RGLS4326 confers efficacy and modulate aberrant signaling and metabolic pathways in PKD mouse models

Tania M. Valencia[1,*], Sole Gatto[1], Annelie E. Schairer[1], Steven Lockton[1], Michael Kim[1], Kara Kersjes[1], Jian Li[1], Andrea N. Flaten[2], Vishal Patel[2], Edmund C. Lee[1]

[1]Regulus Therapeutics Inc., San Diego, California. *Presenting author

[2]Department of Internal Medicine and Division of Nephrology, University of Texas Southwestern Medical Center, Dallas, Texas.

Abstract

Background:
Autosomal dominant polycystic kidney disease (*ADPKD)* is caused by mutations in either *PKD1* or *PKD2* genes, where expansion of fluid-filled cysts and renal fibrosis often leads to end-stage renal disease. MicroRNAs are short non-coding RNAs that modulate several biological processes. We have previously shown that aberrant expression of miR-17 family of microRNAs is involved in human ADPKD pathogenesis. RGLS4326 is a chemically-modified oligonucleotide designed to sterically inhibit miR-17 functions and has been shown to reduce cyst growth *in vitro* and *in vivo*. The goal of this study was to determine the signaling pathways modulated by RGLS4326 treatment in PKD mouse models

Methods:
RGLS4326 is efficacious in the *Pkd2KO* and *Pcy*/CD-1 mouse models of PKD. We performed RNA sequencing and metabolite profiling using kidney samples from both mouse models following RGLS4326 treatment. Ingenuity Pathway Analysis was used to provide novel insights into signaling pathways modulated by RGLS4326 treatment

Results:
Through RNA sequencing, we identified >1000 differentially expressed genes in the PKD kidney samples compared to their age- and strain-matched normal controls. Comparative pathway analysis identified several dysregulated signaling pathways in the two PKD mouse models, including the Pparα, WNT/ β-catenin and PCP signaling, that were in turn modulated following RGLS4326 treatment. Next, we performed kidney global metabolite profiling comparing *Pkd2KO* and normal kidneys, and identified several biochemical alterations in the *Pkd2KO* model, including substantial changes in lipid metabolism. In particular, decrease in β-fatty acid oxidation pathway was observed in the *Pkd2KO* kidneys, which corroborates with previously observed Pparα-dysregulation in PKD mouse models

Conclusion: Our results indicate that RGLS4326 confers efficacy and modulates aberrant PKD signaling and metabolic pathways. These results support the clinical development of RGLS4326 for the treatment of ADPKD

miRNA targets multiple signaling pathways

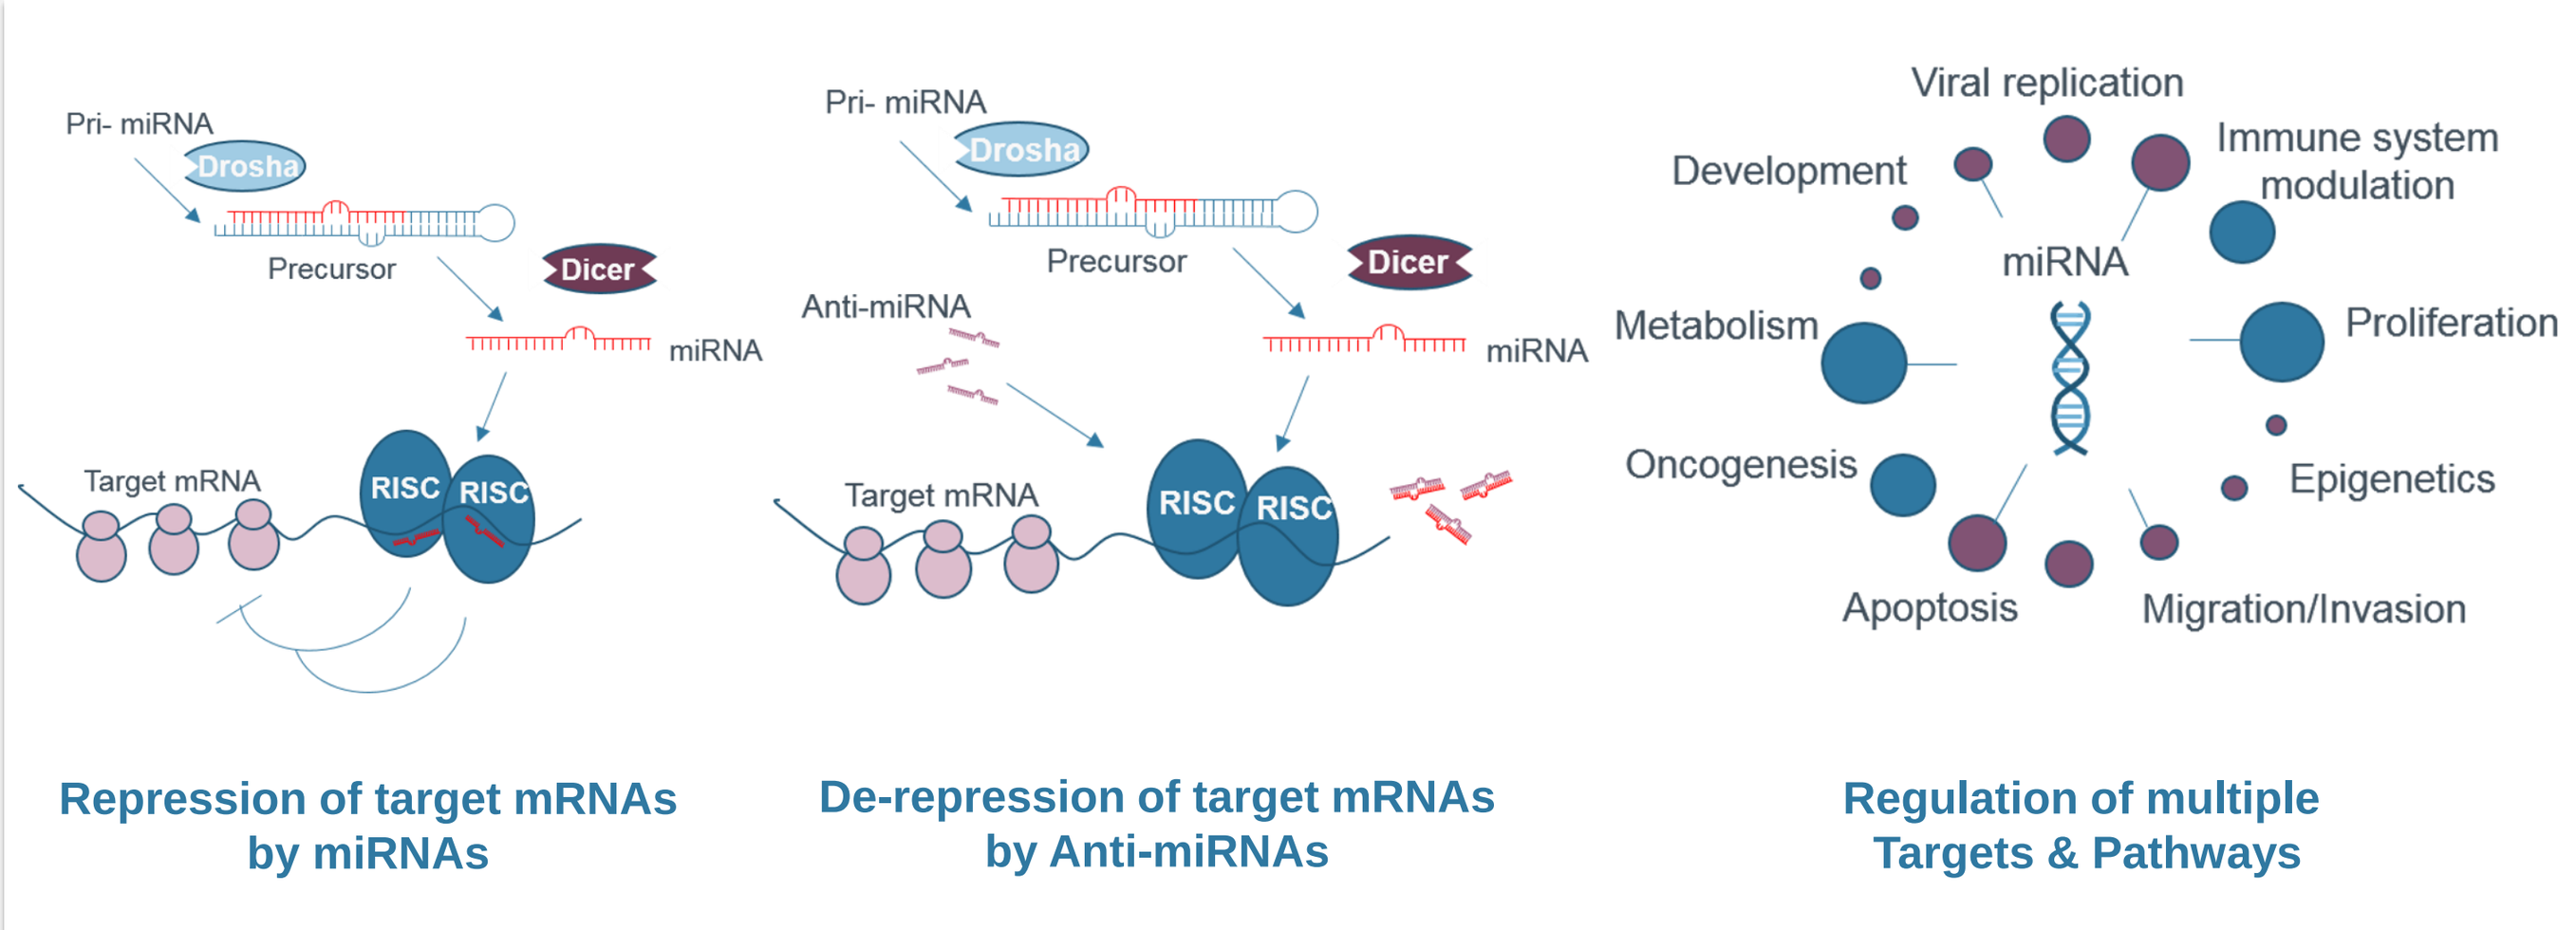
Pri- miRNA
Drosha
Precursor
Dicer
miRNA
Target mRNA
RISC
RISC
Anti-miRNA
Viral replication
Development
Immune system modulation
miRNA
Metabolism
Proliferation
Oncogenesis
Epigenetics
Apoptosis
Migration/Invasion
Repression of target mRNAs by miRNAs
De-repression of target mRNAs by Anti-miRNAs
Regulation of multiple Targets & Pathways

