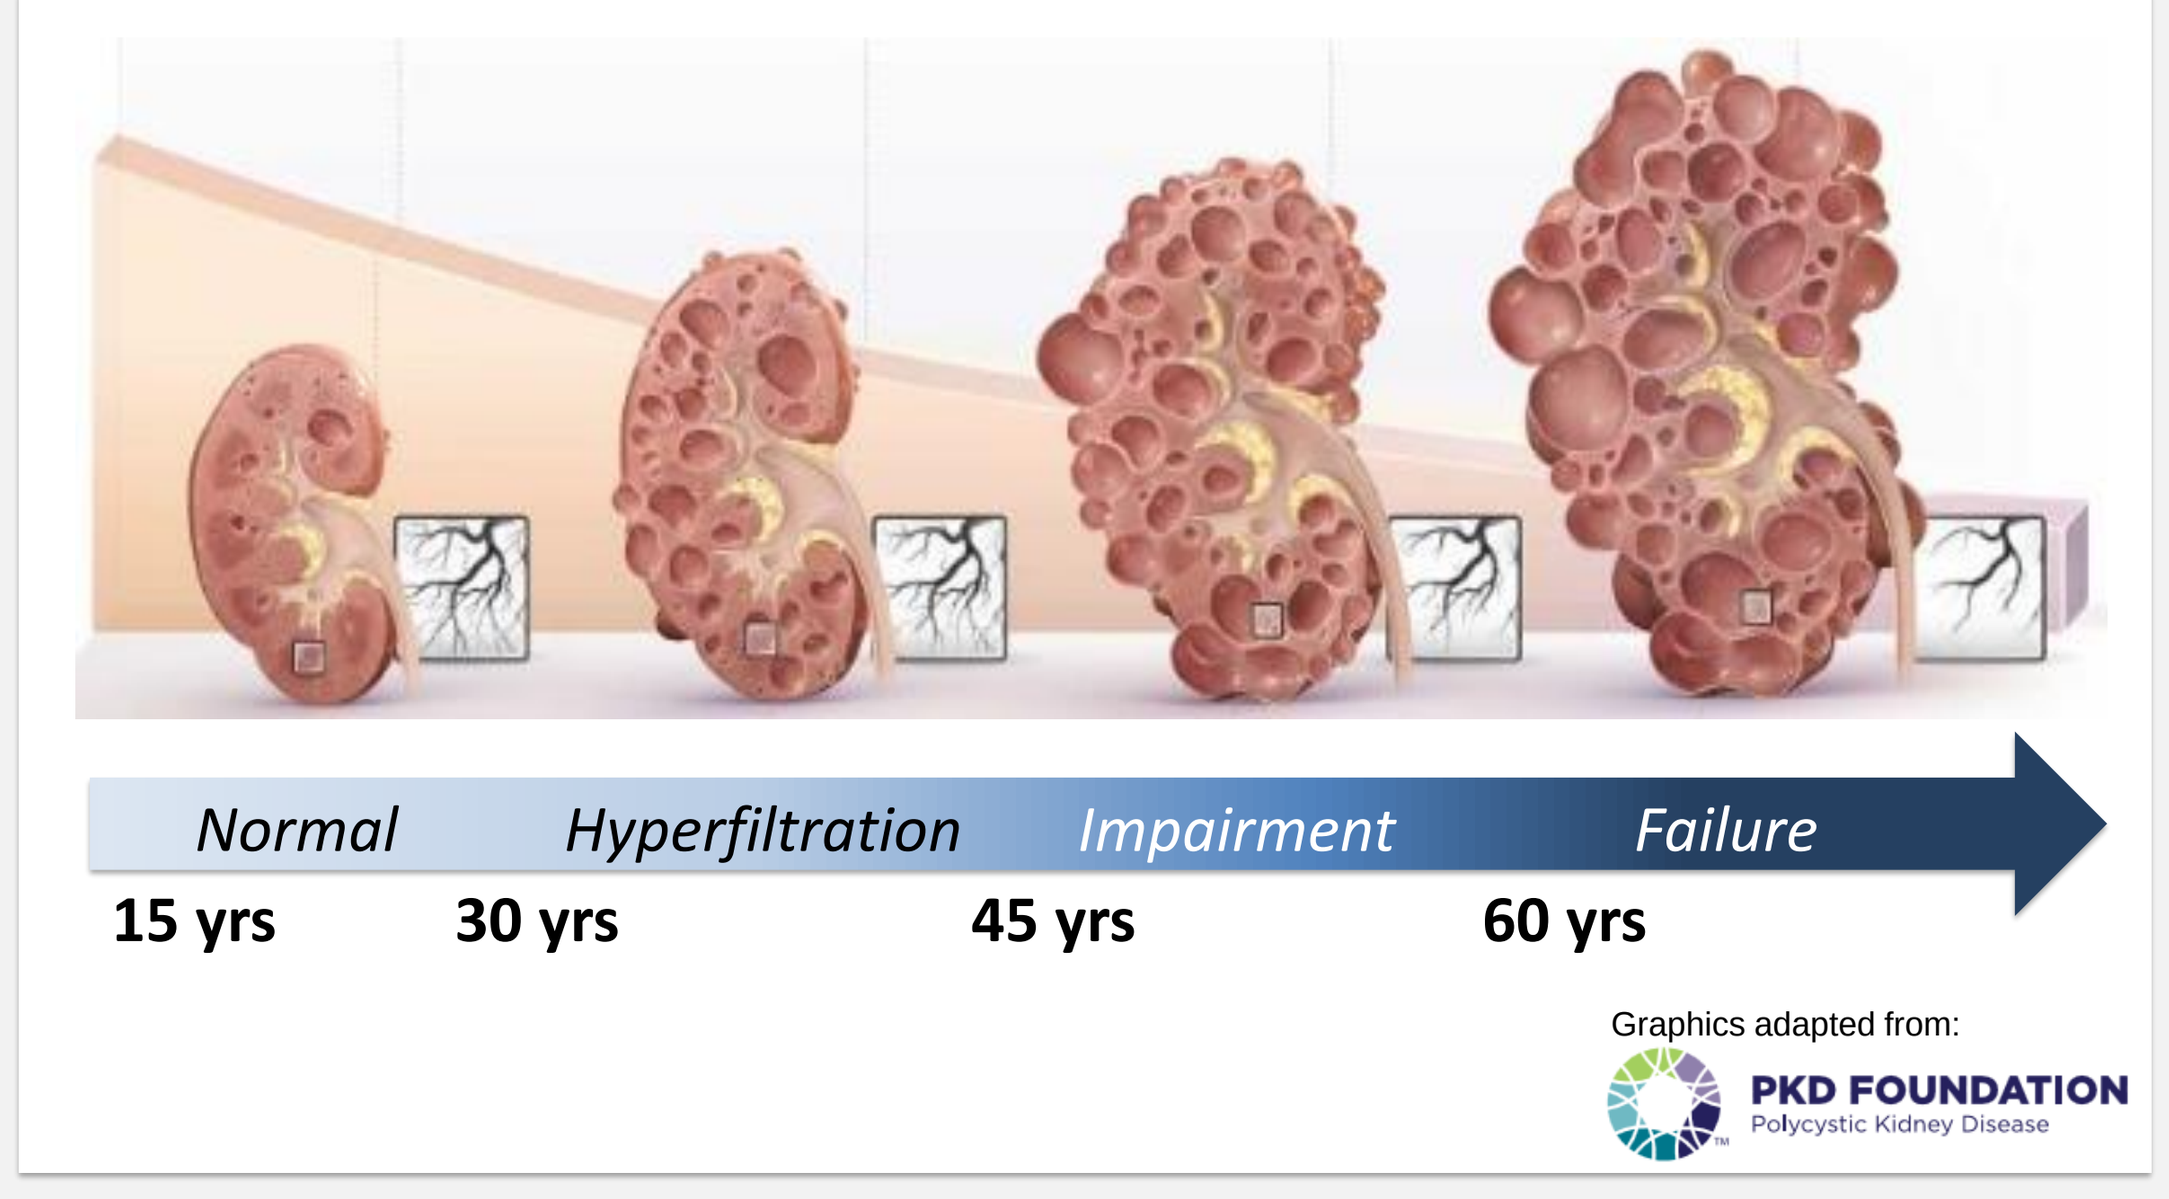
Normal
Hyperfiltration
Impairment
Failure
15 yrs
30 yrs
45 yrs
60 yrs
Graphics adapted from:
PKD FOUNDATION
Polycystic Kidney Disease

Mutations disrupt normal functions of PC1 and PC2 in renal tubular epithelium, causing growth of multiple kidney cysts that displace and destroy normal kidney tissues, ultimately leading to fibrosis, derangement in renal architecture and kidney failure.

What are microRNAs?

- MicroRNAs are highly conserved, short non-coding RNAs (20-22 nucleotides) with unique seed sequence of ~8 nucleotides that bind to complementary target sequences located primary in the 3' untranslated region of targeted mRNAs.
- A single microRNA can bind to and repress translation of different mRNAs, eventually resulting in the degradation of the transcripts.
- **Aberrant microRNAs activity, such as miR-17, has been shown to be important in multiple human diseases, including ADPKD.**
- Anti-miRs are designed to inhibit microRNA function and de-repress their downstream target mRNAs and encoded proteins.

miR-17~92 cluster and its paralogous clusters miR-106a~363 and miR-106b~25.

