Efficacy of RGLS4326 in Human Primary 3D-Cyst Cultures derived from Autosomal Dominant Polycystic Kidney Disease (ADPKD) Donors

Annelie Schairer[1], Tania Valencia[1], Steve Lockton[1], Sole Gatto[1], Michael Kim[1], Darren Wallace[2], Edmund Lee[1,*]
[1]Regulus Therapeutics Inc., San Diego, California. *Presenting Author
[2]The Jared Grantham Kidney Institute, University of Kansas medical Center, Kansas City, Kansas.

Abstract

Background
Autosomal dominant polycystic kidney disease (ADPKD), caused by mutations in the *PKD1* or *PKD2* gene (which encodes for the proteins polycystine-1 (PC1) and polycystine-2 (PC2)), is among the most common monogenetic disorders and a leading genetic cause of end-stage renal disease. Kidney-specific overexpression of miR-17~92 produce kidney cysts in mice, whereas genetic knockdown of miR-17~92 attenuates disease progression in multiple mouse models of PKD. RGLS4326 is designed to specifically bind to miR-17 family of microRNAs, antagonize miR-17 activity and reduce disease progression in mouse models of PKD. In this study, we investigate the effect of RGLS4326 treatment on 3D growth of human primary ADPKD cyst cells derived from ADPKD donor samples.

Methods:
Primary human ADPKD cyst cells (HuADPKD) were transfected with RGLS4326 or control oligo at 20nM, 100nM or 300nM for 24h. RNA samples were harvested for confirmation of miR-17 inhibition by measuring de-repression of a selected set of direct miR-17 target genes (PD-Sig) and RNA sequencing. Following 24h transfection, cells were seeded and further cultured in a 3D cyst formation assay for 8 additional days.

Results:
Kolmogorov-Smironov test statistics on RNA sequencing data comparing log2FC cumulative distribution indicated significant upregulation (i.e. de-repression) of predicted miR-17 target genes after 24h of RGL4326 treatment by transfection. Functional inhibition of miR-17 in HuADPKD cells following RGLS4326 treatment was also confirmed by miR-17 PD-Sig. At the end of 8 additional days of 3D culturing, RGLS4326 consistently reduced growth of HuADPKD cells derived from multiple donors, decreasing cyst count and proliferation.

Conclusion:
RGLS4326 inhibits 3D cyst growth and proliferation of HuADPKD cells *in vitro* compared to oligo control. Our preclinical data supports the clinical development of RGLS4326 for the treatment of ADPKD.

Characteristics of Autosomal Dominant Polycystic Kidney Disease (ADPKD)