RGLS4326 is efficacious in *Pkd2KO* and *Pcy*/CD-1 mouse models

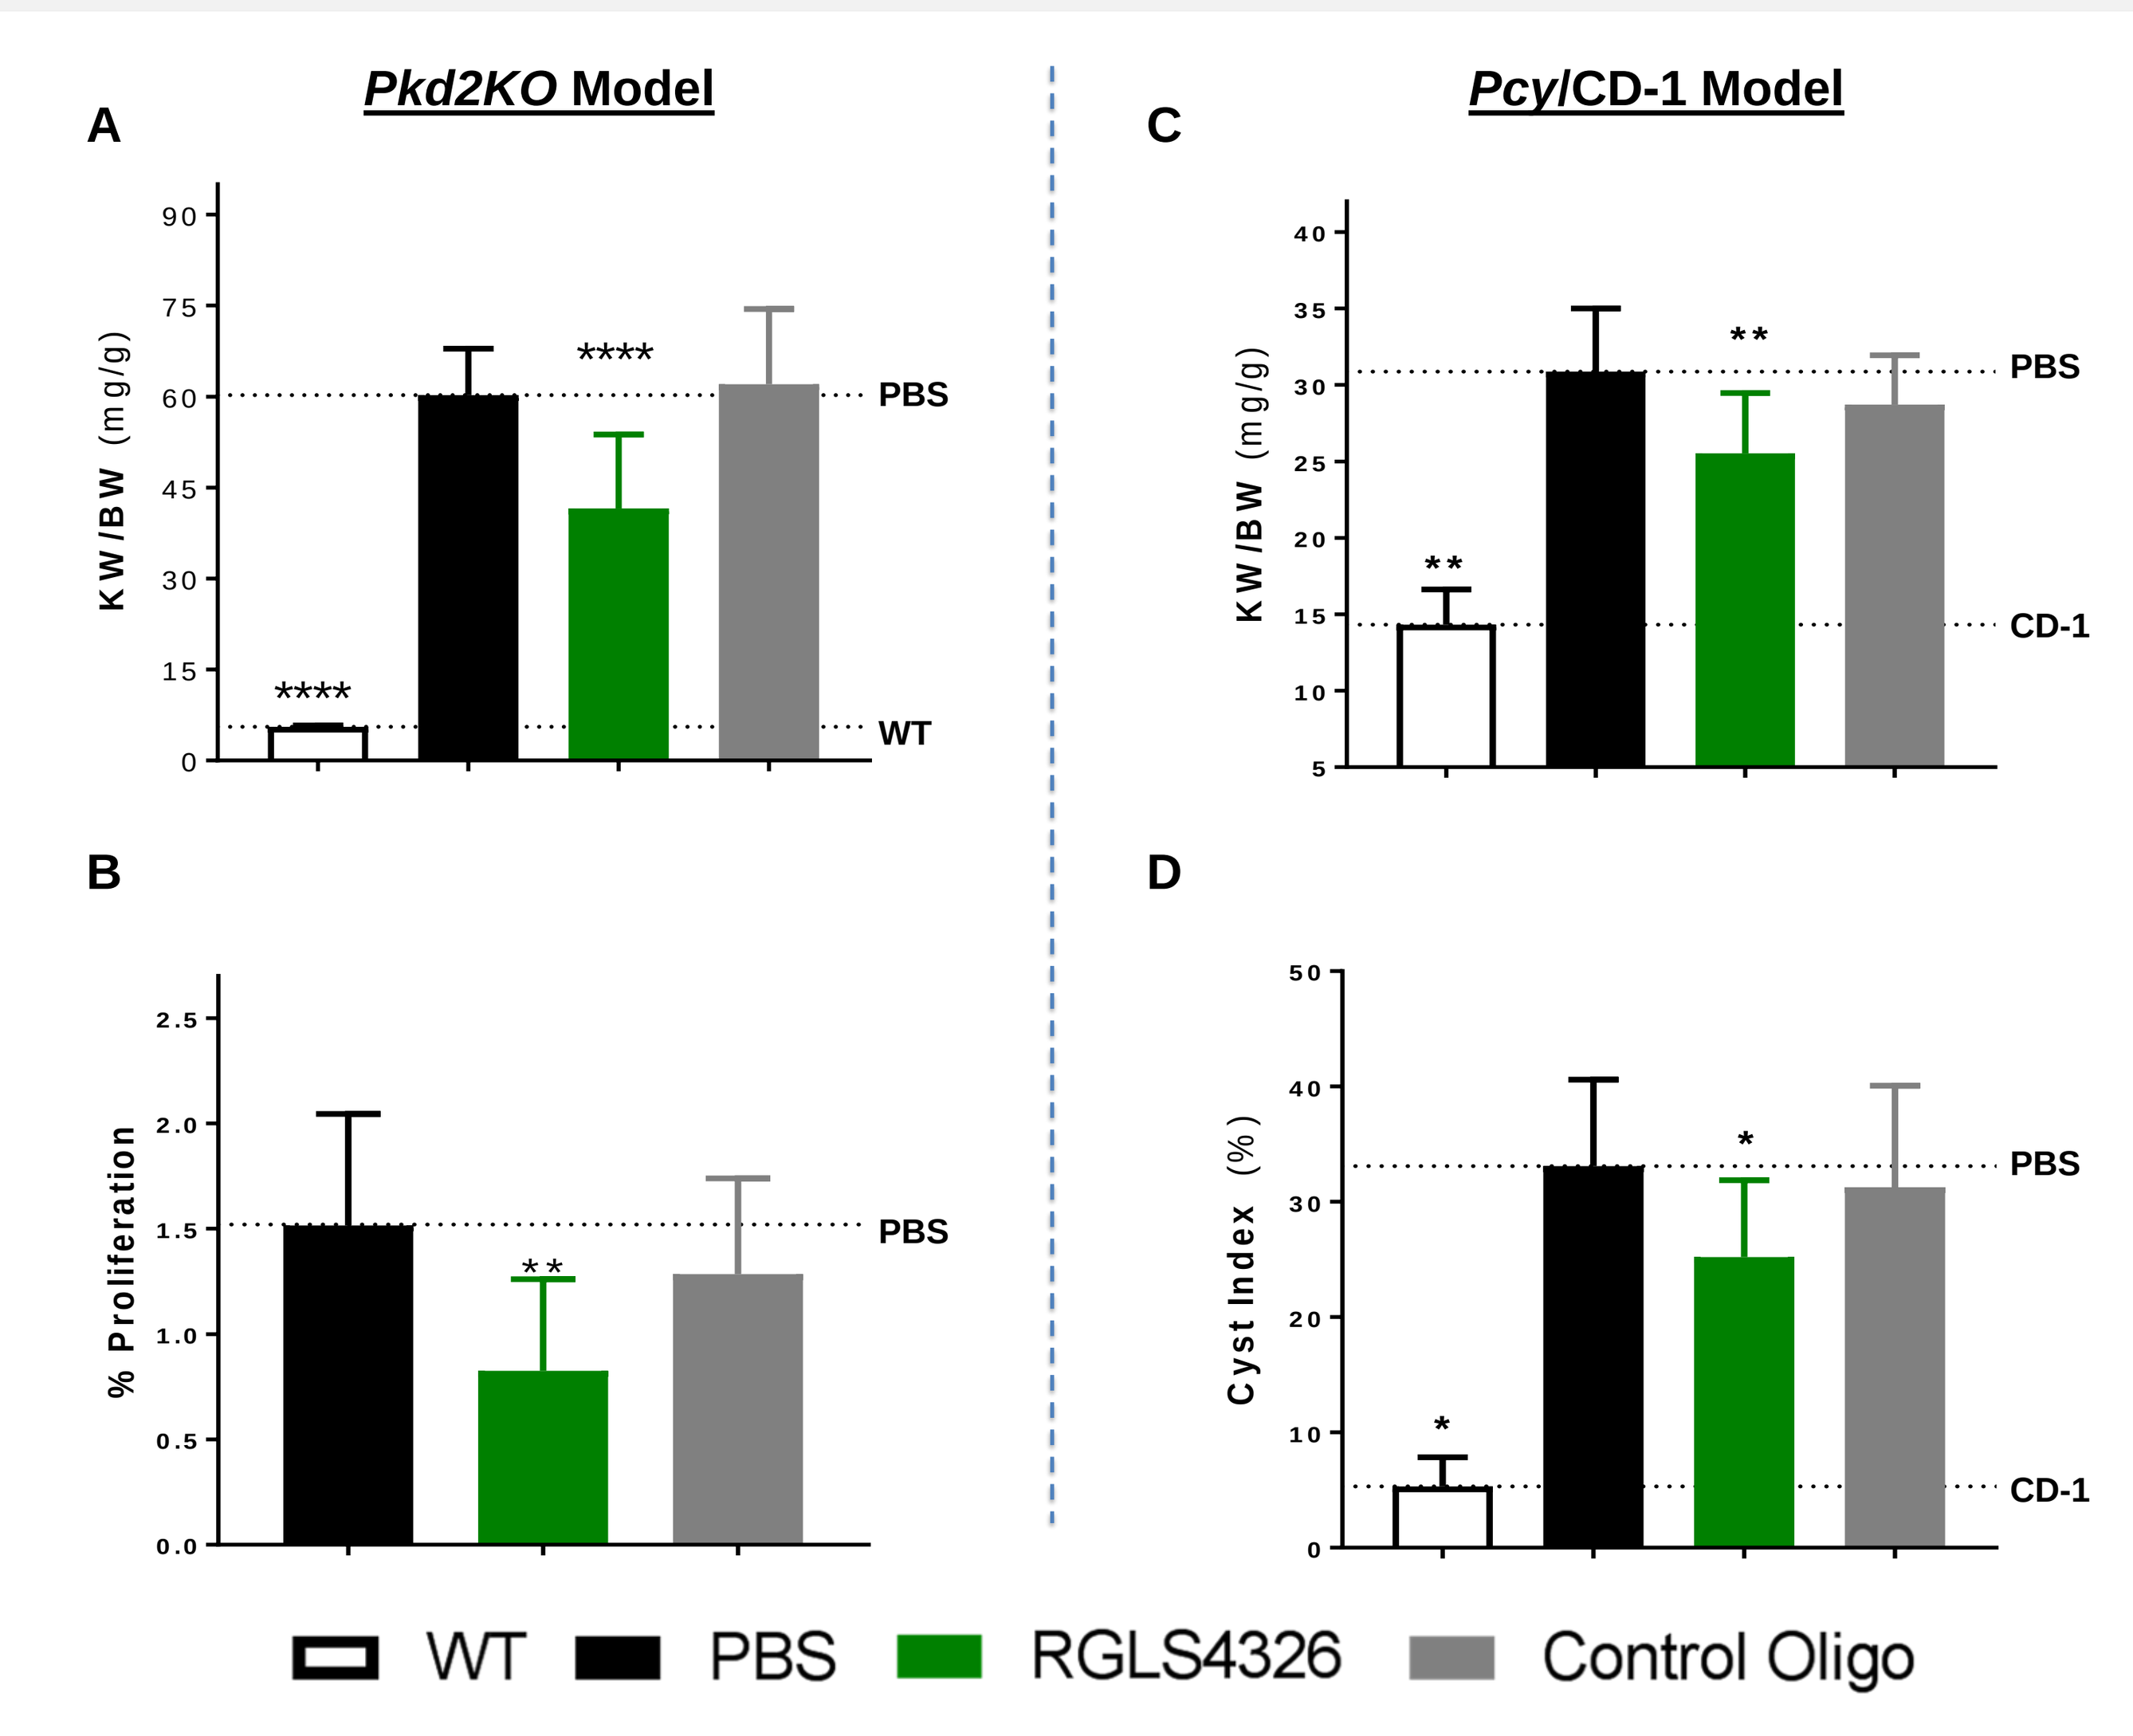
Pkd2KO Model
Pcy/CD-1 Model
A
B
C
D
KW/BW (mg/g)
% Proliferation
Cyst Index (%)
PBS
WT
CD-1

**
*
WT
PBS
RGLS4326
Control Oligo

(A) Decrease in Kidney-Weight-to-Body-Weight (KW/BW) in the *Pkd2KO* efficacy model by subcutaneous administration of RGLS4326 at 20 mg/kg on days P11, P12 and P13 and P19; Kidneys were collected at P28. **(B)** Reduction in proliferation after RGLS4326 treatment as shown by phospho-histone3 staining in the *Pkd2KO* model. **(C)** Decrease in KW/BW in the *Pcy*/CD-1 efficacy model following Q4W dose of RGLS4326 at 25 mg/kg from 5 to 30 weeks of age. **(D)** Reduction of cyst index after RGLS4326 treatment in the *Pcy*/CD-1 efficacy model