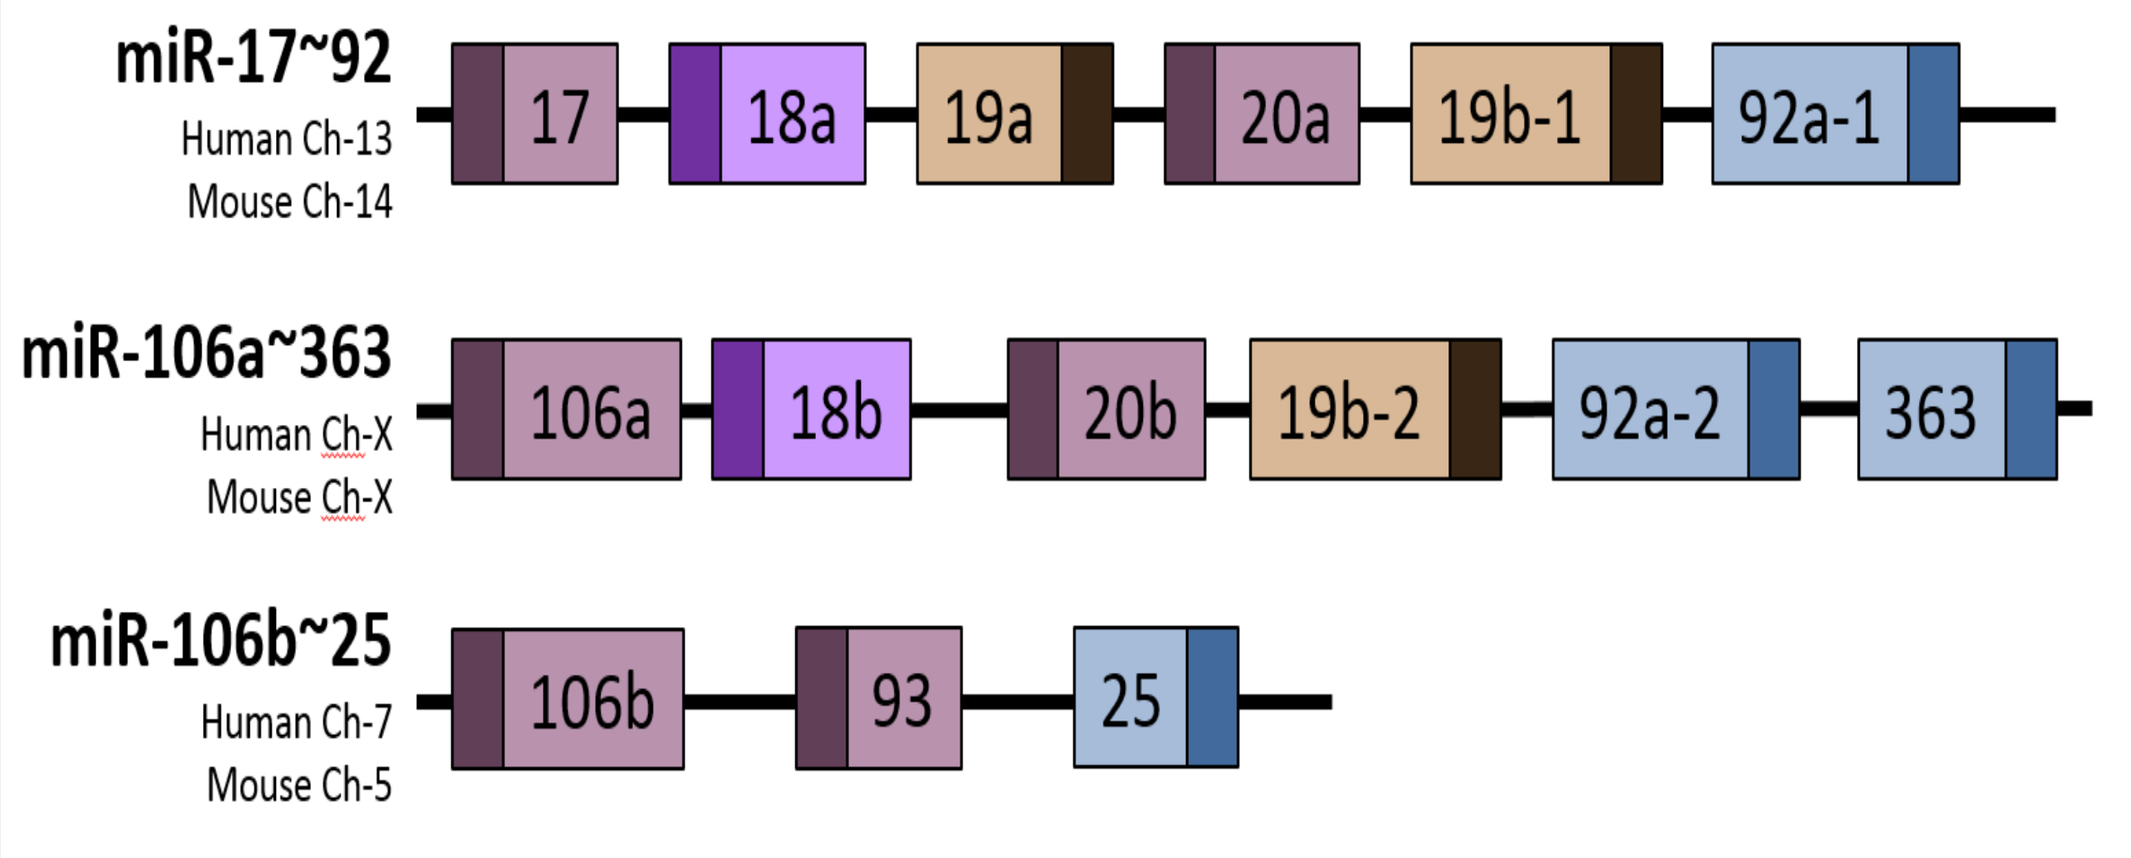
miR-17~92
Human Ch-13
Mouse Ch-14
17
18a
19a
20a
19b-1
92a-1
miR-106a~363
Human Ch-X
Mouse Ch-X
106a
18b
20b
19b-2
92a-2
363
miR-106b~25
Human Ch-7
Mouse Ch-5
106b
93
25

Functions of miR-17 in kidney

- Specific knockdown in mouse nephron progenitors impaired nephrogenesis and renal function.
- **Overexpression in renal tubules promoted kidney cyst growth**
- **Knockdown of miR-17~92 in multiple mouse models of ADPKD, including the *Pkd1*-KO, *Pkd1*$^{F/RC}$, Pkd2$^{RC/RC}$, *Kif3a*-KO and *Pkd2*-KO models, showed reduction of cyst growth.**
- Knockdown of miR-17~92 in renal tubules did not display any appreciable defects in kidney morphology and histology in normal mice.