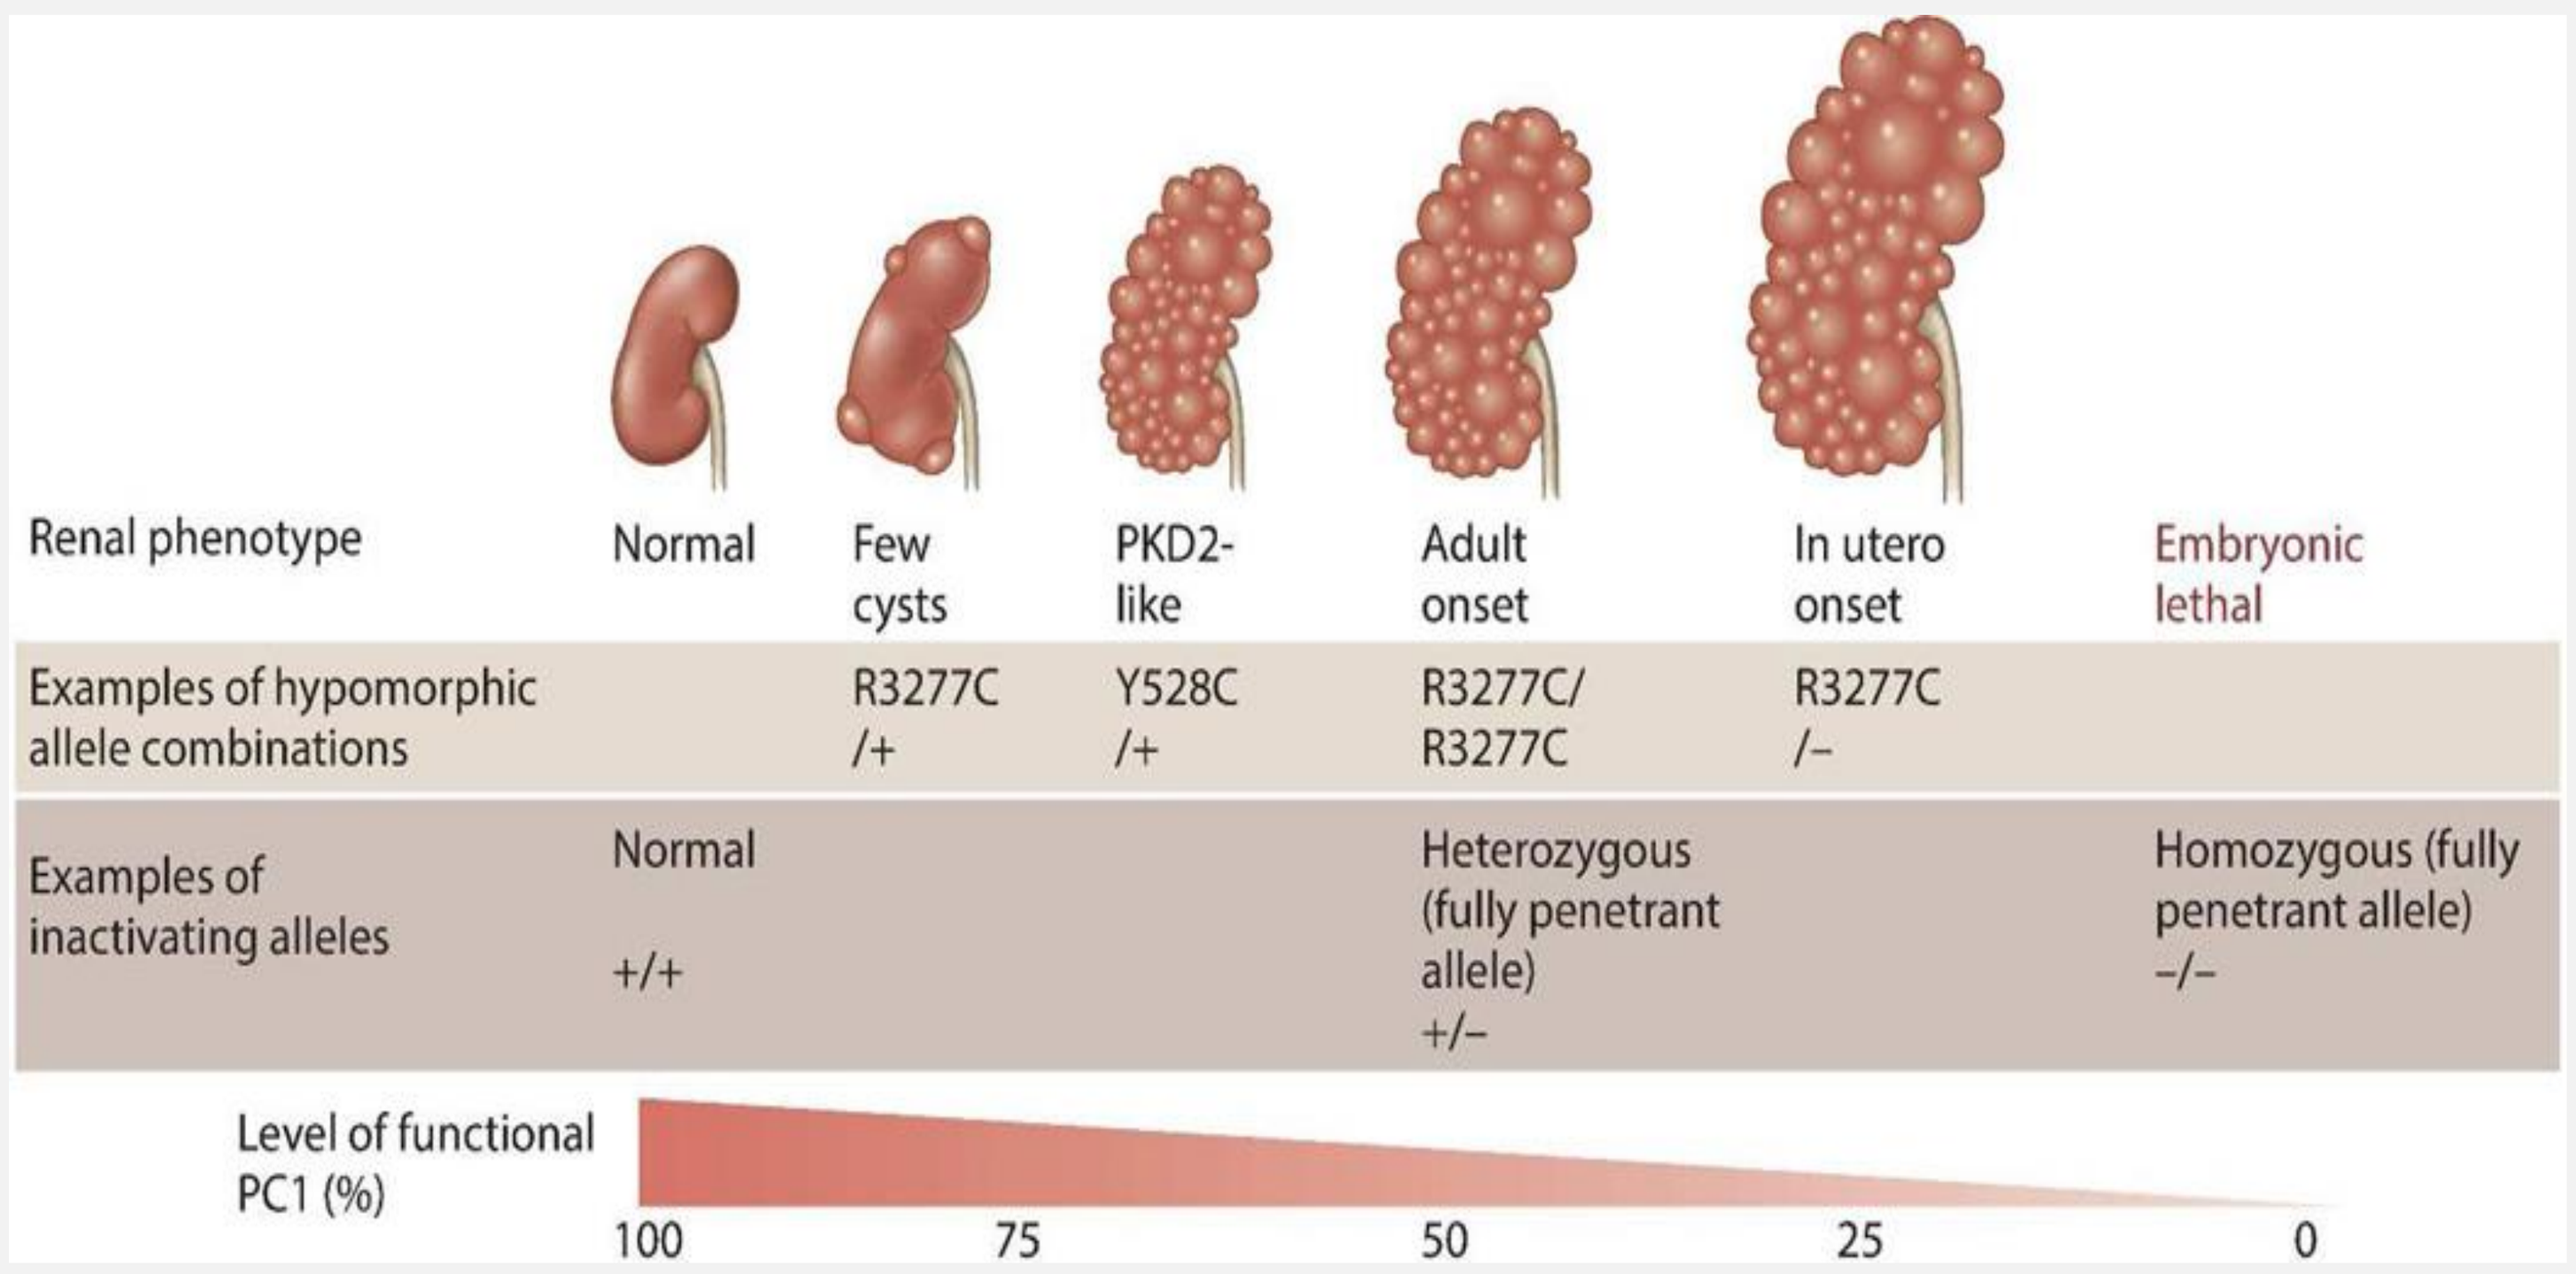
Renal phenotype
Normal
Few cysts
PKD2-like
Adult onset
In utero onset
Embryonic lethal
Examples of hypomorphic allele combinations
R3277C /+
Y528C /+
R3277C/ R3277C
R3277C /–
Examples of inactivating alleles
Normal +/+
Heterozygous (fully penetrant allele) +/–
Homozygous (fully penetrant allele) –/–
Level of functional PC1 (%)
100
75
50
25
0