RNA Seq differential expression analysis in efficacy models

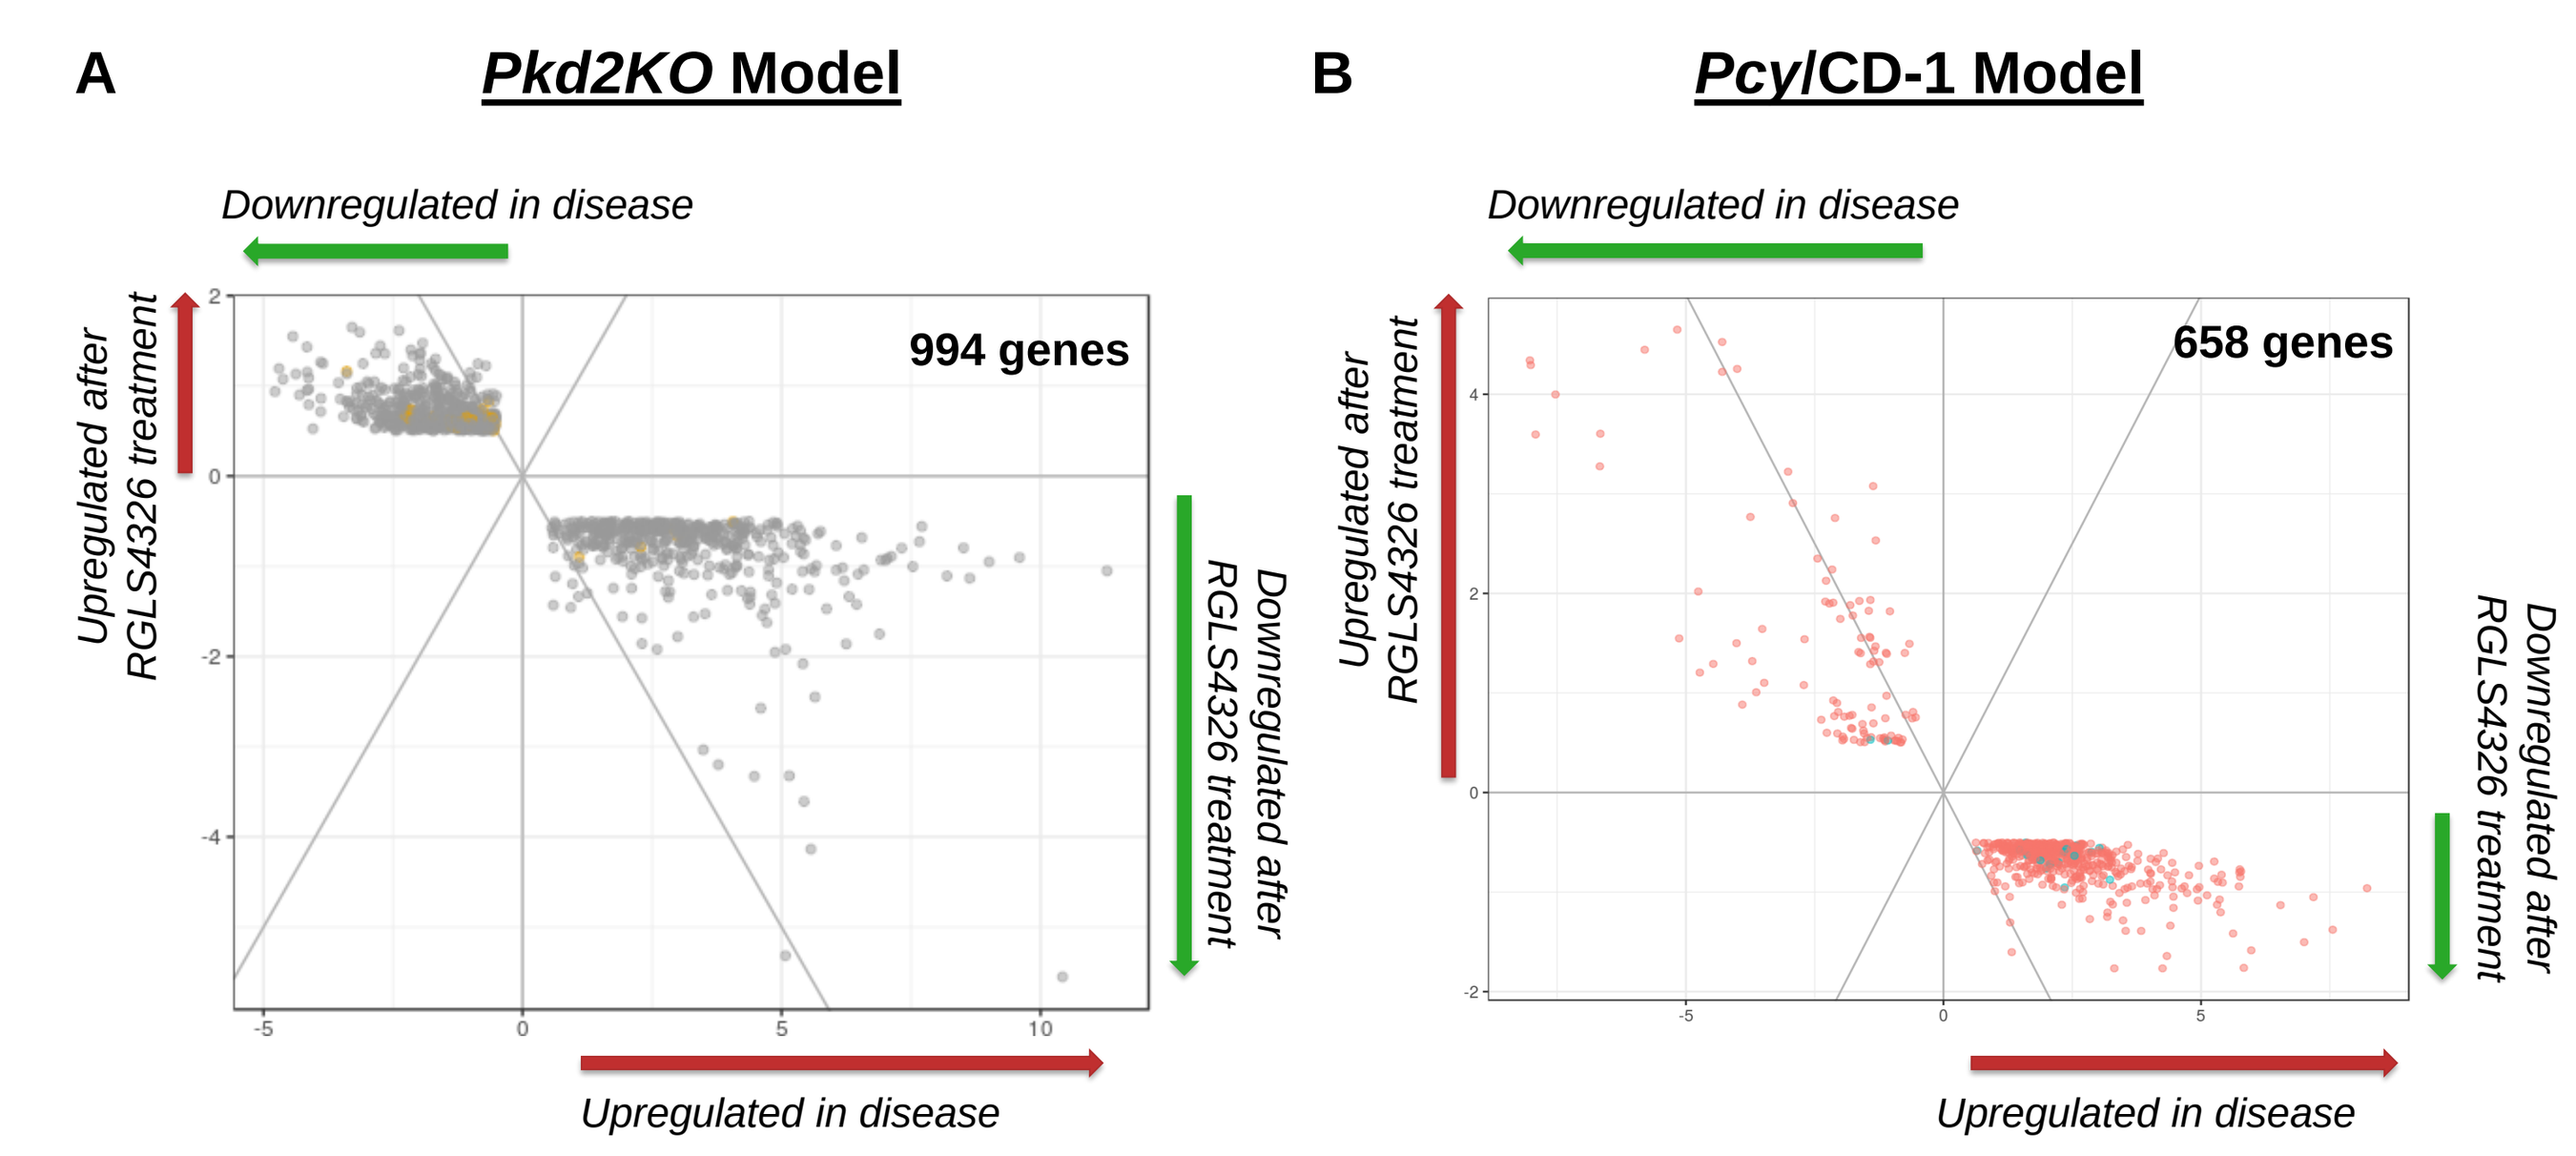
A
Pkd2KO Model
B
Pcy/CD-1 Model
Downregulated in disease
Upregulated after RGLS4326 treatment
Downregulated after RGLS4326 treatment
Upregulated in disease
994 genes
658 genes

RNAseq differential expression (DE) analysis of *Pkd2KO* and *Pcy*/CD-1 efficacy models. DE genes upregulated by disease and downregulated by RGLS4326 treatment and vice-versa for the *Pkd2KO* **(A)** and *Pcy*/CD-1 **(B)** efficacy models. Criteria FDR<0.05, Log2FC>0.5 in disease and <-0.5 in RGLS4326 vs PBS or Log2FC<-0.5 in disease and >0.5 in RGLS4326 vs PBS.