RGLS4326 inhibits miR-17 family of microRNAs in luciferase assays *in vitro*

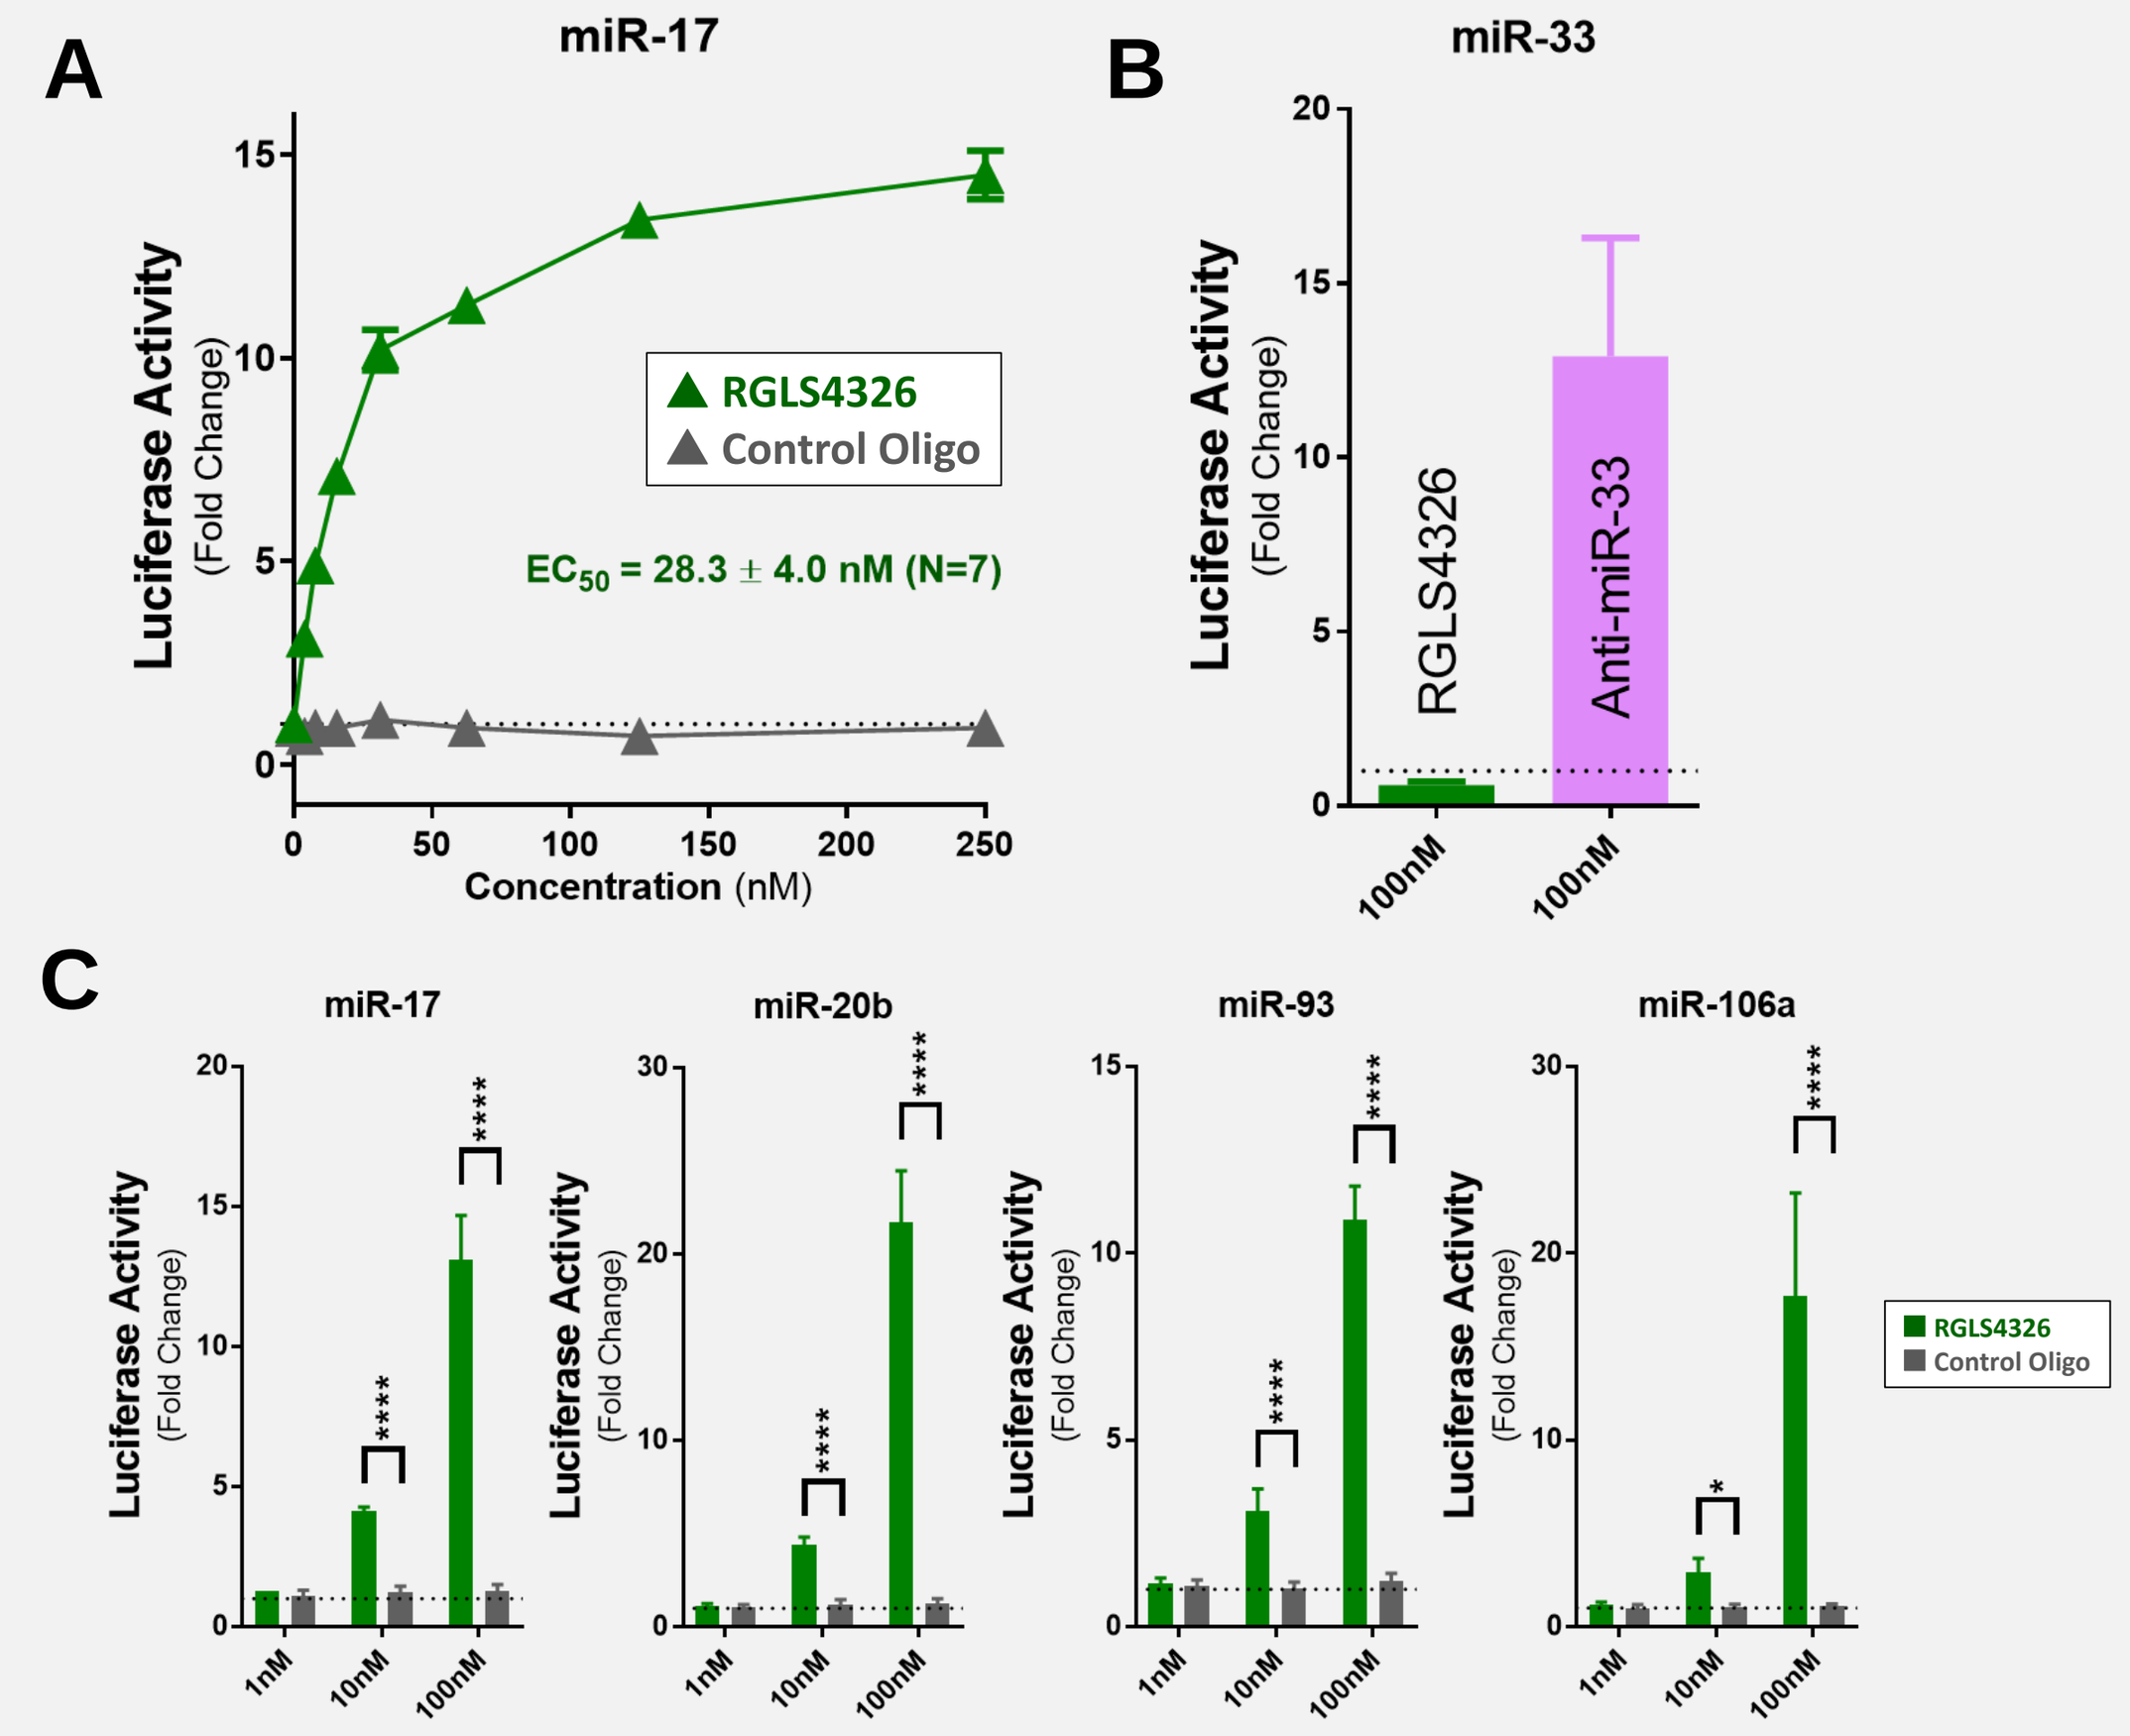
A
miR-17
Luciferase Activity (Fold Change)
RGLS4326
Control Oligo
EC50 = 28.3 ± 4.0 nM (N=7)
Concentration (nM)
B
miR-33
RGLS4326
Anti-miR-33
100nM
C
miR-17
miR-20b
miR-93
miR-106a
1nM
10nM
100nM

(A) Concentration-dependent de-repression of miR-17 luciferase activity by RGLS4326 in HeLa cells 24h following transfection.
(B) RGLS4326 has no activity against miR-33.
(C) As expected from its sequence complementarity to the miR-17 family seed region, RGLS4326 also inhibited miR-20a, miR-93 and miR-106b activity in corresponding luciferase assays.

RGLS4326 de-represses miR-17 downstream target genes, including *Pkd1* and *Pkd2*