Graphics adapted from: Ong et al, Kidney Int 2015

Mutations disrupt normal functions of PC1 and PC2 in renal tubular epithelium, cause growth of multiple kidney cysts that displace and destroy normal kidney tissues, and ultimately lead to fibrosis, derangement in renal architecture and kidney failure.

What are microRNAs?

- MicroRNAs are highly conserved, short non-coding RNAs (20-22 nts) with unique seed sequence of ~8 nts that bind to complementary target sequences located primary in the 3' untranslated region of targeted mRNAs.
- **Aberrant microRNAs activity, such as miR-17, has been shown to be important in multiple human diseases, including ADPKD.**
- Anti-miRs inhibits microRNAs function and de-represses their downstream target mRNAs and encoded proteins.

Knockdown of miR-17 reduces renal cyst in multiple preclinical models of ADPKD

Preclinical Model	Disease Progression	Intervention Modality	Reduction in Renal Cyst Growth
Genetic Models			
Pkd1-KO mice	Fast	x *miR-17~92*-KO	Y
Pkd1$^{F/RC}$ mice	Slow	x *miR-17~92*-KO	Y
Pkd1$^{RC/RC}$ mice	Slow	x *miR-17~92*-KO	Y
Kif3a-KO mice	Slow	x *miR-17~92*-KO	Y
Pkd2-KO mice	Fast	x *miR-17~92*-KO	Y

Patel et al. (2013) PNAS. Jun 25;110(26):10765; Hajarnis et al. (2017) Nat Commun. Feb 16;8:14395

miR-17 inhibition by RGLS4326 treatment has no affect on cell viability in non-ADPKD collecting duct cells