Comparative pathway analysis –*Pkd2KO* and *Pcy* efficacy models

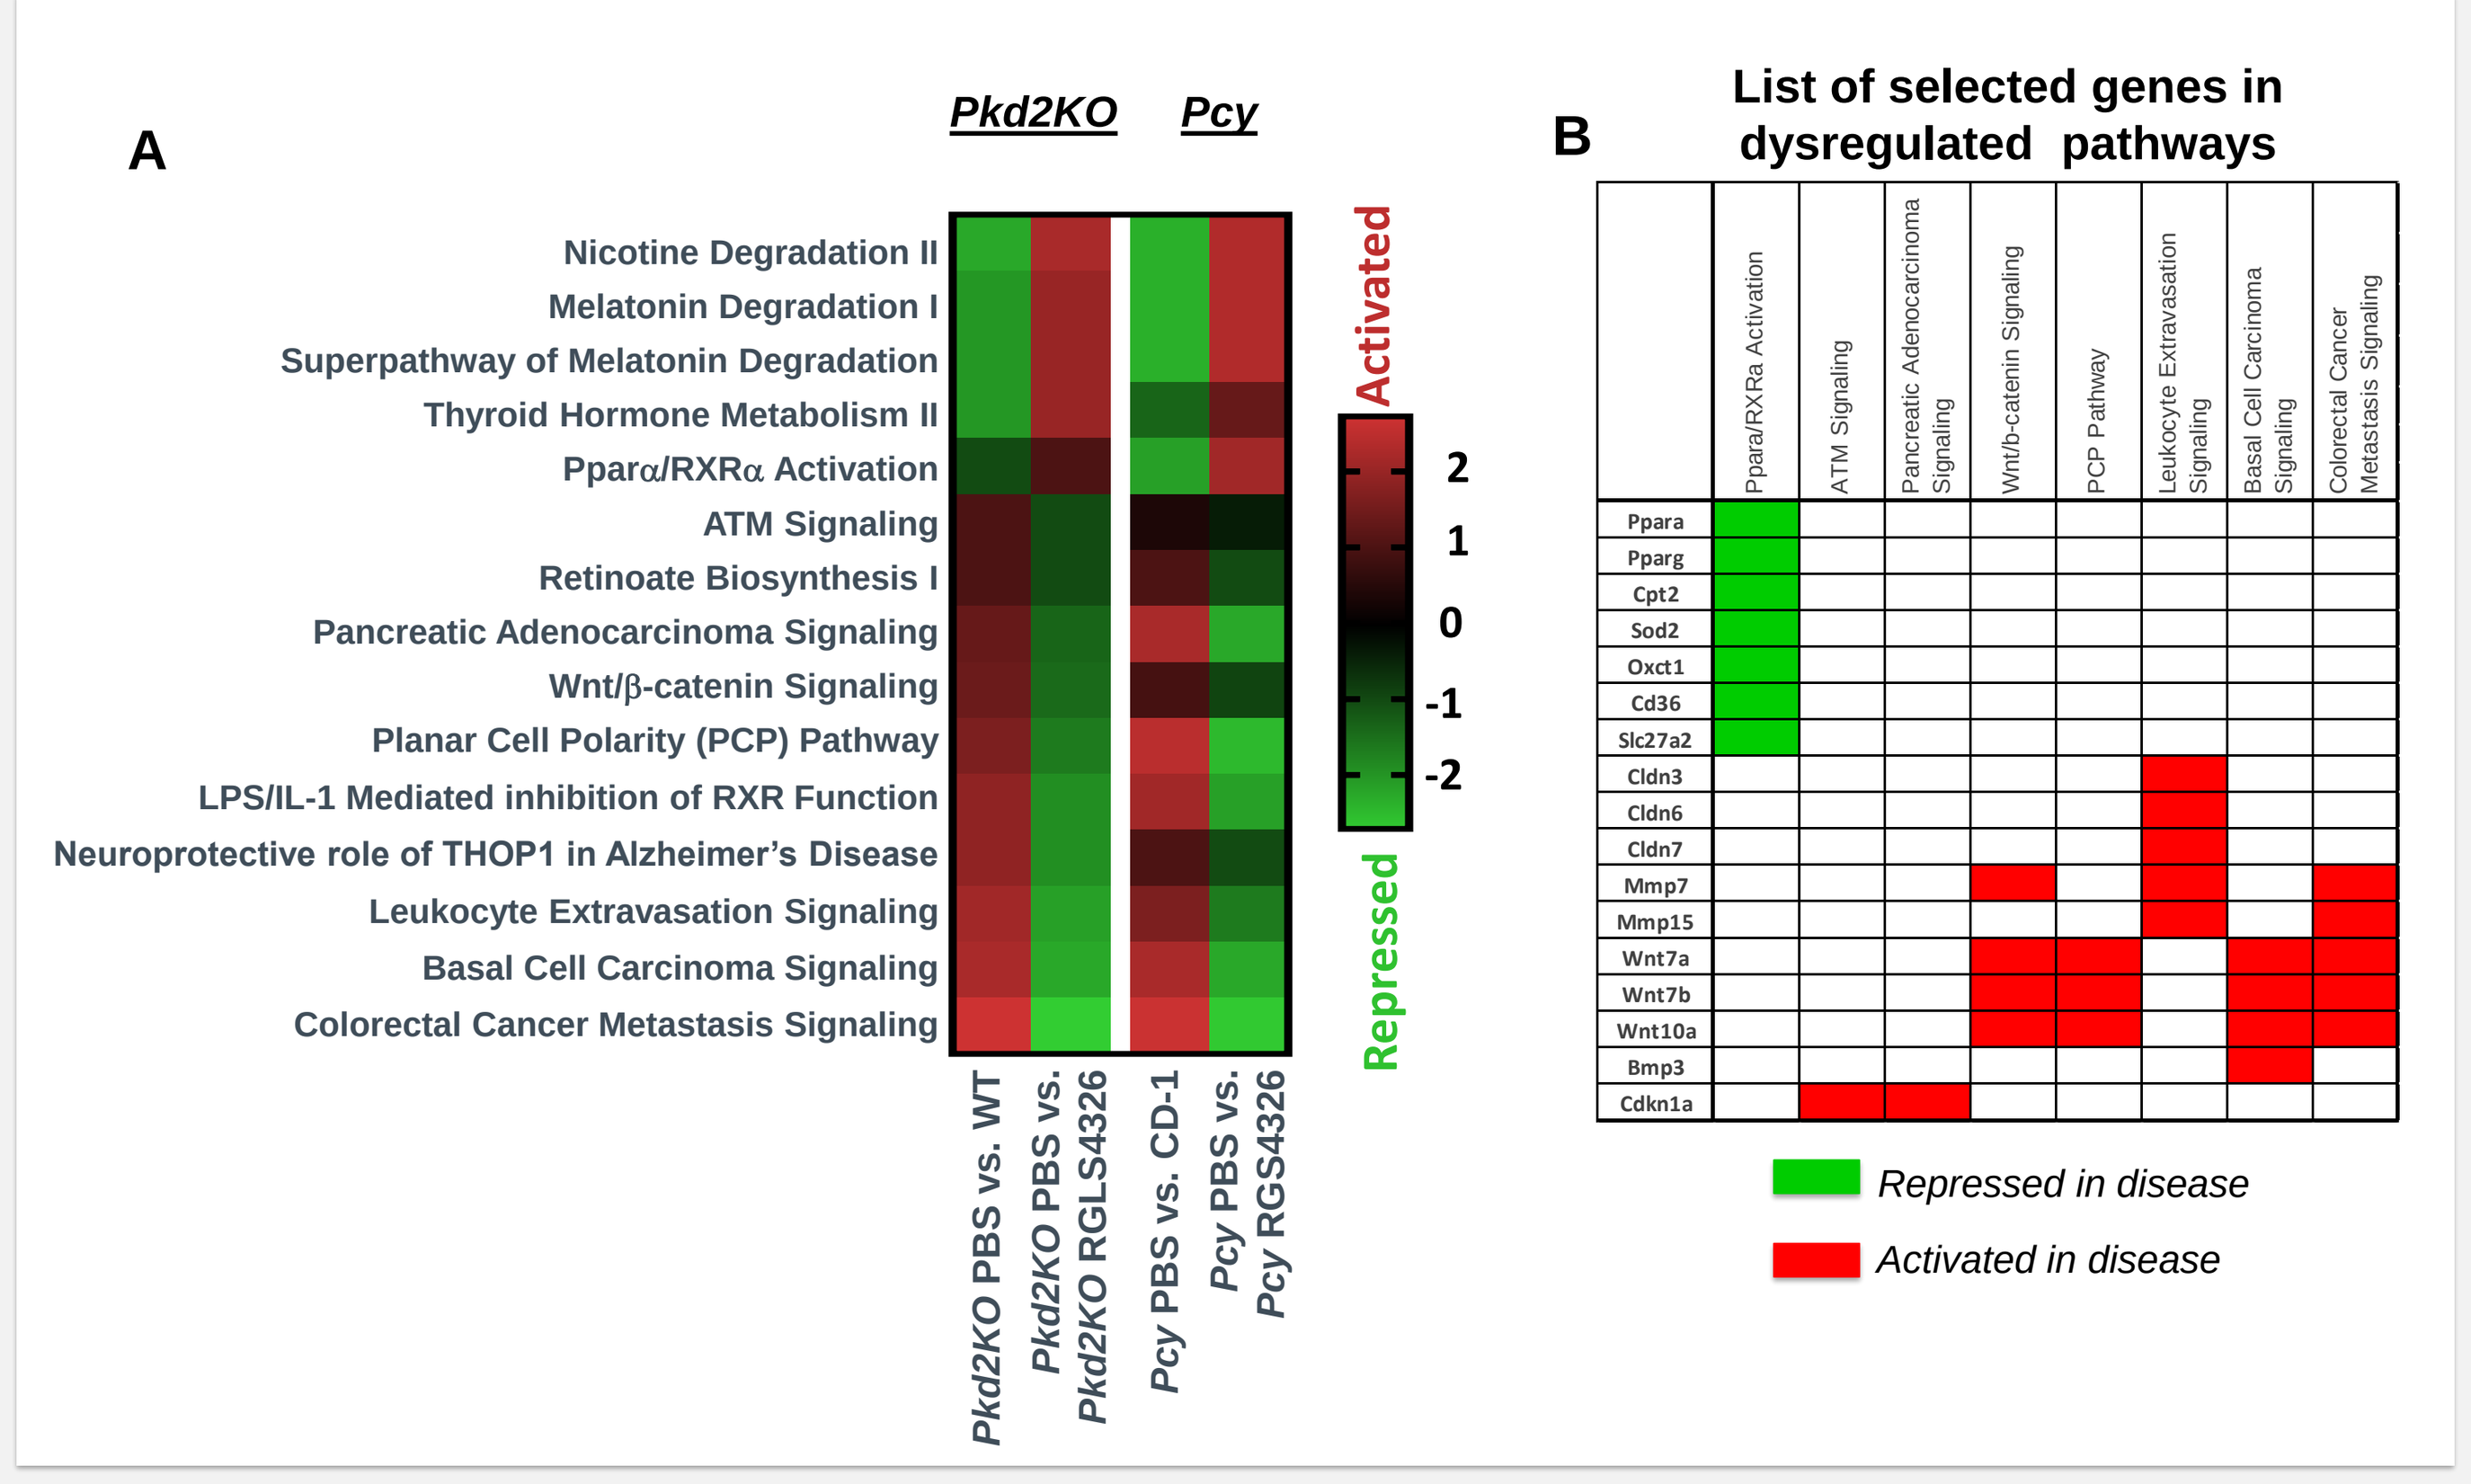
A
Pkd2KO
Pcy
Nicotine Degradation II
Melatonin Degradation I
Superpathway of Melatonin Degradation
Thyroid Hormone Metabolism II
Pparα/RXRα Activation
ATM Signaling
Retinoate Biosynthesis I
Pancreatic Adenocarcinoma Signaling
Wnt/β-catenin Signaling
Planar Cell Polarity (PCP) Pathway
LPS/IL-1 Mediated inhibition of RXR Function
Neuroprotective role of THOP1 in Alzheimer's Disease
Leukocyte Extravasation Signaling
Basal Cell Carcinoma Signaling
Colorectal Cancer Metastasis Signaling
Pkd2KO PBS vs. WT
Pkd2KO PBS vs. *Pkd2KO* RGLS4326
Pcy PBS vs. CD-1
Pcy PBS vs. *Pcy* RGS4326
Activated
Repressed
2
1
0
-1
-2
B
List of selected genes in dysregulated pathways
Ppara/RXRa Activation
ATM Signaling
Pancreatic Adenocarcinoma Signaling
Wnt/b-catenin Signaling
PCP Pathway
Leukocyte Extravasation Signaling
Basal Cell Carcinoma Signaling
Colorectal Cancer Metastasis Signaling
Ppara
Pparg
Cpt2
Sod2
Oxct1
Cd36
Slc27a2
Cldn3
Cldn6
Cldn7
Mmp7
Mmp15
Wnt7a
Wnt7b
Wnt10a
Bmp3
Cdkn1a
Repressed in disease
Activated in disease