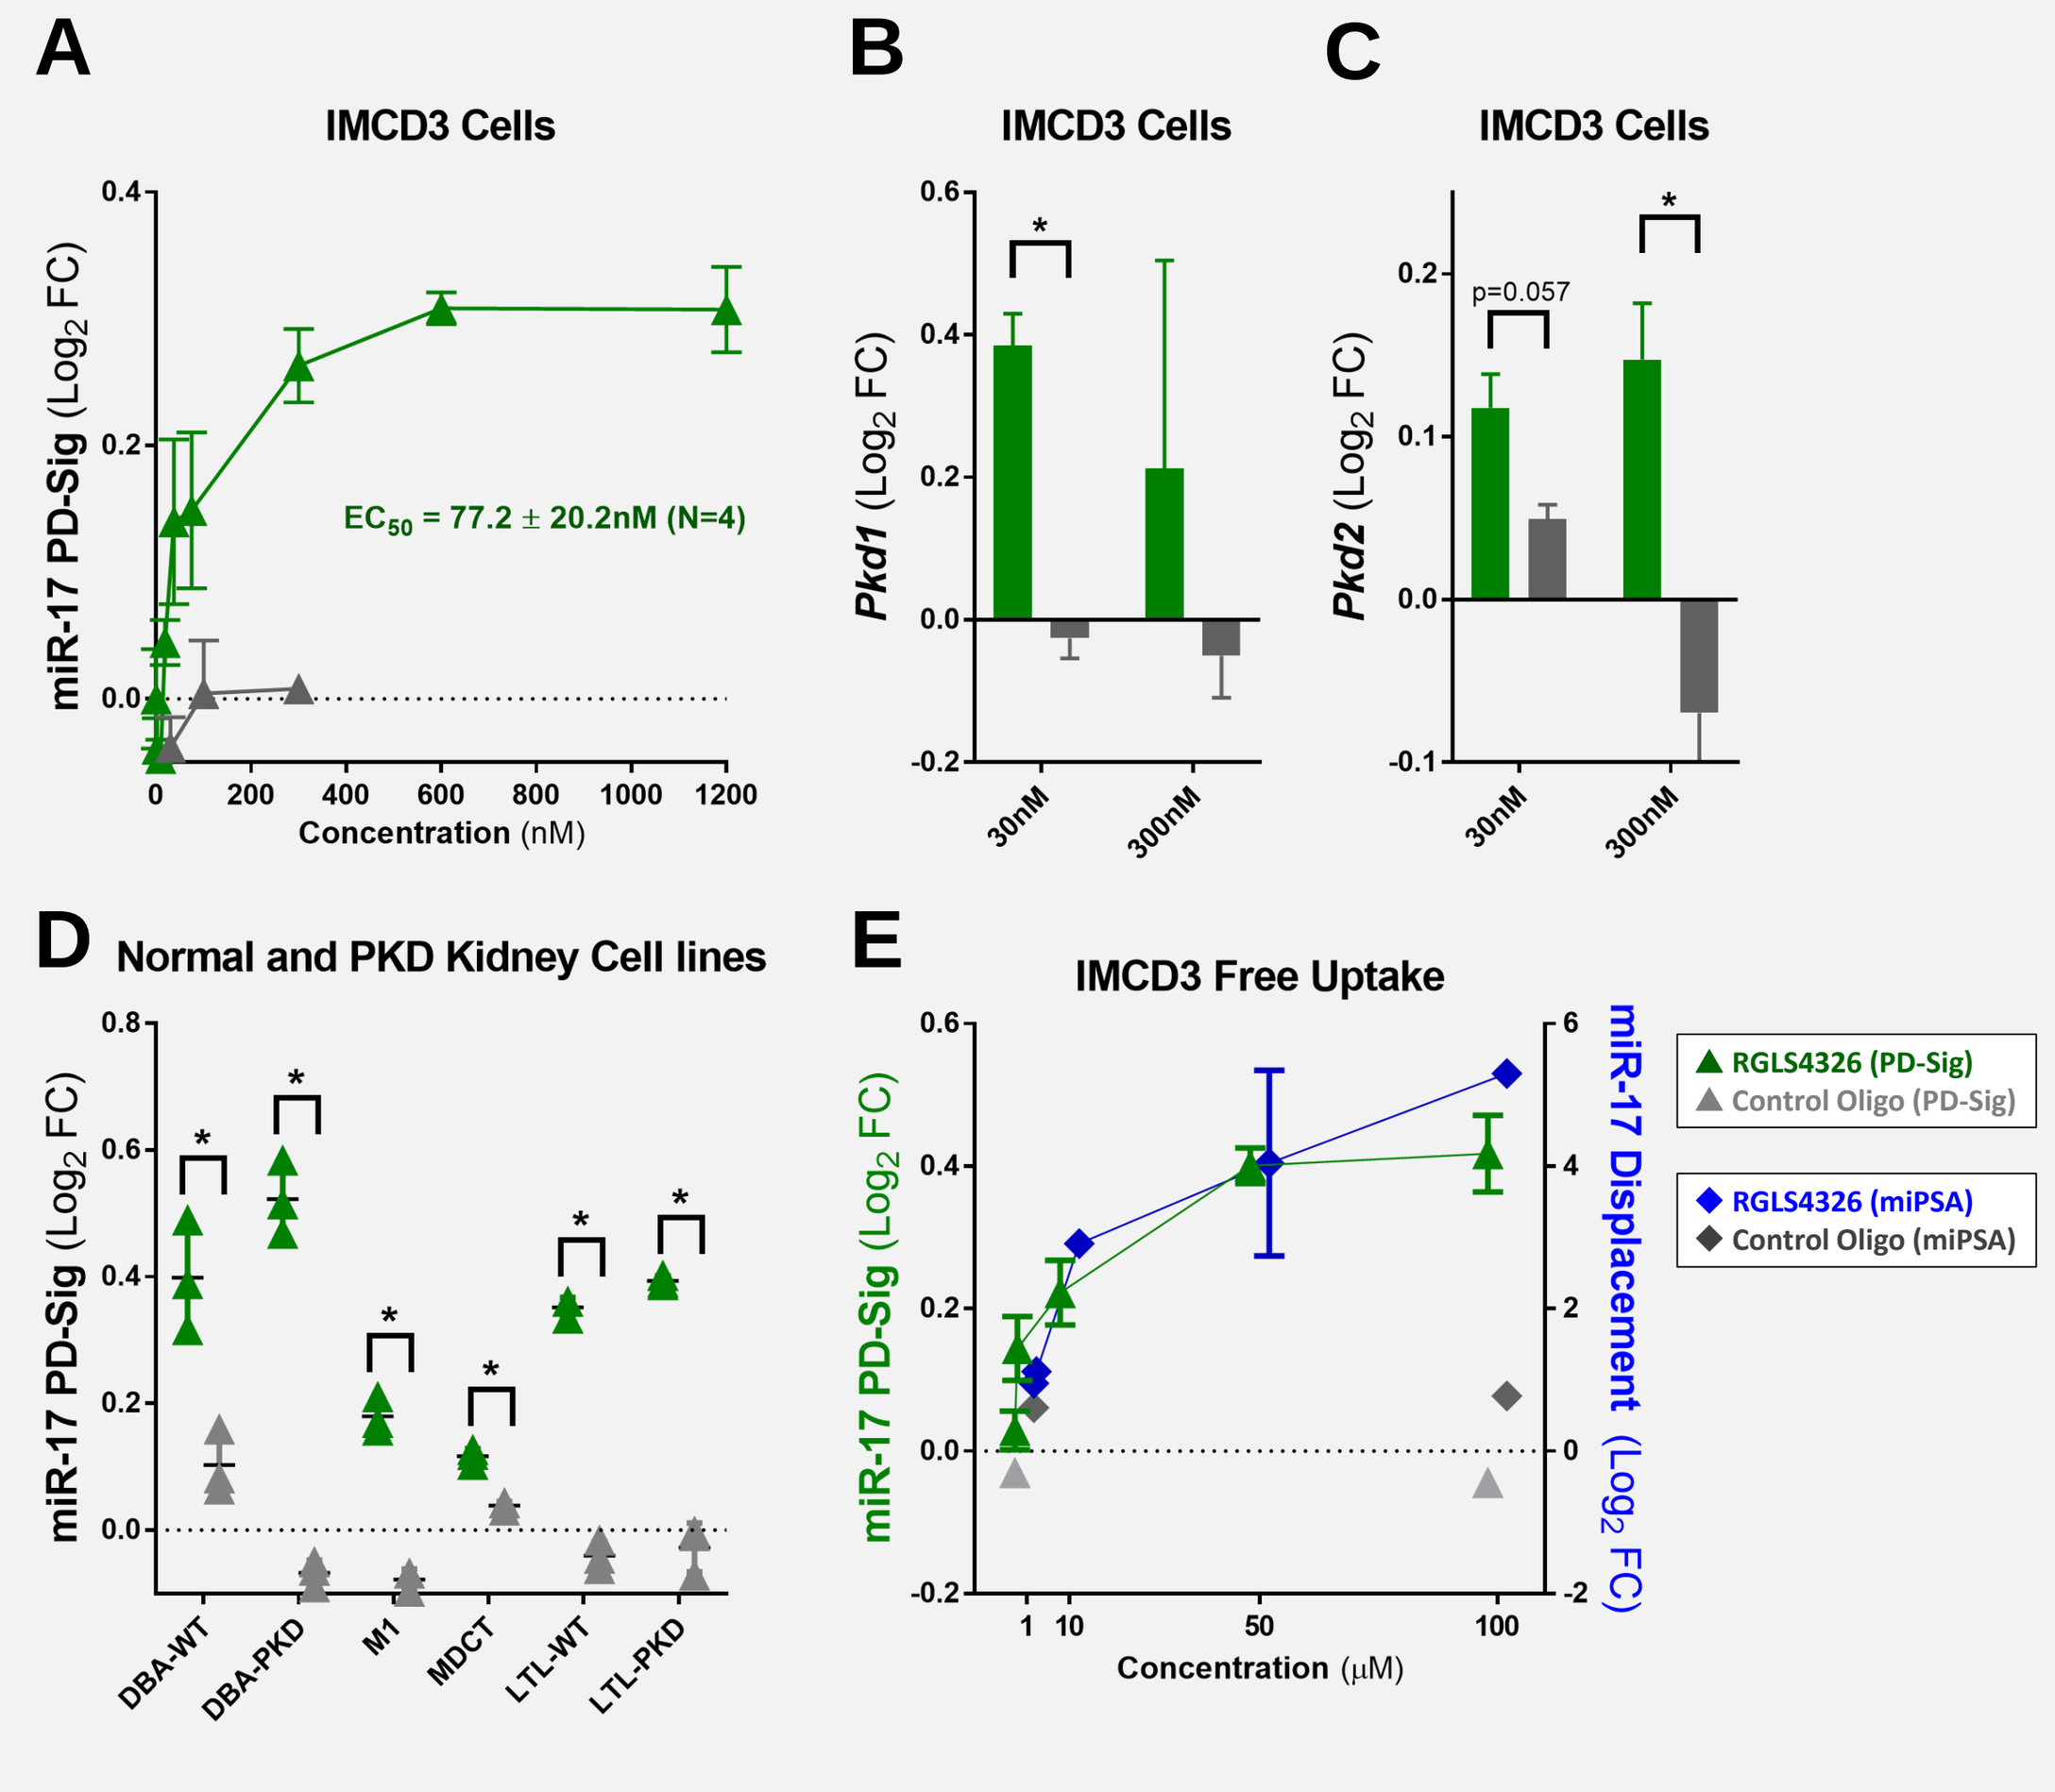
A
IMCD3 Cells
miR-17 PD-Sig (Log2 FC)
EC50 = 77.2 ± 20.2nM (N=4)
Concentration (nM)
B
IMCD3 Cells
Pkd1 (Log2 FC)
C
IMCD3 Cells
Pkd2 (Log2 FC)
p=0.057
30nM
300nM
D
Normal and PKD Kidney Cell lines
DBA-WT
DBA-PKD
M1
MDCT
LTL-WT
LTL-PKD
E
IMCD3 Free Uptake
miR-17 Displacement (Log2 FC)
RGLS4326 (PD-Sig)
Control Oligo (PD-Sig)
RGLS4326 (miPSA)
Control Oligo (miPSA)
Concentration (μM)

(A) RGLS4326 transfection de-repressed multiple miR-17 downstream target genes (measured by miR-17 PD-Sig, which contains 18 miR-17 target genes normalized by 6 housekeeping genes) in IMCD3 cells after 24h.
(B-C) RGLS4326 transfection also de-repressed expression of the miR-17 direct target genes *Pkd1* and *Pkd2* in IMCD3 cells after 24h.
(D) De-repression of miR-17 PD-Sig in different mouse kidney cell lines after transfection with RGLS4326 at 30nM.
(E) Target engagement of miR-17 (as measured by miPSA; Androsavich 2016) corresponded with de-repression of miR-17 PD-Sig after 24h of RGLS4326 treatment by free-uptake.