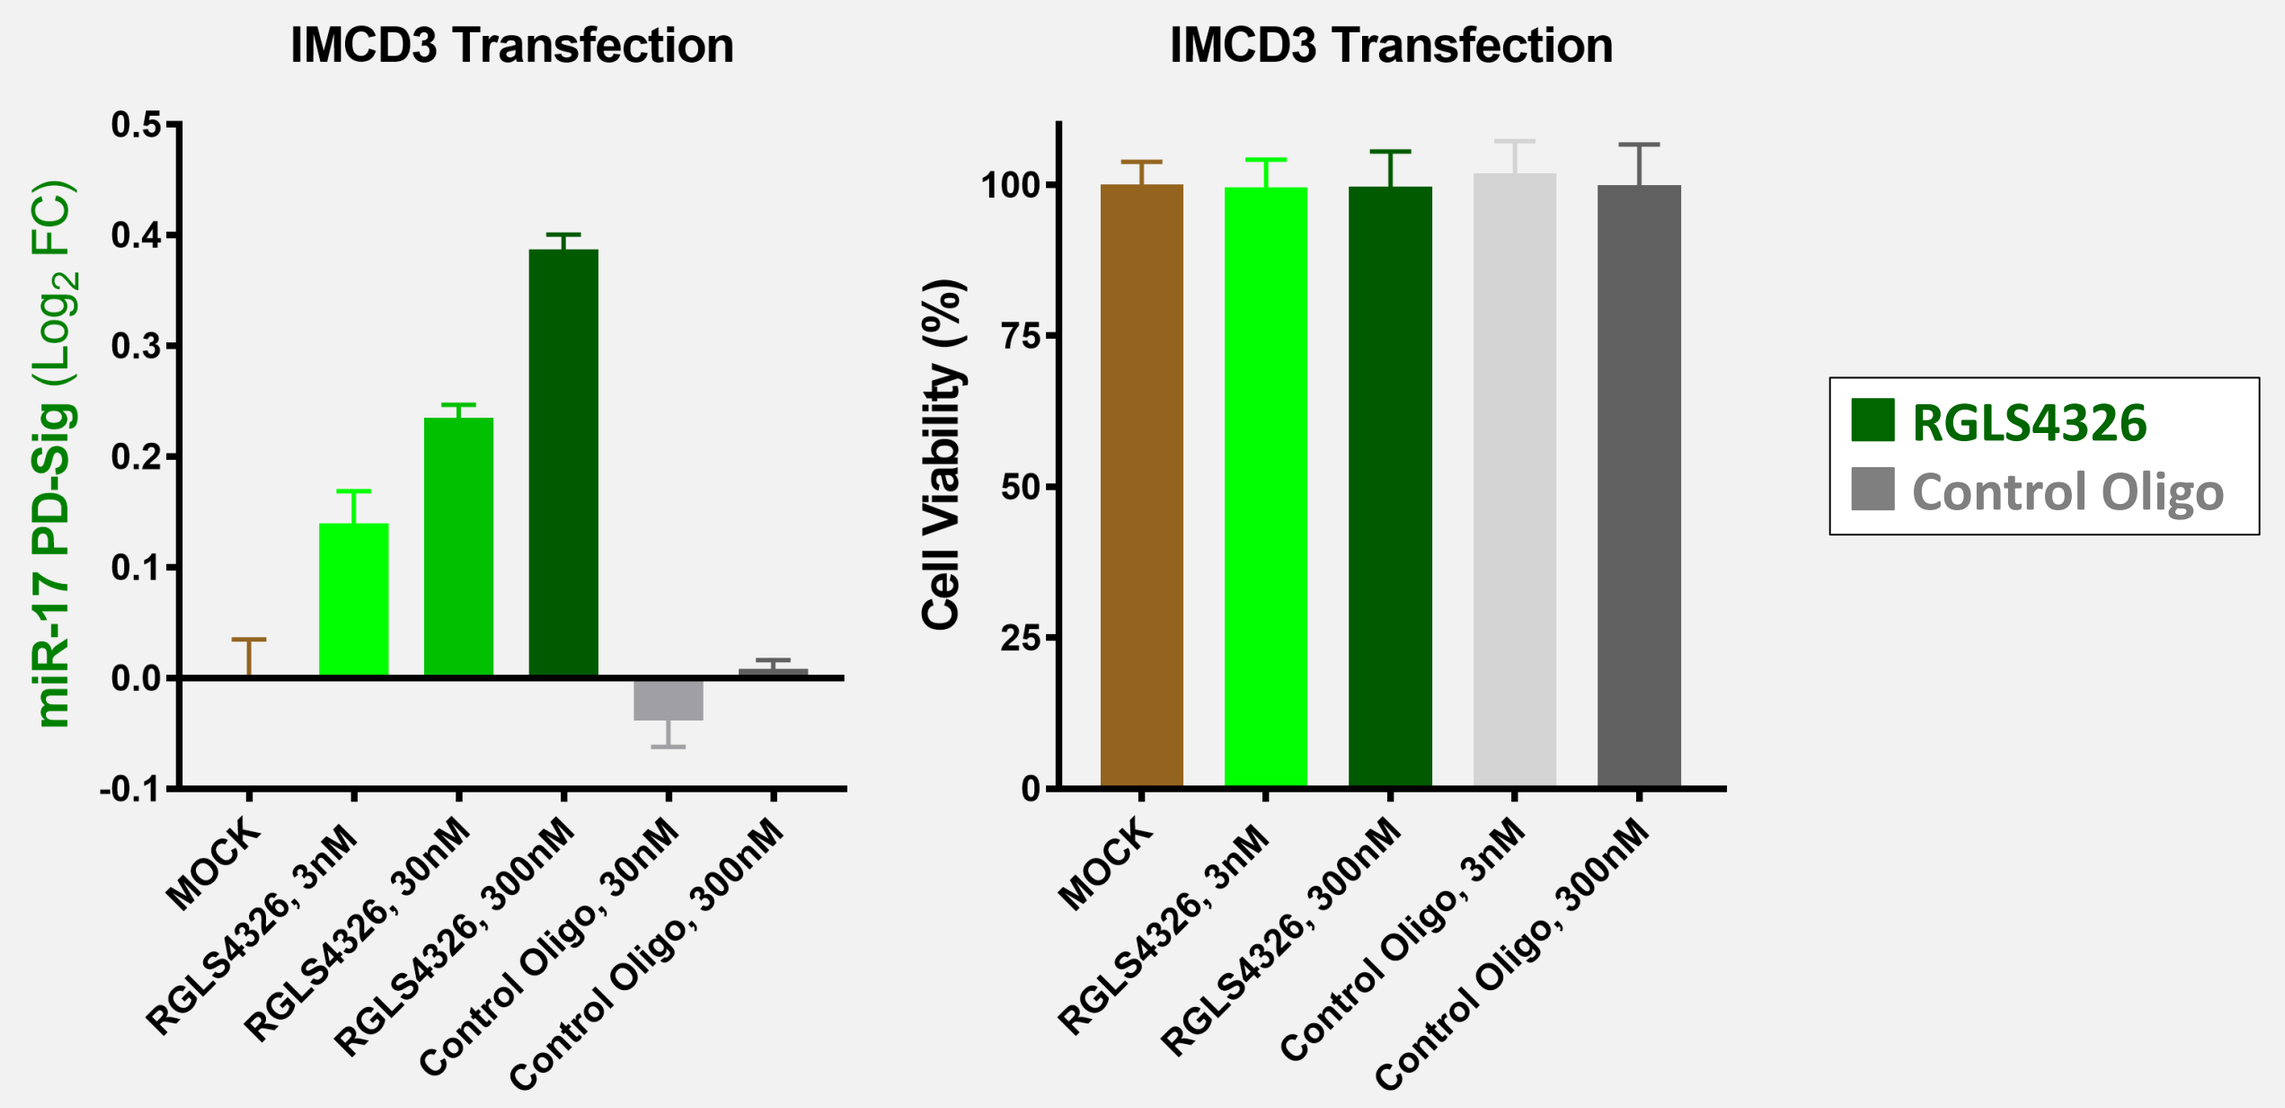
IMCD3 Transfection
miR-17 PD-Sig (Log2 FC)
MOCK
RGLS4326, 3nM
RGLS4326, 30nM
RGLS4326, 300nM
Control Oligo, 30nM
Control Oligo, 300nM
IMCD3 Transfection
Cell Viability (%)
MOCK
RGLS4326, 3nM
RGLS4326, 300nM
Control Oligo, 3nM
Control Oligo, 300nM
RGLS4326
Control Oligo