Ingenuity pathway analysis of the DE genes from *Pkd2KO* and *Pcy*/CD-1 efficacy models
(A) Comparative pathway analysis revealed that dysregulated pathways in the disease models (when compared to age matched WT controls) are restored upon RGLS4326 treatment in both models
(B) Genes from selected dysregulated pathways identified in IPA comparative analysis were assessed by qPCR in both models

Kidney expression of selected genes following RLGS4326 treatment in *Pkd2KO* and *Pcy* efficacy models

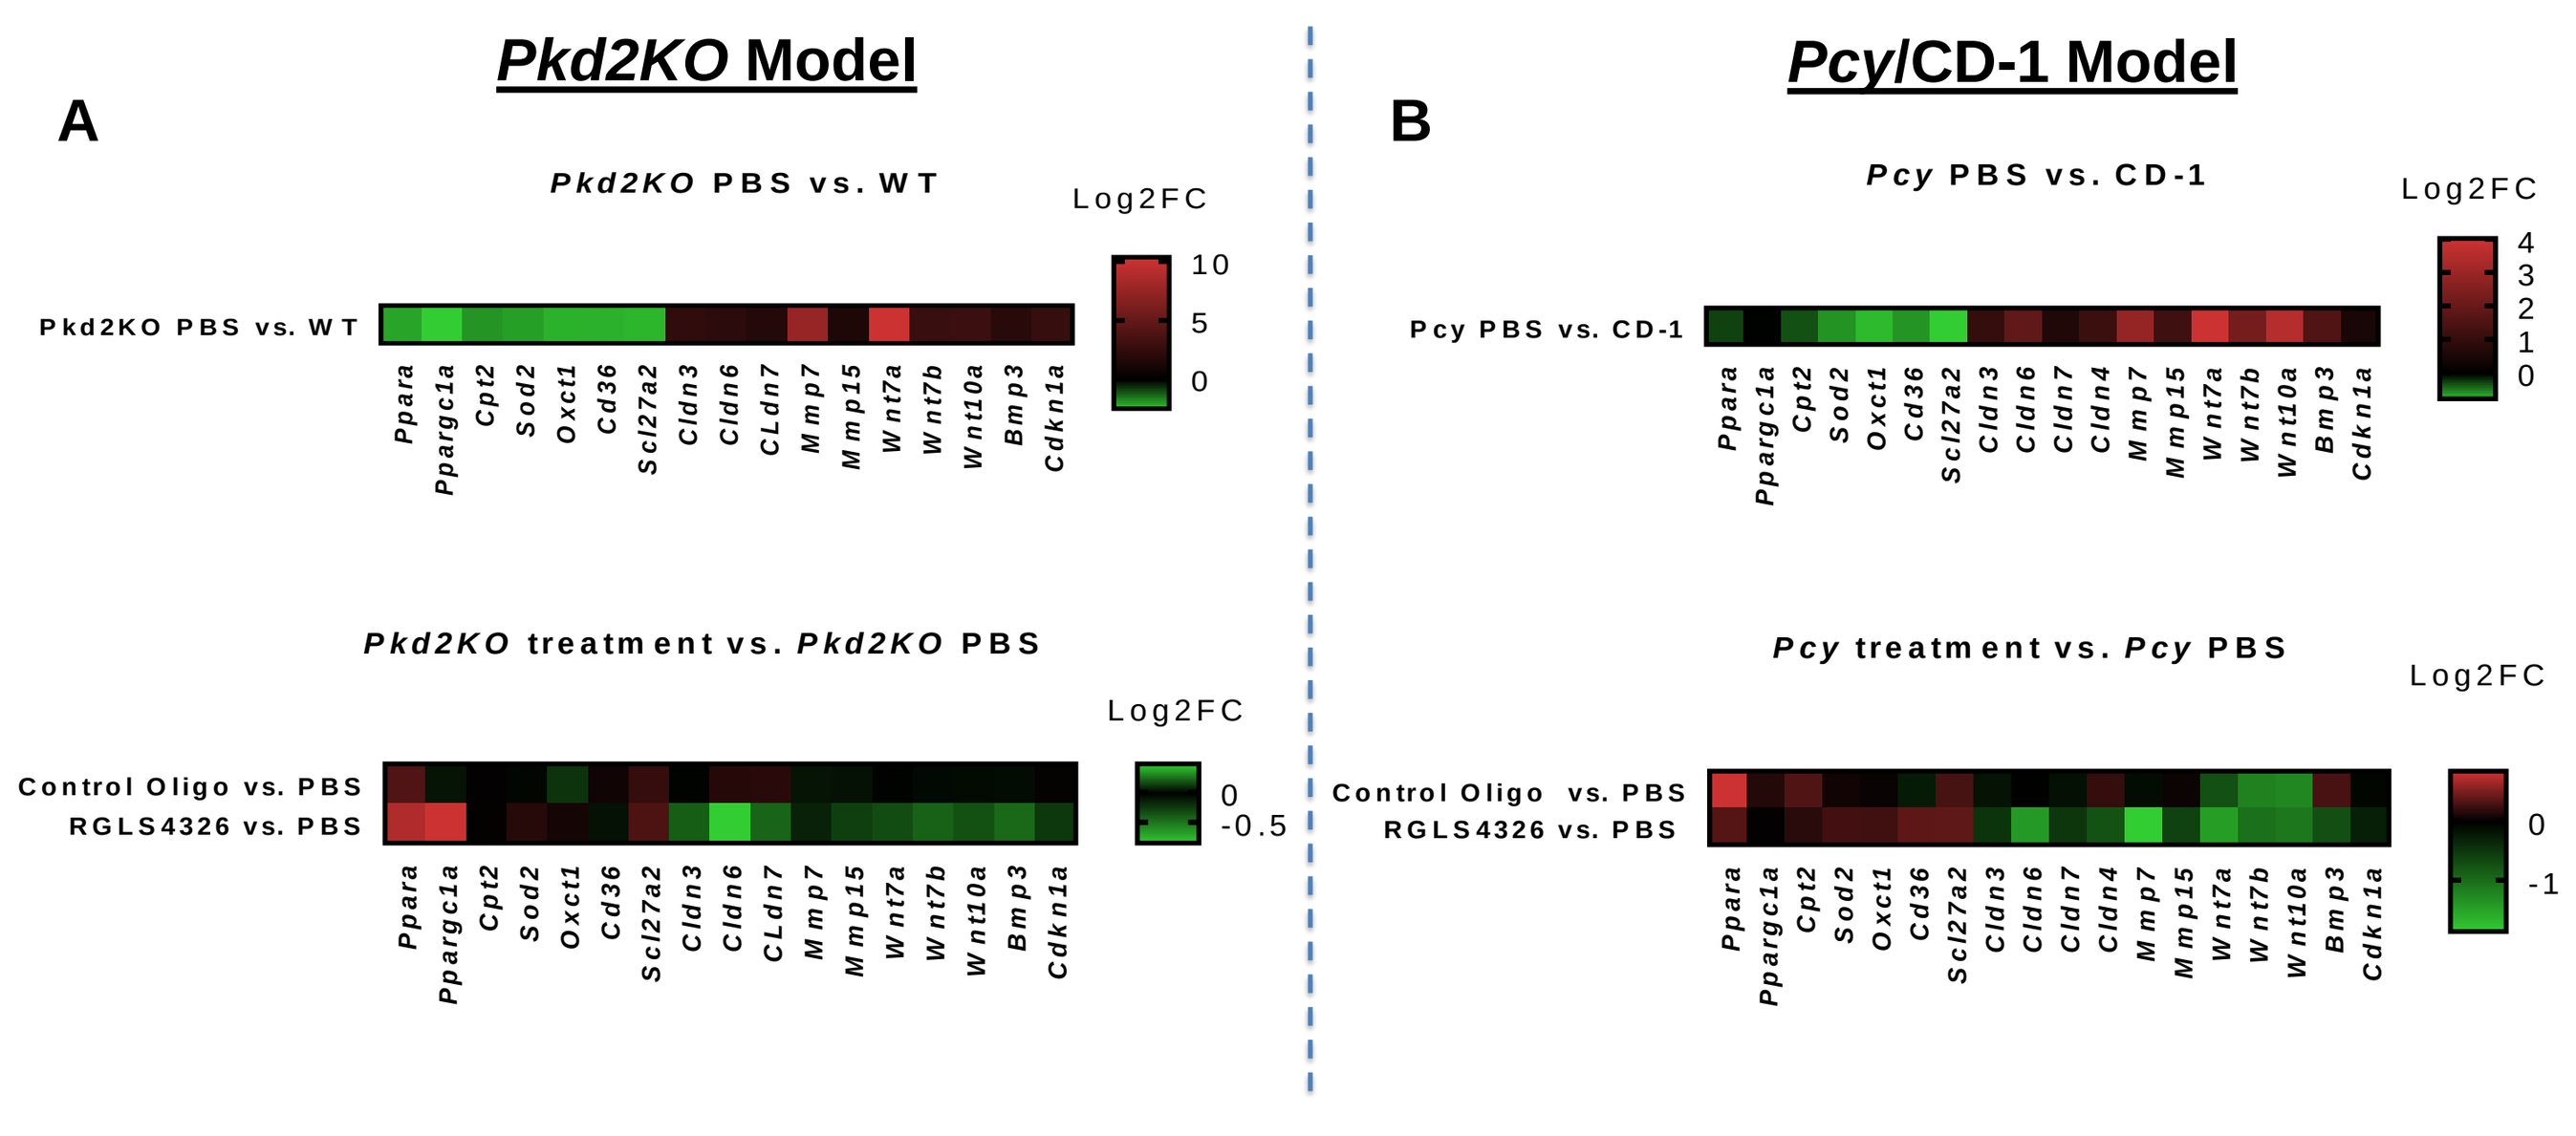
Pkd2KO Model
A
Pkd2KO PBS vs. WT
Log2FC
10
5
0
Pkd2KO treatment vs. *Pkd2KO* PBS
Control Oligo vs. PBS
RGLS4326 vs. PBS
0
-0.5
Pcy/CD-1 Model
B
Pcy PBS vs. CD-1
4
3
2
1
0
Pcy treatment vs. *Pcy* PBS
-1
Ppara
Ppargc1a
Cpt2
Sod2
Oxct1
Cd36
Scl27a2
Cldn3
Cldn6
CLdn7
Cldn4
Mmp7
Mmp15
Wnt7a
Wnt7b
Wnt10a
Bmp3
Cdkn1a

- Selected genes identified key PKD-disease related pathways in *Pkd2KO* **(A)** and *Pcy*/CD-1 **(B)** and their modulation after RGLS4326 treatment
- Upregulation of Pparα upon RGLS4326 treatment in the *Pkd2KO* model corroborates with previous findings where genetic deletion of miR17-92 cluster in the *Pkd2KO* kidney restores the de-repressed Pparα pathway (Hajarnis *et al*. 2017)
- Overexpression of claudins in ADPKD patients compared to normal tissue has been reported previously (Almeida *et al*. 2016). Consistently we detected elevated levels of claudins in disease kidney and downregulation following RGLS4326 treatment in both efficacy models
- Aberrant regulation of PCP and Wnt signaling in Pkd1 Knockout cystic kidneys has been already described (Luyten *et al*. 2010; Quin *et al*. 2012). In agreement with this, we also observed the overexpression of Wnt7a and Wnt7b in *Pkd2KO* and *Pcy* mouse models, where their levels were significantly downregulated after RGLS4326 treatment