RGLS4326 shows favorable pharmacokinetic and pharmacodynamic profiles

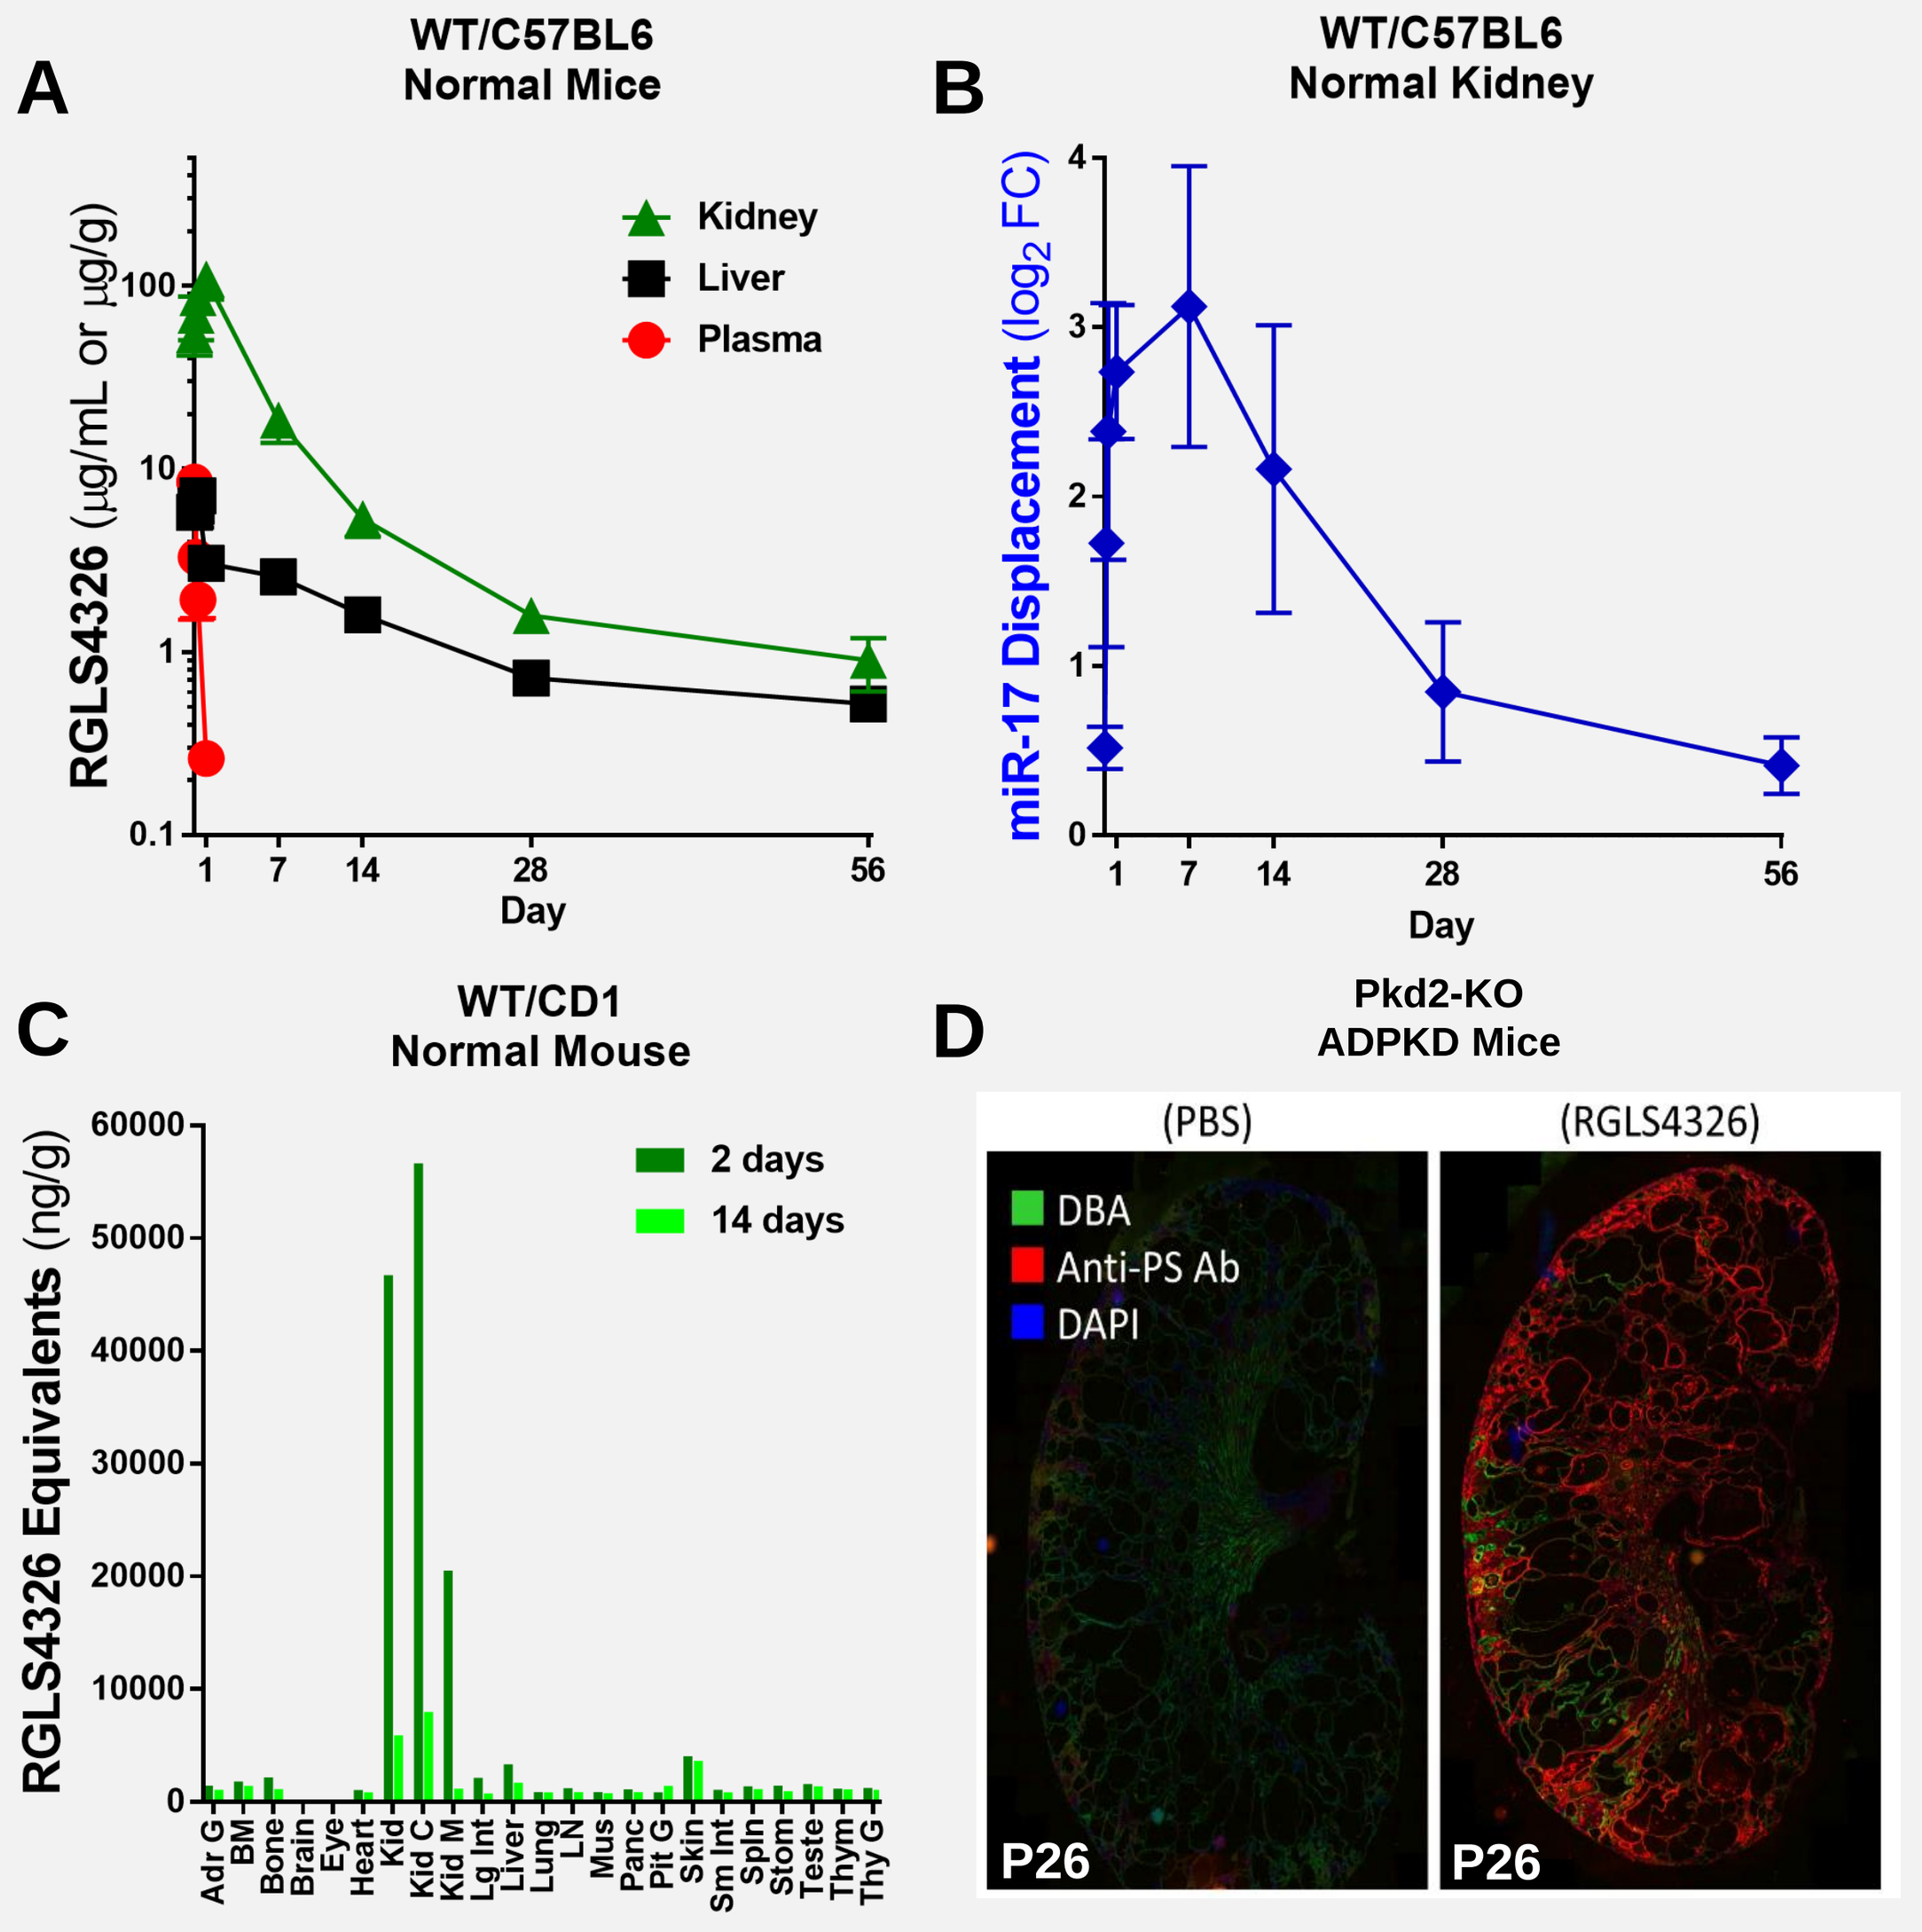
A
WT/C57BL6
Normal Mice
RGLS4326 (μg/mL or μg/g)
Kidney
Liver
Plasma
Day
B
WT/C57BL6
Normal Kidney
miR-17 Displacement (log2 FC)
Day
C
WT/CD1
Normal Mouse
RGLS4326 Equivalents (ng/g)
2 days
14 days
Adr G
BM
Bone
Brain
Eye
Heart
Kid
Kid C
Kid M
Lg Int
Liver
Lung
LN
Mus
Panc
Pit G
Skin
Sm Int
Spln
Stom
Teste
Thym
Thy G
D
Pkd2-KO
ADPKD Mice
(PBS)
(RGLS4326)
DBA
Anti-PS Ab
DAPI
P26
P26