Kidney global metabolite profiling

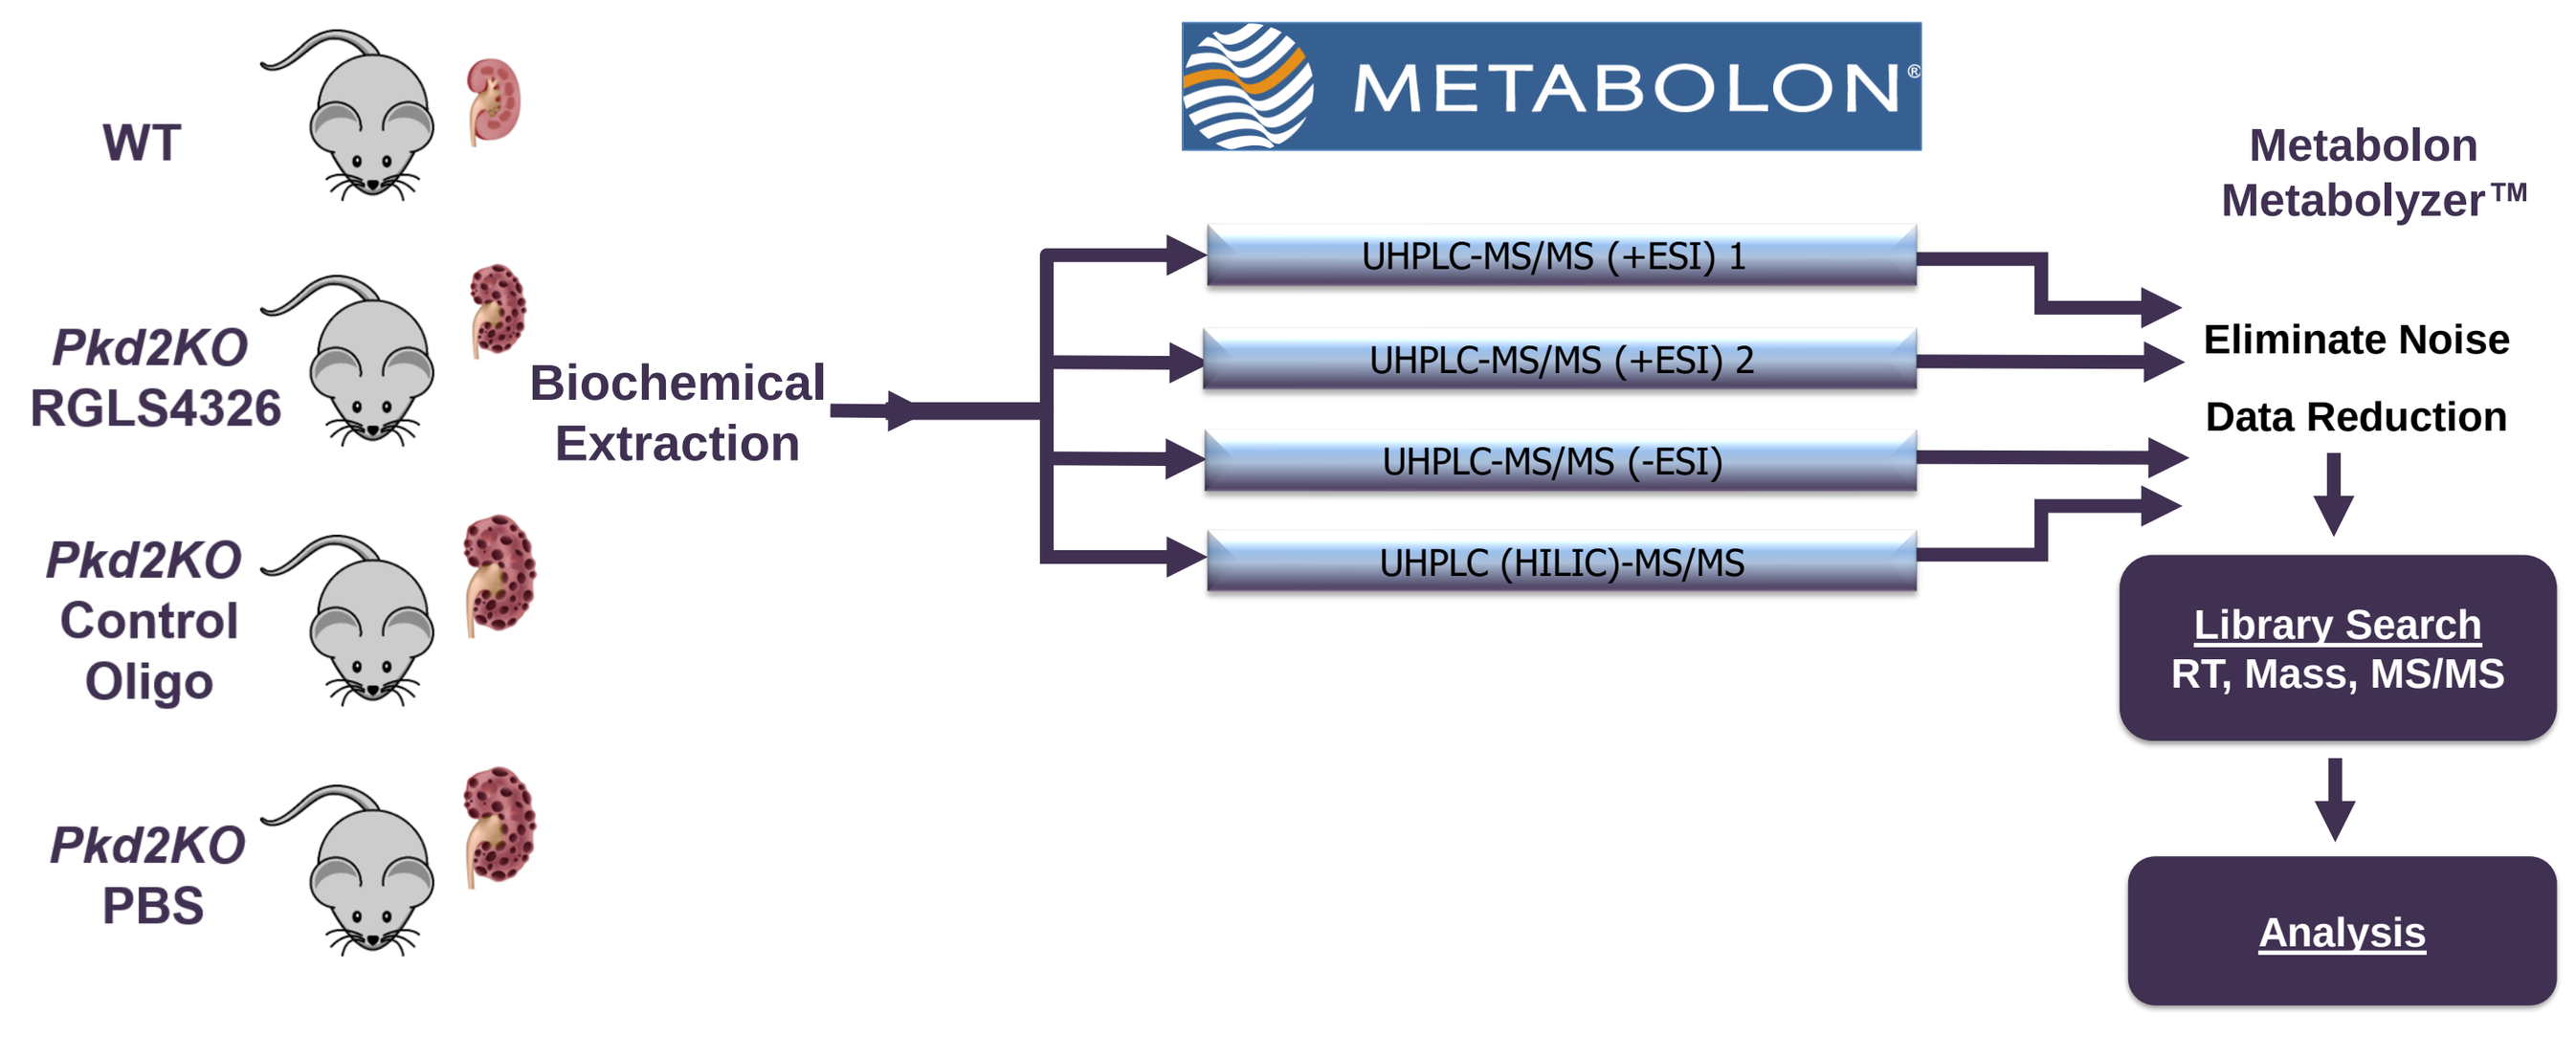
WT
Pkd2KO RGLS4326
Pkd2KO Control Oligo
Pkd2KO PBS
Biochemical Extraction
METABOLON
UHPLC-MS/MS (+ESI) 1
UHPLC-MS/MS (+ESI) 2
UHPLC-MS/MS (-ESI)
UHPLC (HILIC)-MS/MS
Metabolon Metabolyzer™
Eliminate Noise
Data Reduction
Library Search
RT, Mass, MS/MS
Analysis