(A) After a single SC 30mg/kg dose, RGLS4326 was rapidly absorbed into and cleared from plasma. RGLS4326 distributed primarily to kidney, with kidney-to-liver ratio of >10-fold by C_{max}.
(B) RGLS4326 potently engaged kidney miR-17, with peak target engagement (by miPSA) observed at 7 days after a single SC dose at 30mg/kg.
(C) Quantitative whole-body autoradiography of [^{35}S]-RGLS4326-derived radioactivity in male WT/CD1 mice showed RGLS4326 preferentially distributed to kidney over other organs after a single 30mg/kg SC dose.
(D) Immunofluorescence data showing localization of RGLS4326 (RED) to kidney collecting duct cyst cells (GREEN) in *Pkd2*-KO mice after repeat SC dosing at 20mg/kg on postnatal day (P)21, P22 and P23 where numerous kidneys cyst have already formed.

RGLS4326 shows efficacy in the *Pkd2*-KO mouse model of ADPKD

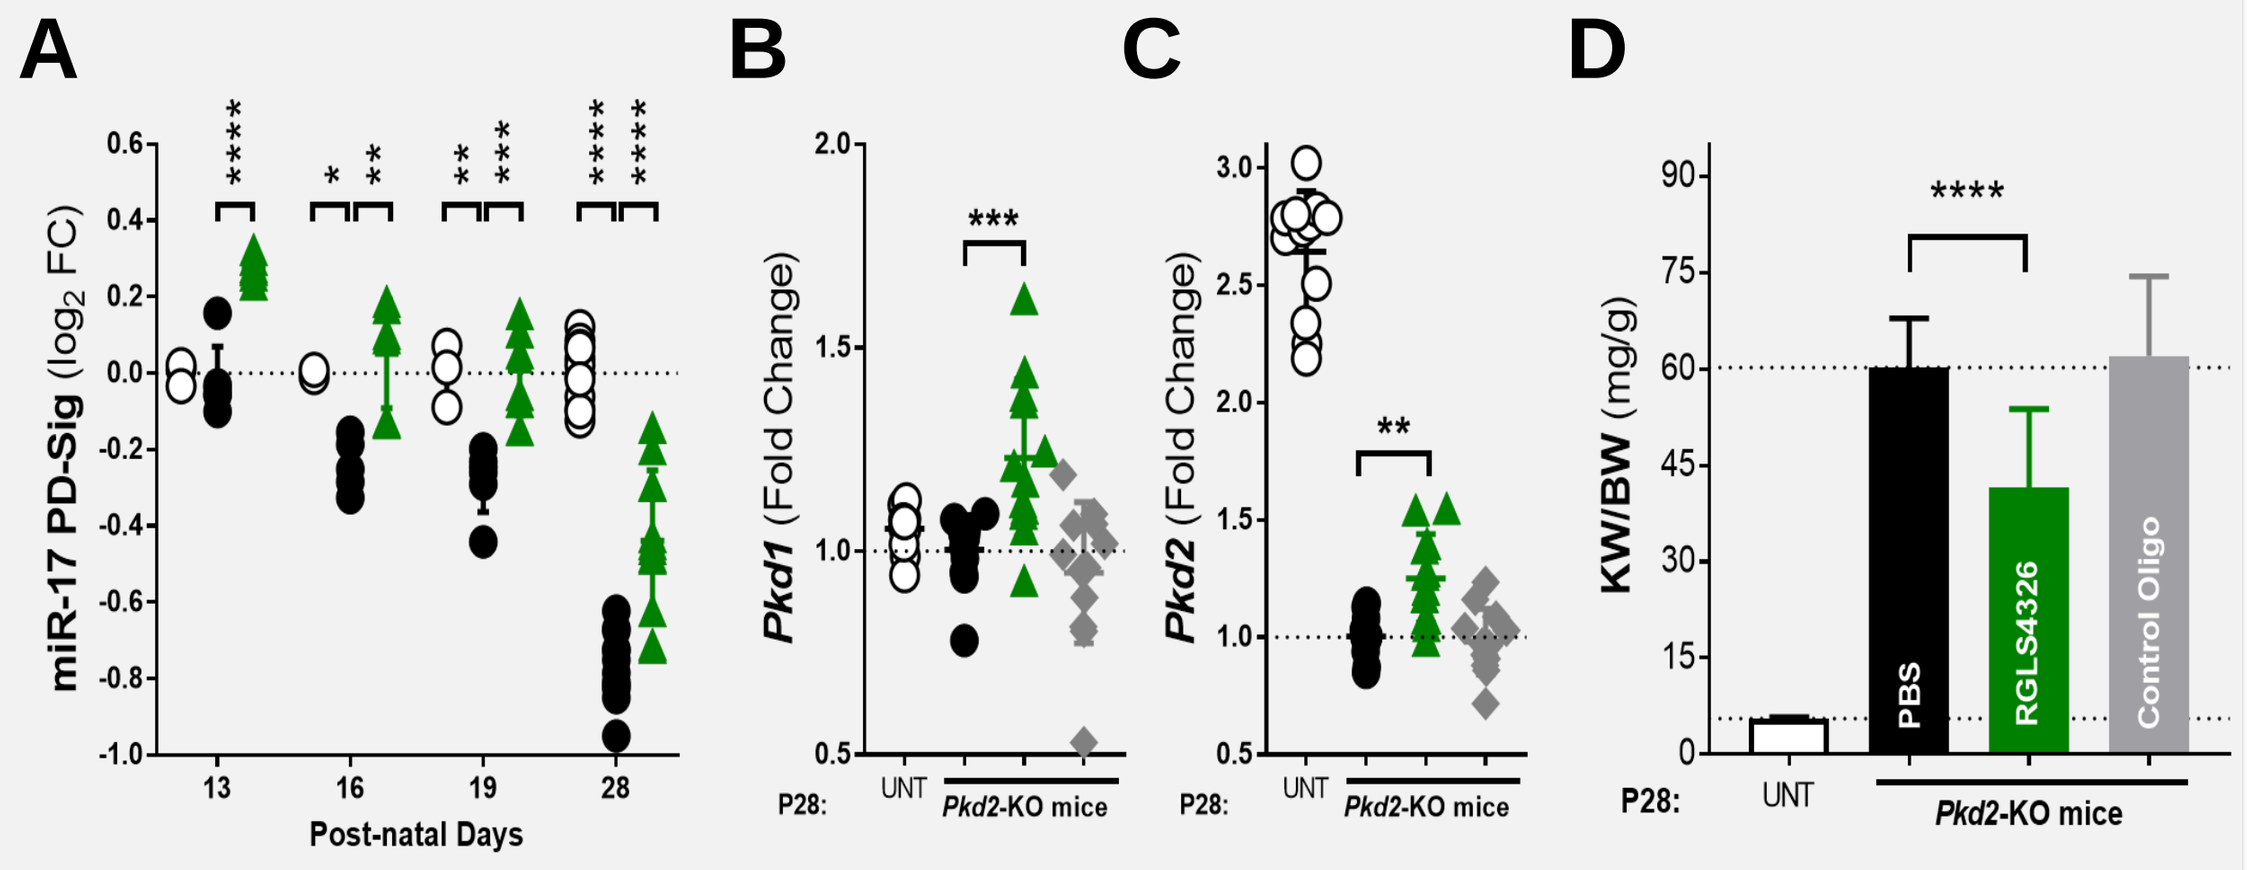
A
miR-17 PD-Sig (log2 FC)
Post-natal Days
B
Pkd1 (Fold Change)
C
Pkd2 (Fold Change)
D
KW/BW (mg/g)
PBS
RGLS4326
Control Oligo
P28:
UNT
Pkd2-KO mice

Pkd2-KO mice were dosed SC with PBS or 20 mg/kg of RGLS4326 or control oligo on postnatal day (P)10, P11, P12 and P19. Kidneys were harvested on P13, P16, P19 and P28. Age-matched untreated non-transgenic (UNT) control mice were also included for comparison.
(A-C) *Pkd2*-KO kidneys showed low PD-sig compared to UNT, indicative of high baseline miR-17 functional activity, while RGLS4326 treatment in *Pkd2*-KO mice de-repressed multiple miR-17 downstream target genes (miR-17 PD-Sig), including *Pkd1* and *Pkd2*.
(D) Treatment with RGLS4326, but not control oligo, conferred efficacy and reduced kidney-weight-to-body-weight (KW/BW) ratio in *Pkd2*-KO model.

RGLS4326 shows efficacy in the *Pcy*/DBA mouse model of PKD

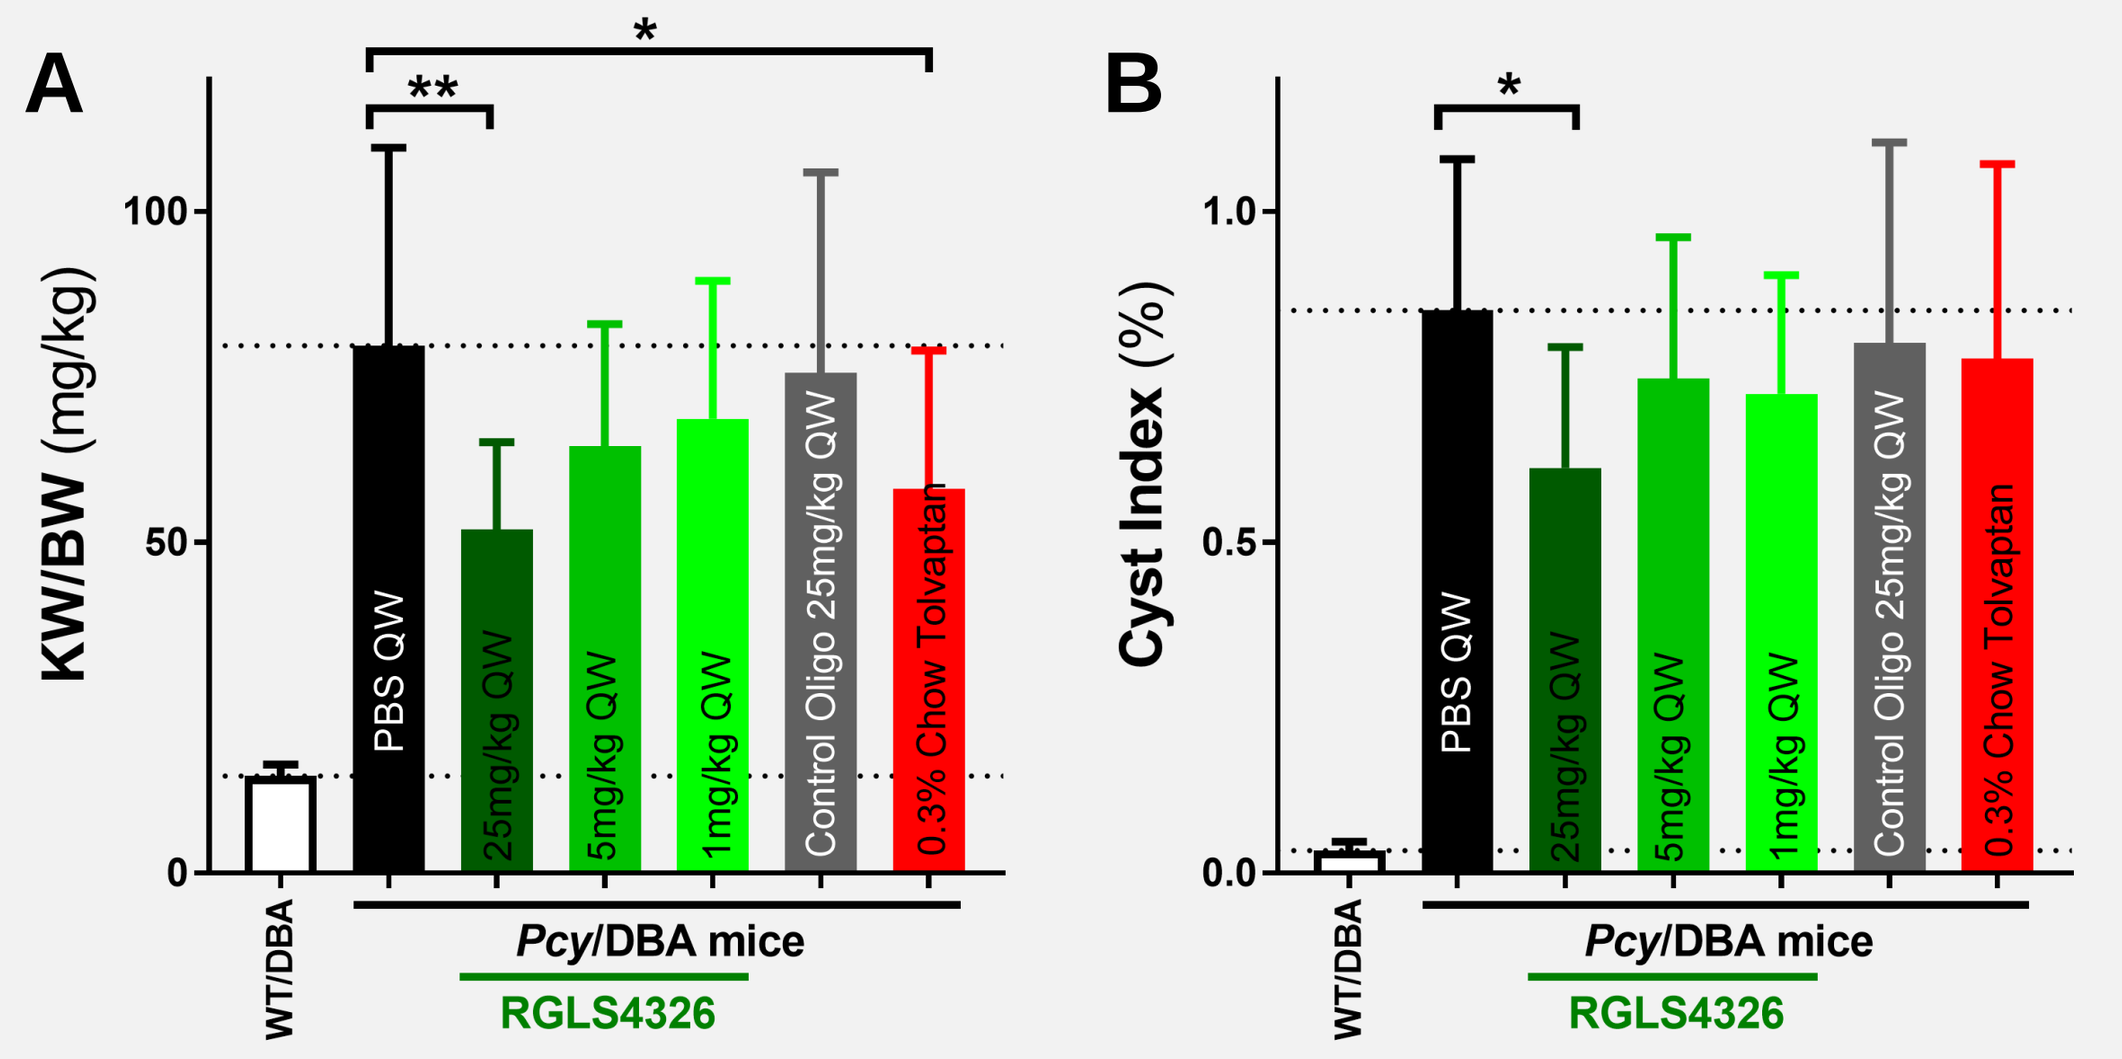
A
KW/BW (mg/kg)
B
Cyst Index (%)
PBS QW
25mg/kg QW
5mg/kg QW
1mg/kg QW
Control Oligo 25mg/kg QW
0.3% Chow Tolvaptan
WT/DBA
Pcy/DBA mice
RGLS4326

Six-weeks-old *Pcy*/DBA mice were dosed SC QW with PBS, or 25, 5 or 1 mg/kg of RGLS4326, or 25 mg/kg of control oligos for 9 weeks. An additional group of mice were treated with 0.3% Tolvaptan via diet from 6 to 15 weeks of age. All mice were euthanized at 15-weeks of age.
(A-B) KW/BW ratio and cyst index were significantly reduced following weekly RGLS4326, but not control oligo, treatment compared to PBS. Tolvaptan conferred efficacy based on KW/BW ratio, but not cyst index.