Global metabolic profile: *Pkd2KO* vs WT

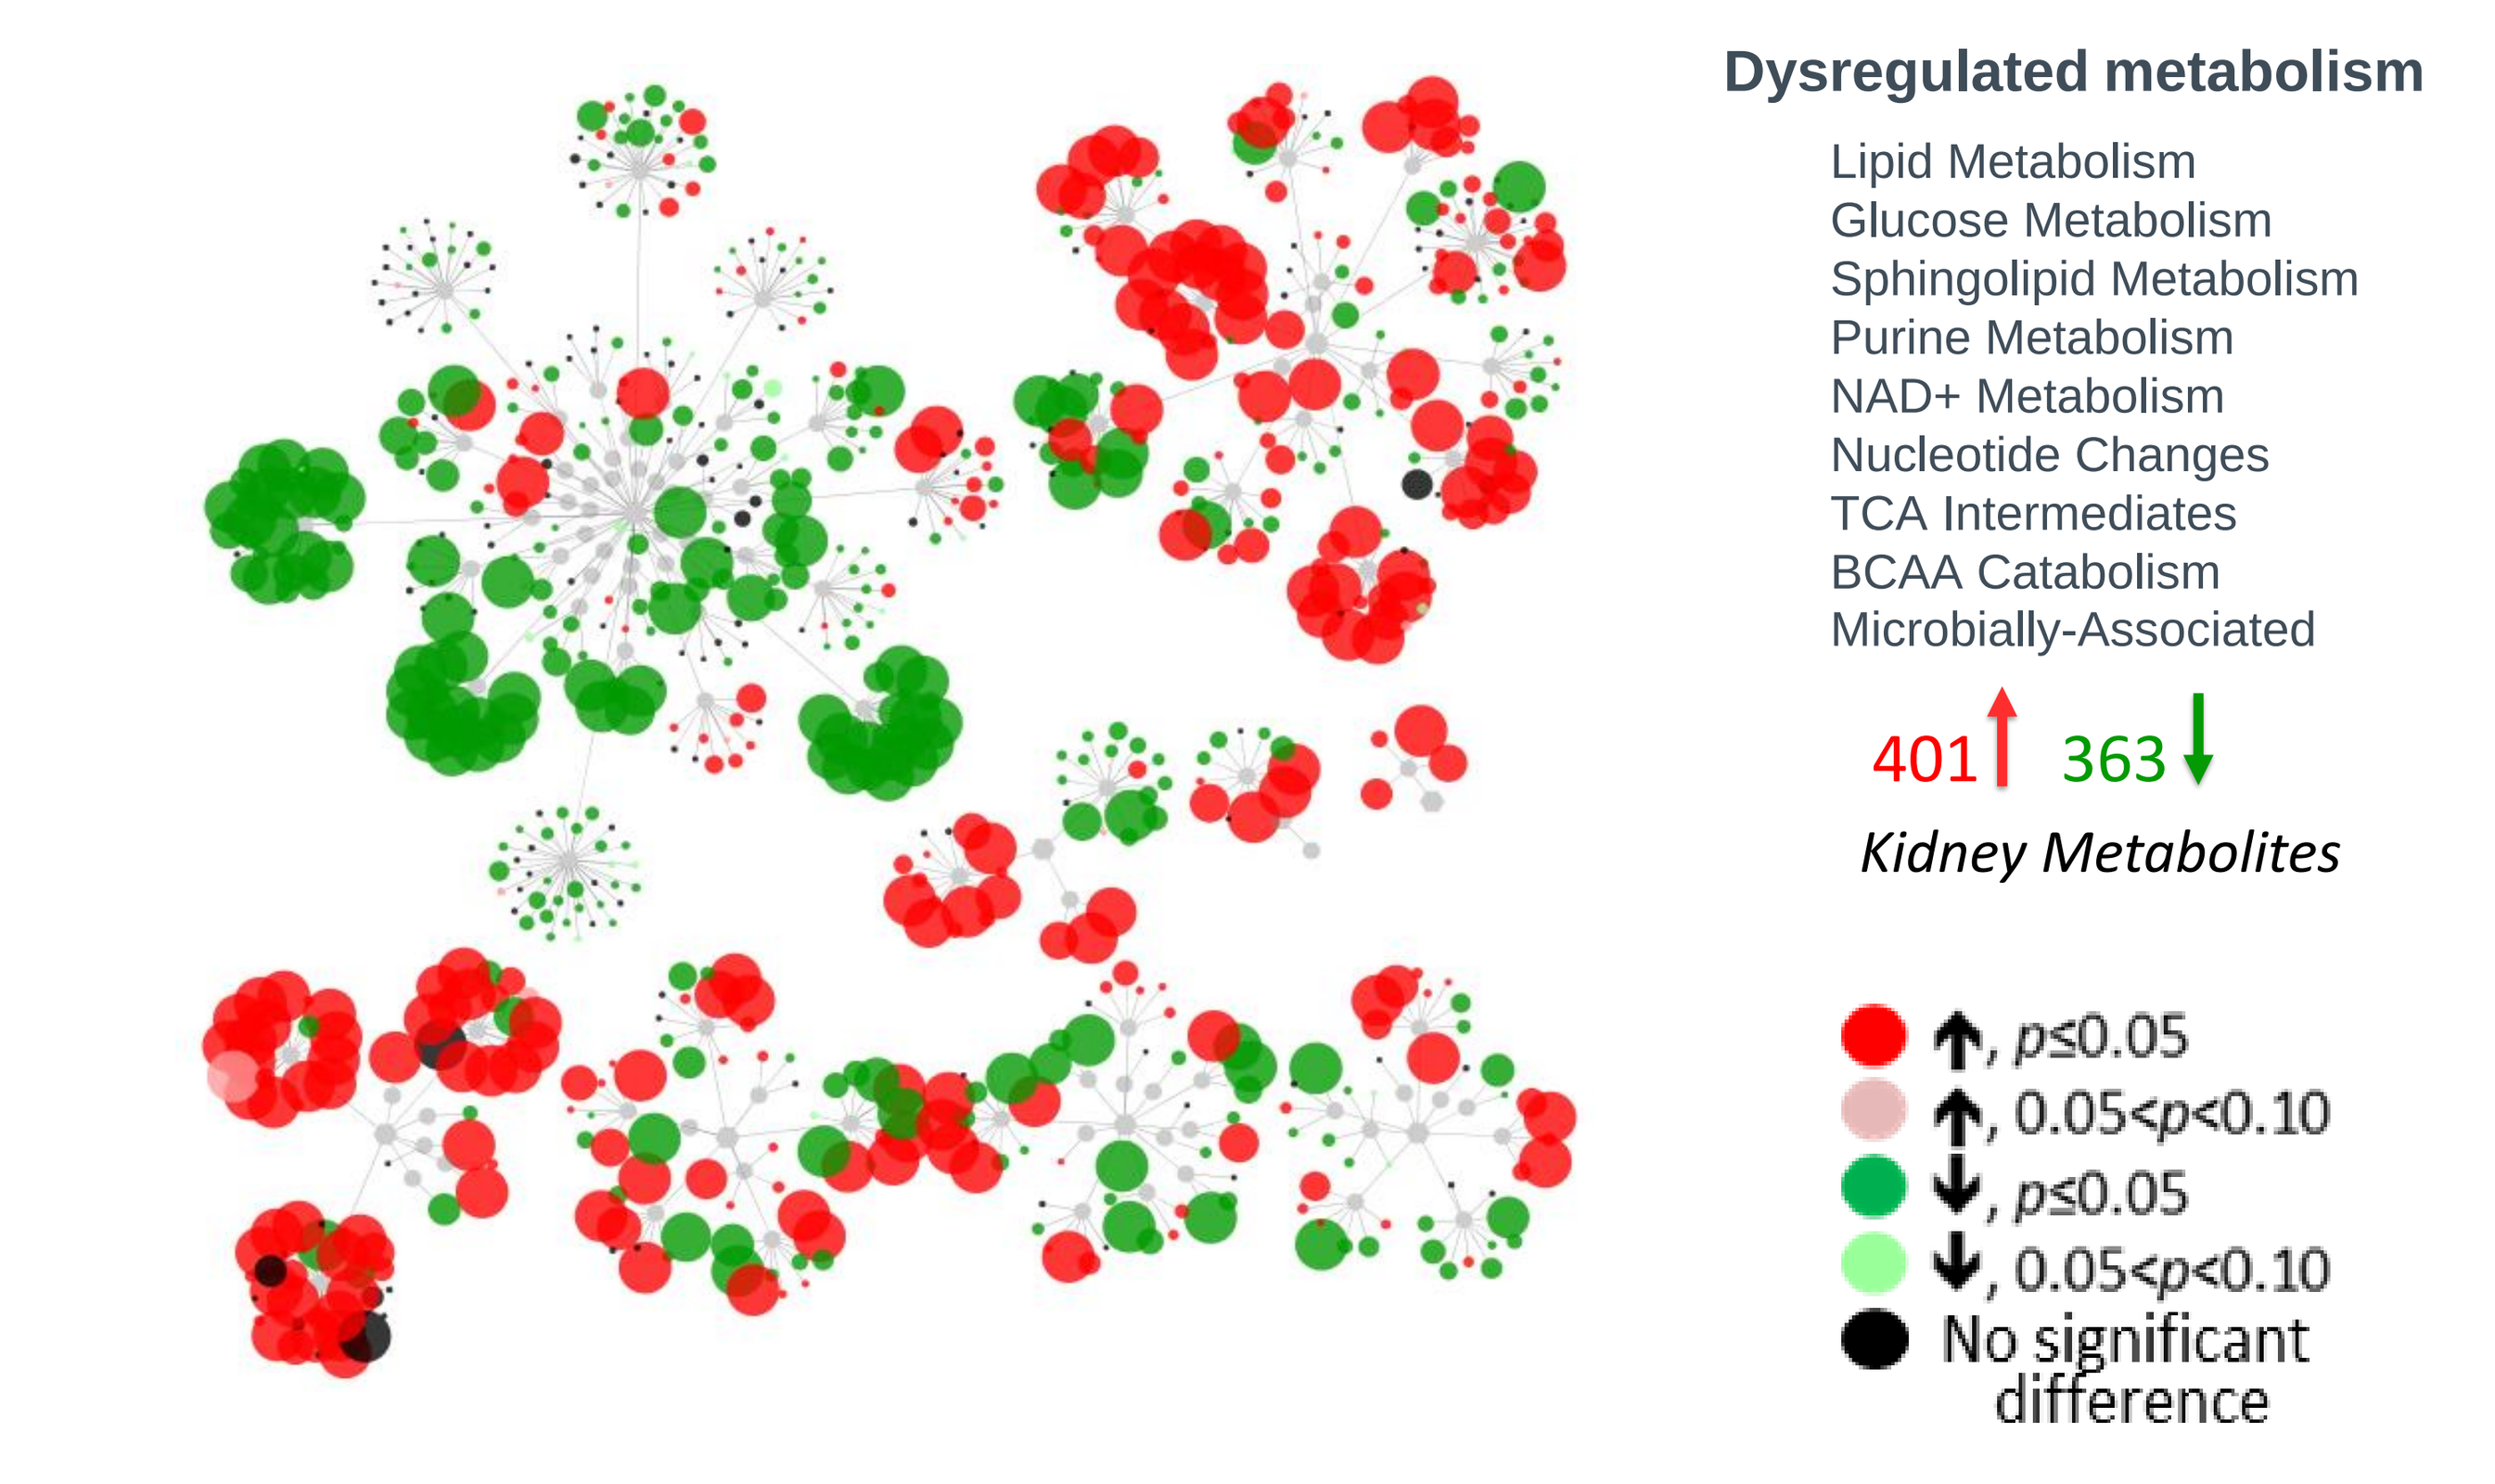
Dysregulated metabolism
Lipid Metabolism
Glucose Metabolism
Sphingolipid Metabolism
Purine Metabolism
NAD+ Metabolism
Nucleotide Changes
TCA Intermediates
BCAA Catabolism
Microbially-Associated
401 363
Kidney Metabolites
↑, p≤0.05
↑, 0.05<p<0.10
↓, p≤0.05
↓, 0.05<p<0.10
No significant difference

Metabolic profiles between disease *Pkd2KO* and healthy WT kidney displayed significant differences in metabolites (401 upregulated and 363 downregulated) across all biochemical nodes, indicating a broadly altered metabolism in the disease kidney such as glucose, lipid, sphingolipids, Purine, NAD+ and amino acids metabolism