RGLS4326 reduces rate of bwTKV increase by MRI in the *Pcy*/DBA mouse model of PKD

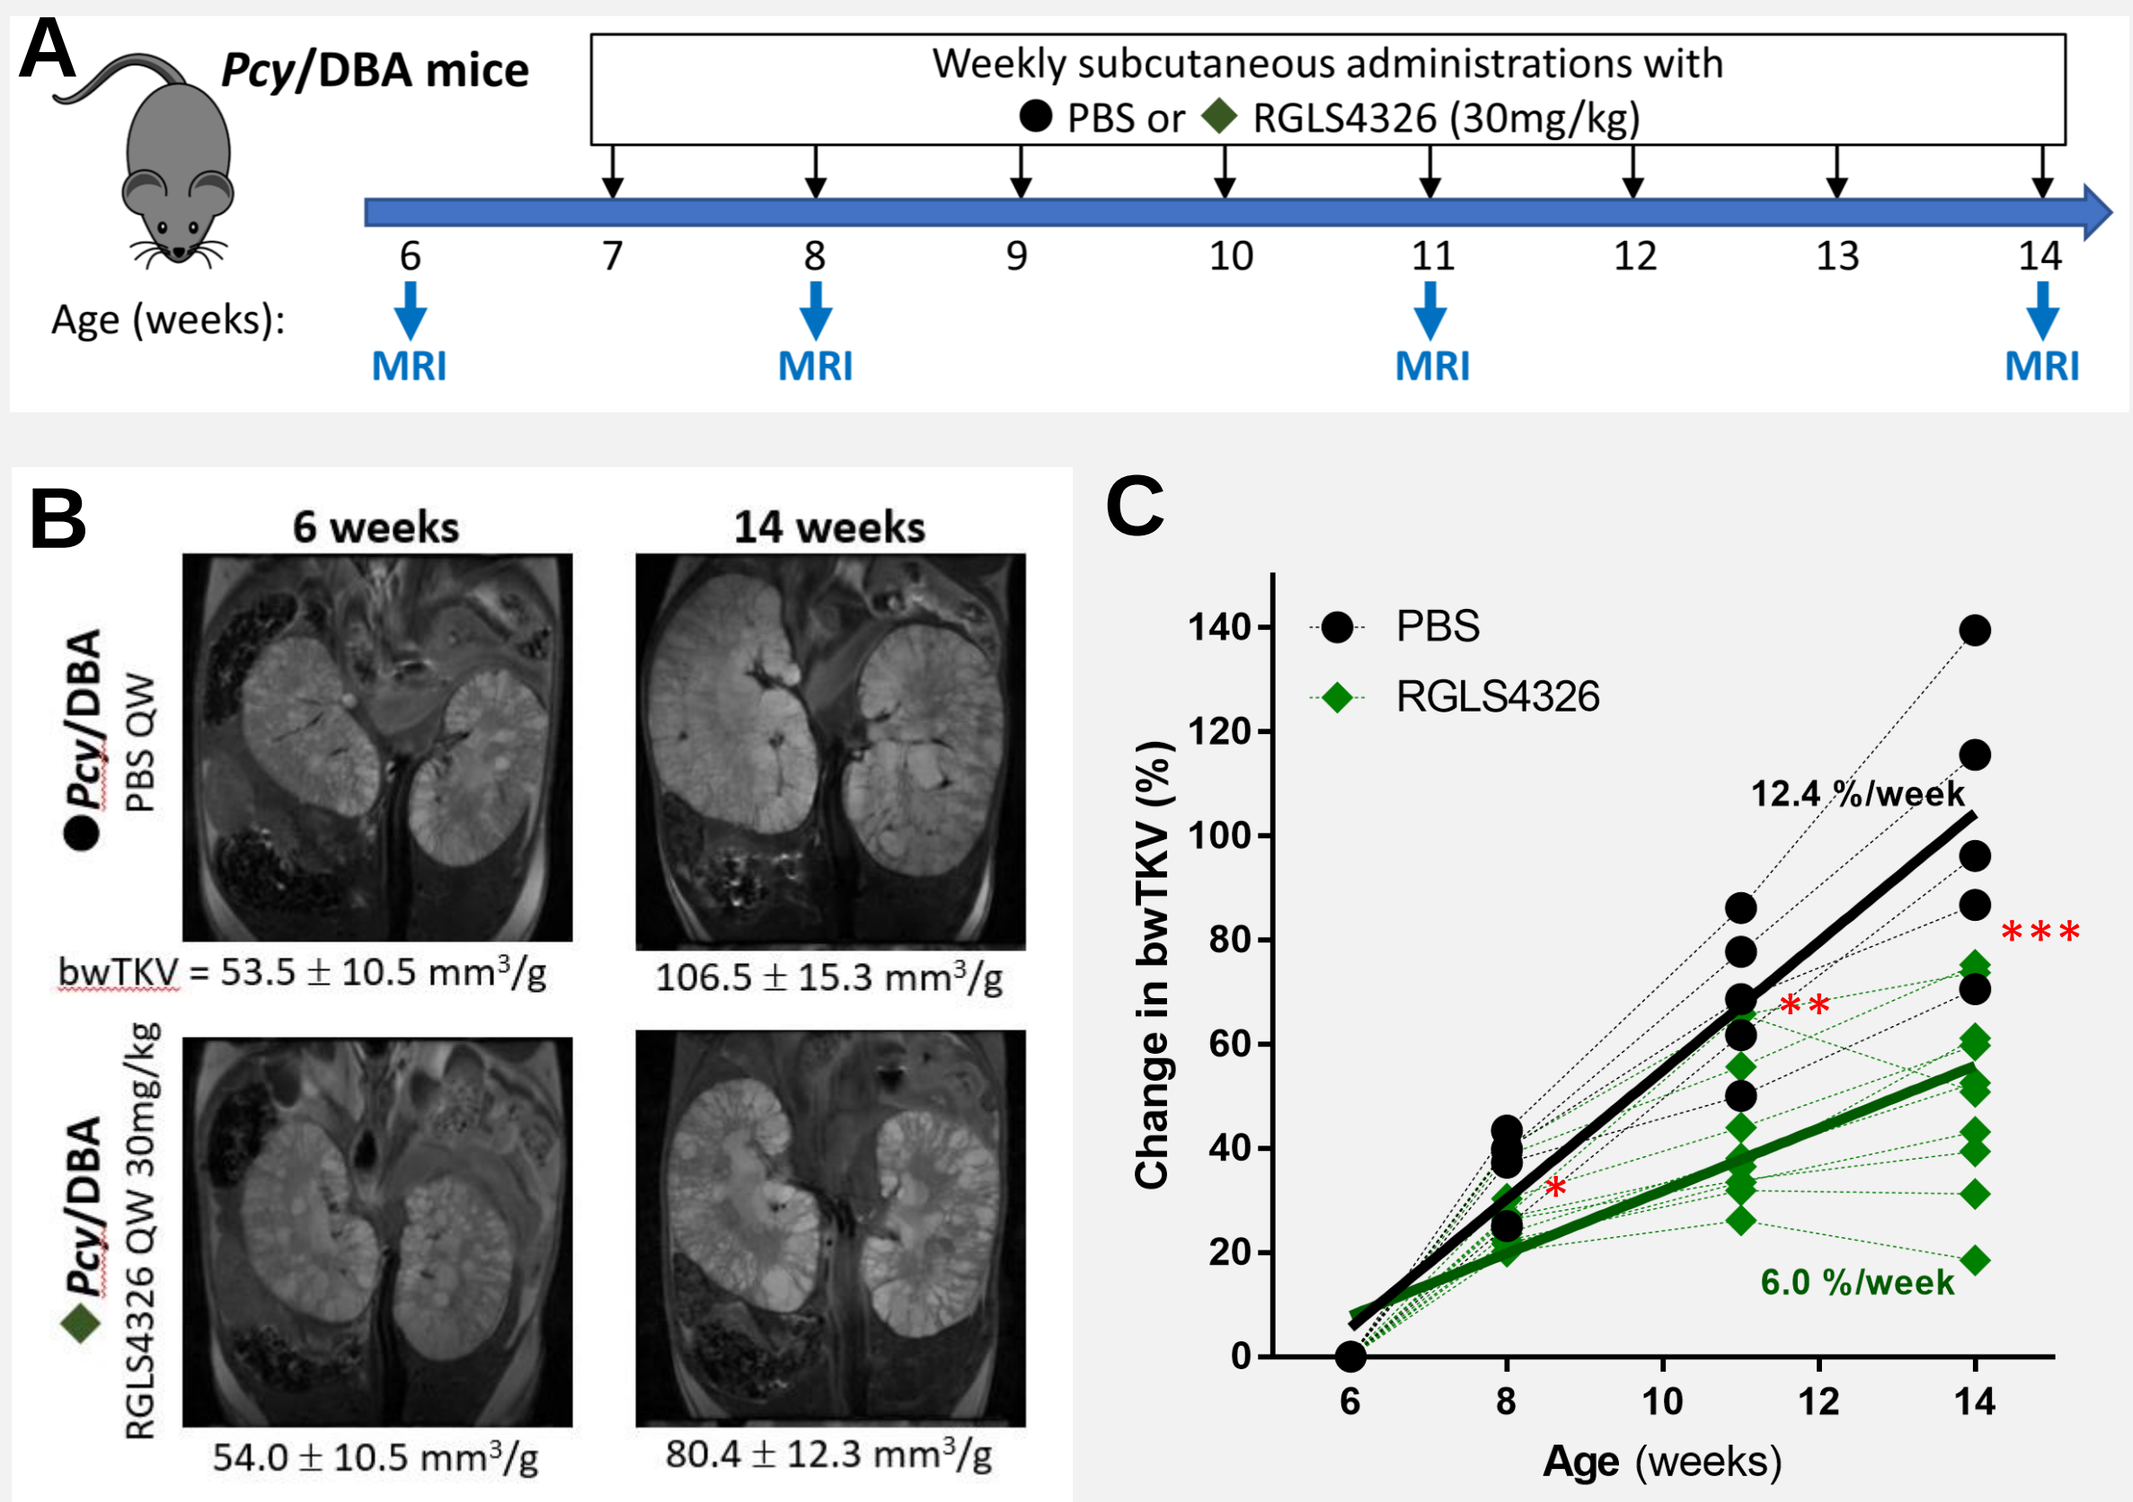
A
Pcy/DBA mice
Weekly subcutaneous administrations with
PBS or RGLS4326 (30mg/kg)
Age (weeks):
MRI
B
6 weeks
14 weeks
Pcy/DBA PBS QW
bwTKV = 53.5 ± 10.5 mm3/g
106.5 ± 15.3 mm3/g
Pcy/DBA RGLS4326 QW 30mg/kg
54.0 ± 10.5 mm3/g
80.4 ± 12.3 mm3/g
C
PBS
RGLS4326
12.4 %/week
6.0 %/week
Change in bwTKV (%)
Age (weeks)

(A) Baseline total kidney volume (TKV) were obtained from 6-weeks-old male Pcy/DBA mice by T2-weighted MRI and used to assign treatment groups.
(B) Representative MRI images at 6 and 14 weeks. Body weight-adjusted TKV (bwTKV; Mean±SD) were shown.
(C) Changes in bwTKV from baseline at week 8, 11 and 14 was significantly reduced following RGLS4326 treatment by 25%, 37% and 50%, respectively (p<0.05 for all). RGLS4326 treatment reduced rate of weekly bwTKV increase by 52% compared to PBS (from 12.4%/week to 6.0%/week; p<0.0001).

American Society of Nephrology - Kidney Week, San Diego, CA (Oct 23 - 28, 2018)

Abstract Number: 3022832; Poster Board Number: TH-PO685