Lipid metabolism in the kidney: *Pkd2KO* vs WT

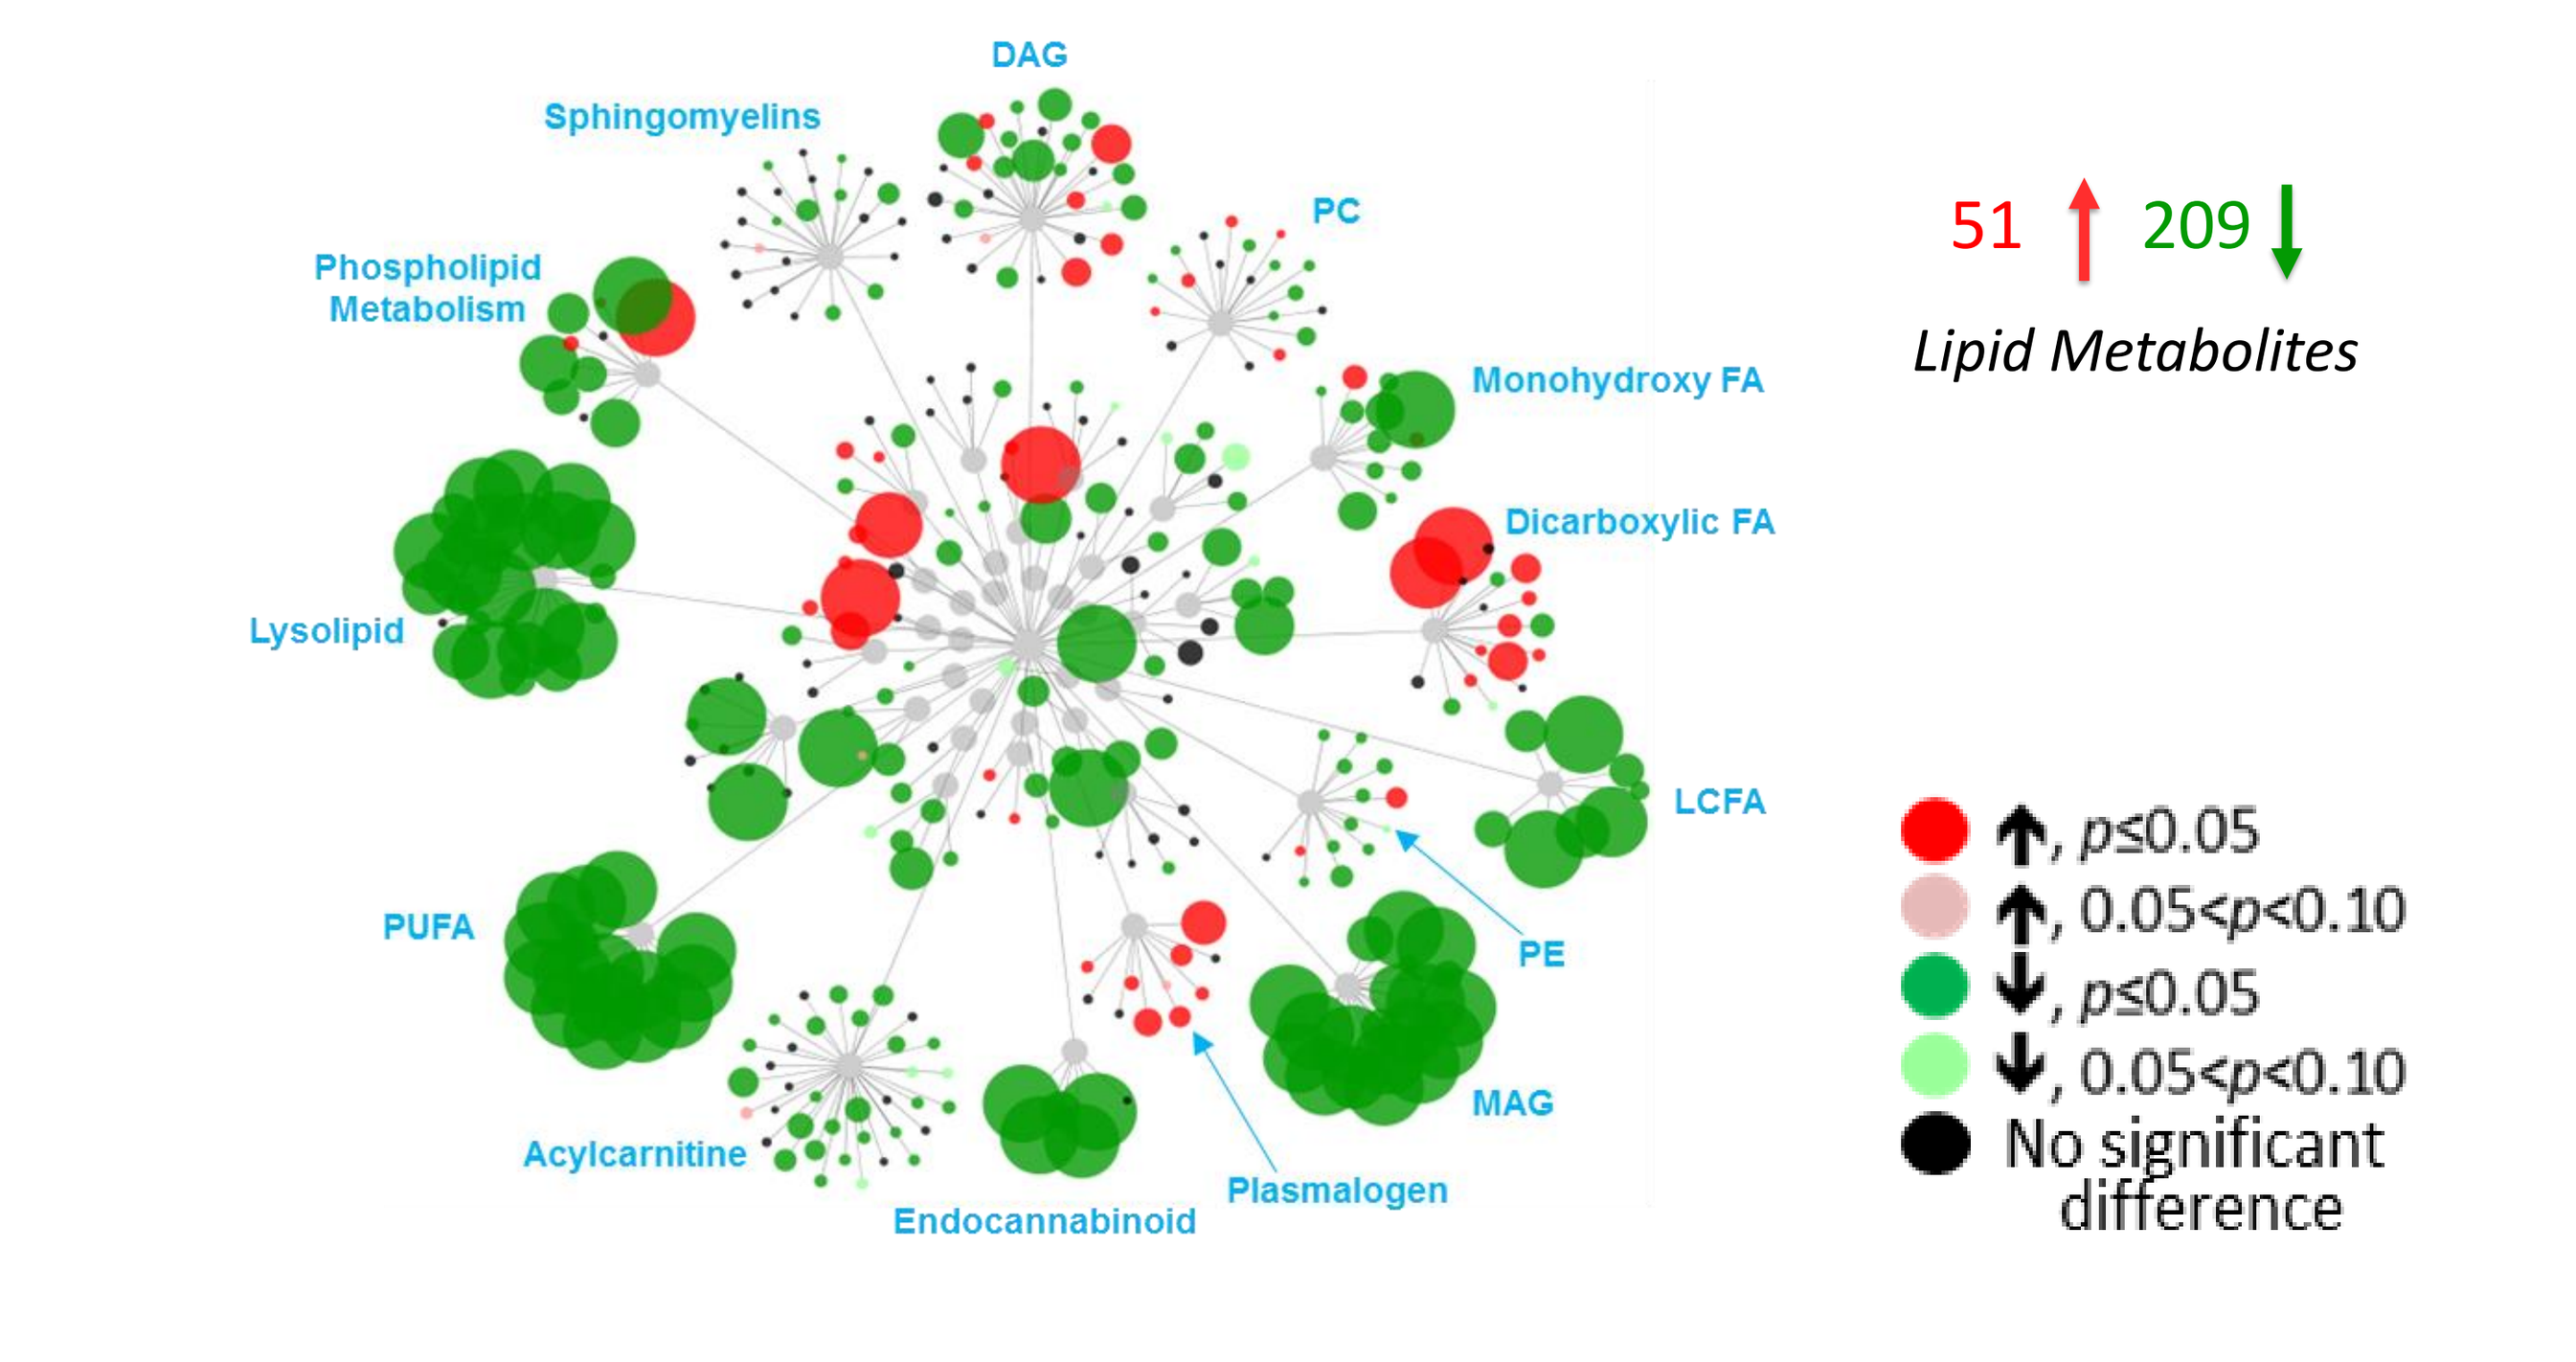
DAG
Sphingomyelins
PC
Phospholipid Metabolism
Monohydroxy FA
Dicarboxylic FA
Lysolipid
LCFA
PUFA
PE
MAG
Acylcarnitine
Plasmalogen
Endocannabinoid
51 209
Lipid Metabolites
↑, p≤0.05
↑, 0.05<p<0.10
↓, p≤0.05
↓, 0.05<p<0.10
No significant difference

Altered lipid metabolism noted in *Pkd2KO* kidney reflected the differential uptake, synthesis or utilization of lipids. Several lipid classes were significantly decreased in *Pkd2KO* vs WT including monoacylglyerols (MAG), long chain fatty acids (LCFA), polyunsaturated fatty acids (PUFA), endocannabinoids and lysolipids. Preliminary data demonstrated that treatment with RGLS4326 modulated some dysregulated metabolites.

References

- Almeida RMC., Clendenon SG., Richards WG., Boedigheimer M., Damore M., Rossetti S., Harris PC., Herbert B., Xu WM., Wandinger-Ness A., Wrd HH., Glazier JA., Bacallao RL., 2016. Transcriptome analysis reveals manifold mechanisms of cyst development in ADPKD. Hum Genomics, 10: 37.
- Hajarnis S., Lakhia R., Yheskel M., Williams D., Sorourian M., Lui X., Aboudehen K., Zhang S., Kersjes K., Galasso R., Li J., Kimal V., Lockton S., Davis S., Flaten A., Johnson JA., Holland WL., Kusminski CM., Scherer PE., Harris PC., Trudel M., Wallace DP., Igarashi P., Lee EC., Androsavich JR., Patel V., 2017. microRNA-17 family promotes polycystic kidney disease progression through modulation of mitochondrial metabolism. Nature comm. 8: 14395.
- Luyten A., Su X., Gondela S., Chen Y., Rompani S., Takakura A., Zhou J., 2010. Aberrant regulation of Planar cell polarity in polycystic kidney disease. J am Soc Nephrol, 21(9): 1521-1532.
- Quin S, Taglienti M., Cai L., Zhou J., Kreidberg JA., 2012. c-Met and NF-κB–Dependent overexpression of Wnt7a and -7b and Pax2 promotes cystogenesis in polycystic kidney disease. J Am Soc Nephrol, 23(8): 1309-1318.

American Society of Nephrology - Kidney Week, San Diego, CA (Oct 23 - 28, 2018)

Abstract Number: 3021851; Poster Board Number: TH-PO687