- Concentration-dependent de-repression of multiple miR-17 downstream target genes (as measured by mouse miR-17 PD-Sig) by RGLS4326 in mouse IMCD3 collecting duct cells 24h following transfection. No change in cell viability was observed 4 days following initial RGLS4326 treatment *in vitro*.

Primary human ADPKD (HuADPKD) cyst growth assay to measure anti-cyst activity of RGLS4326

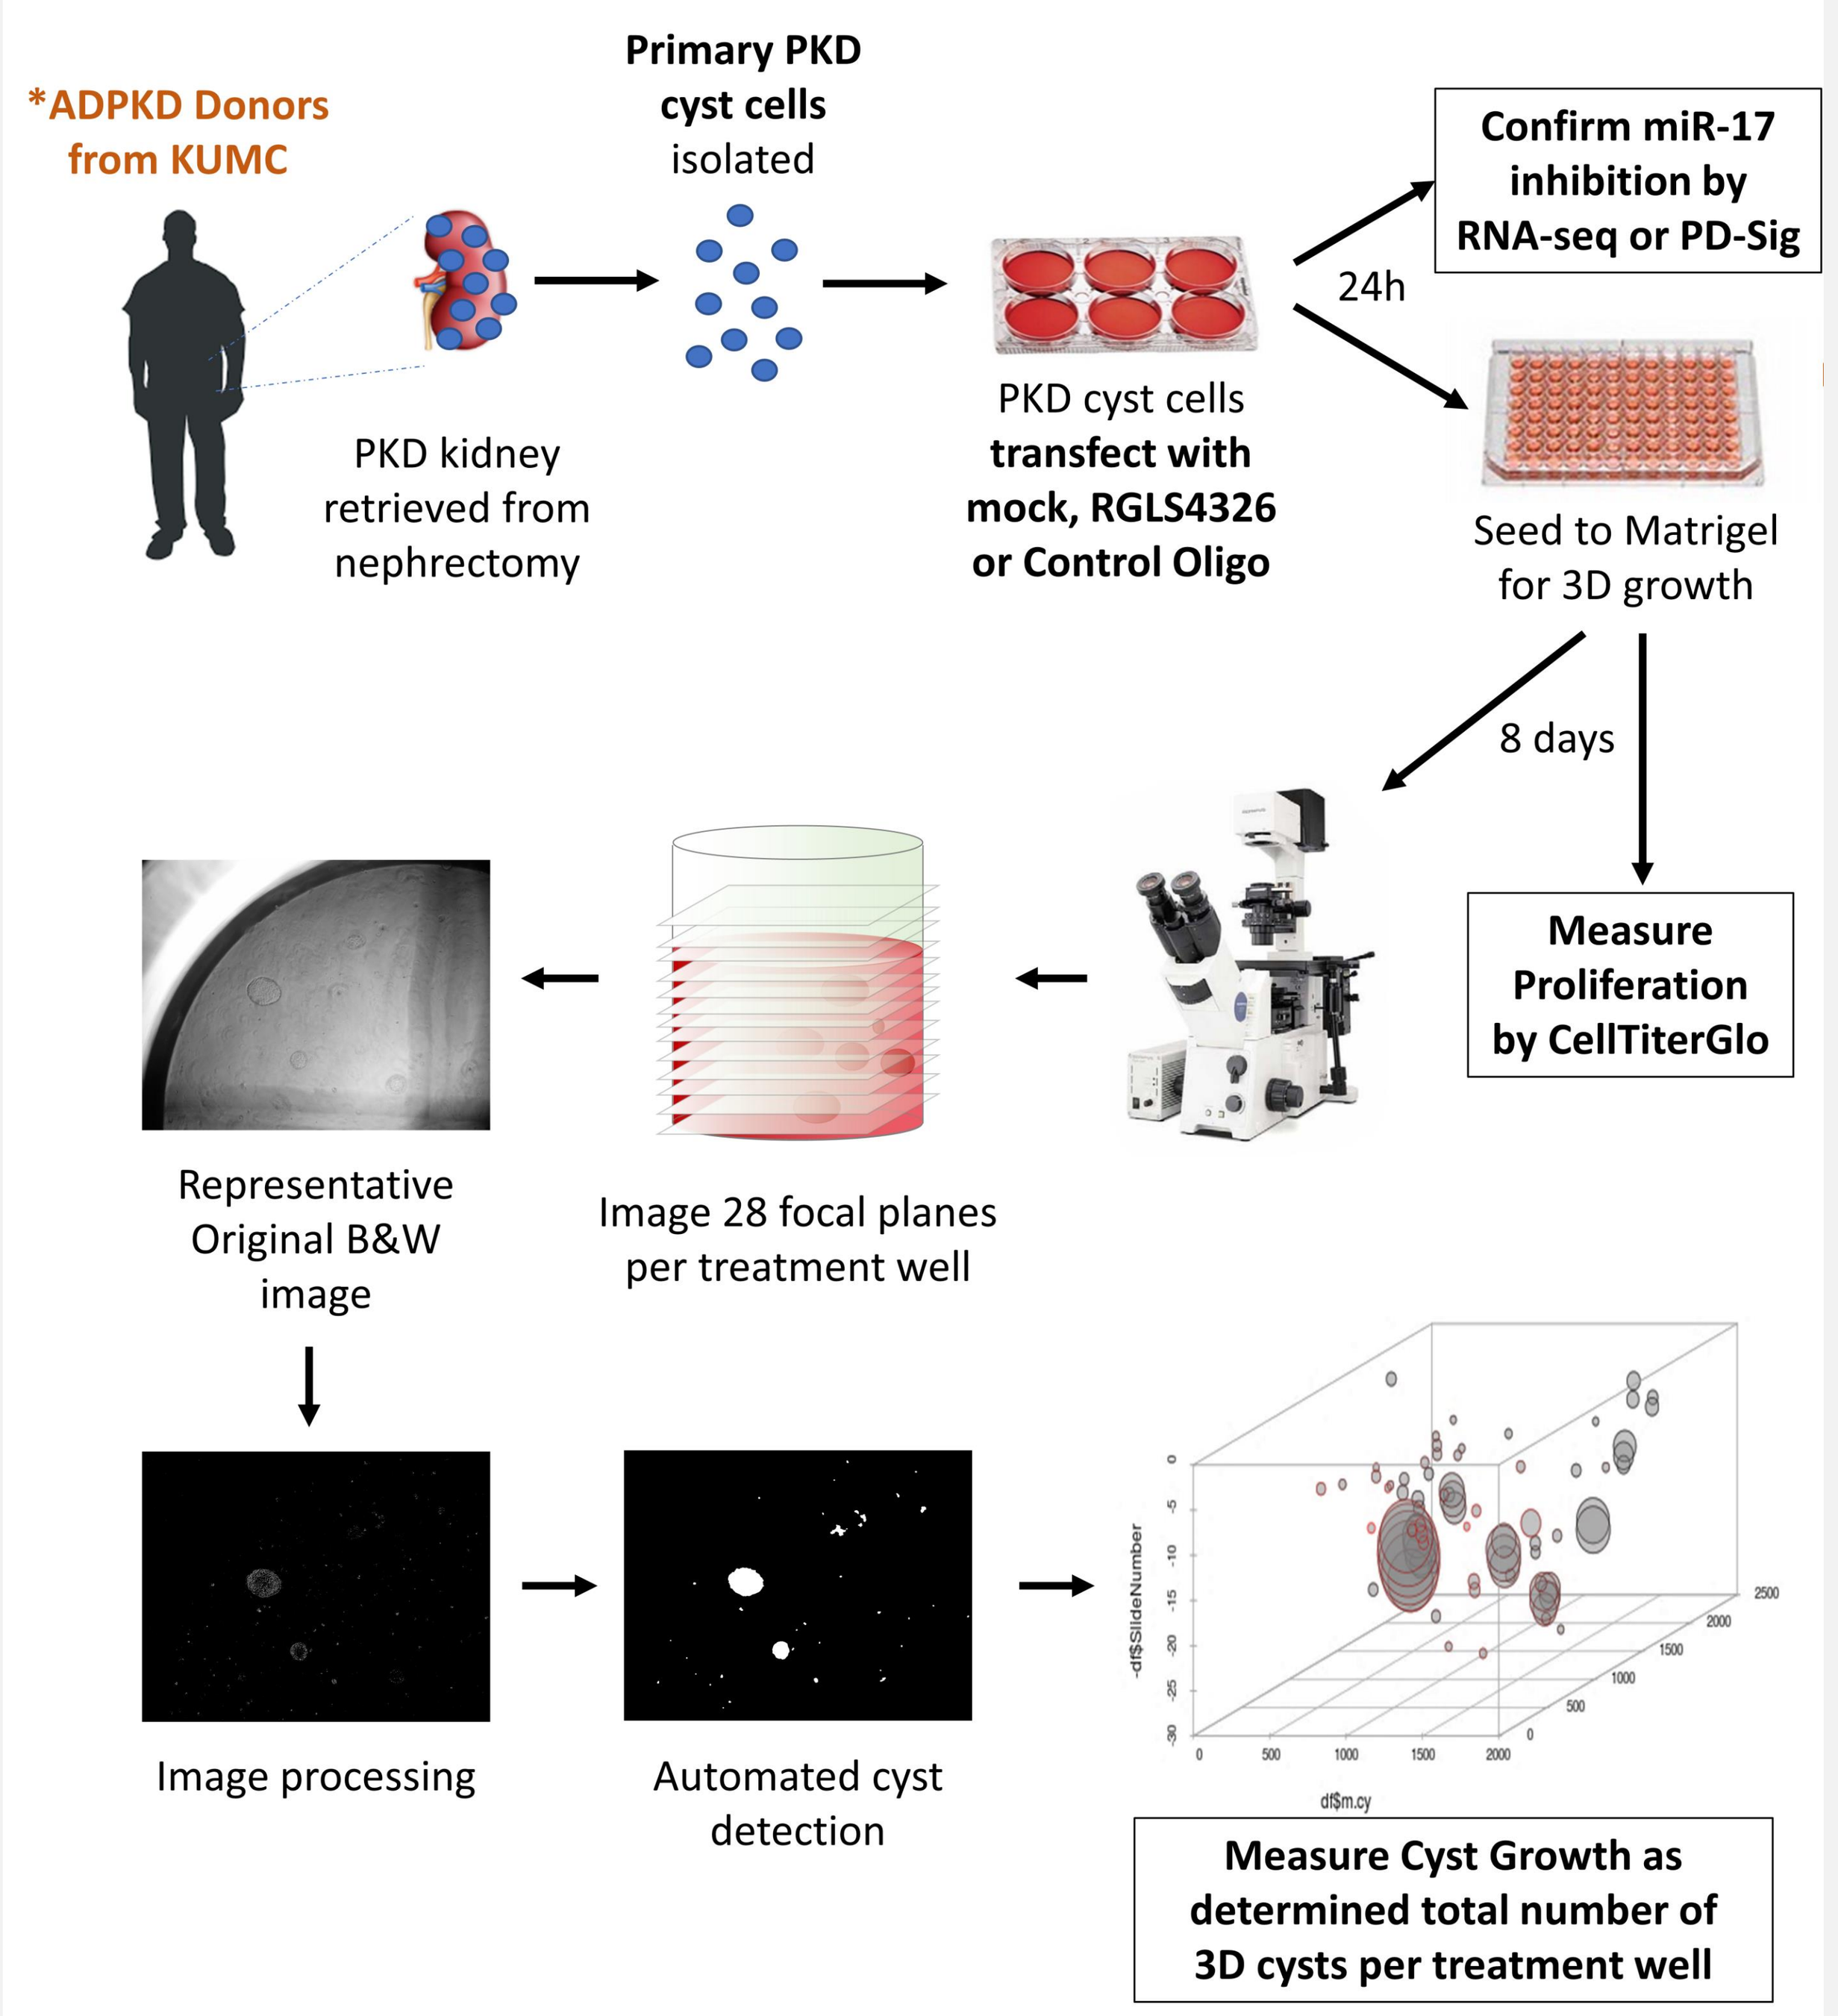
*ADPKD Donors from KUMC
PKD kidney retrieved from nephrectomy
Primary PKD cyst cells isolated
PKD cyst cells transfect with mock, RGLS4326 or Control Oligo
24h
Confirm miR-17 inhibition by RNA-seq or PD-Sig
Seed to Matrigel for 3D growth
8 days
Measure Proliferation by CellTiterGlo
Image 28 focal planes per treatment well
Representative Original B&W image
Image processing
Automated cyst detection
Measure Cyst Growth as determined total number of 3D cysts per treatment well

RGLS4326 de-represses miR-17 downstream target genes in primary ADPKD cyst derived from human donors *in vitro*

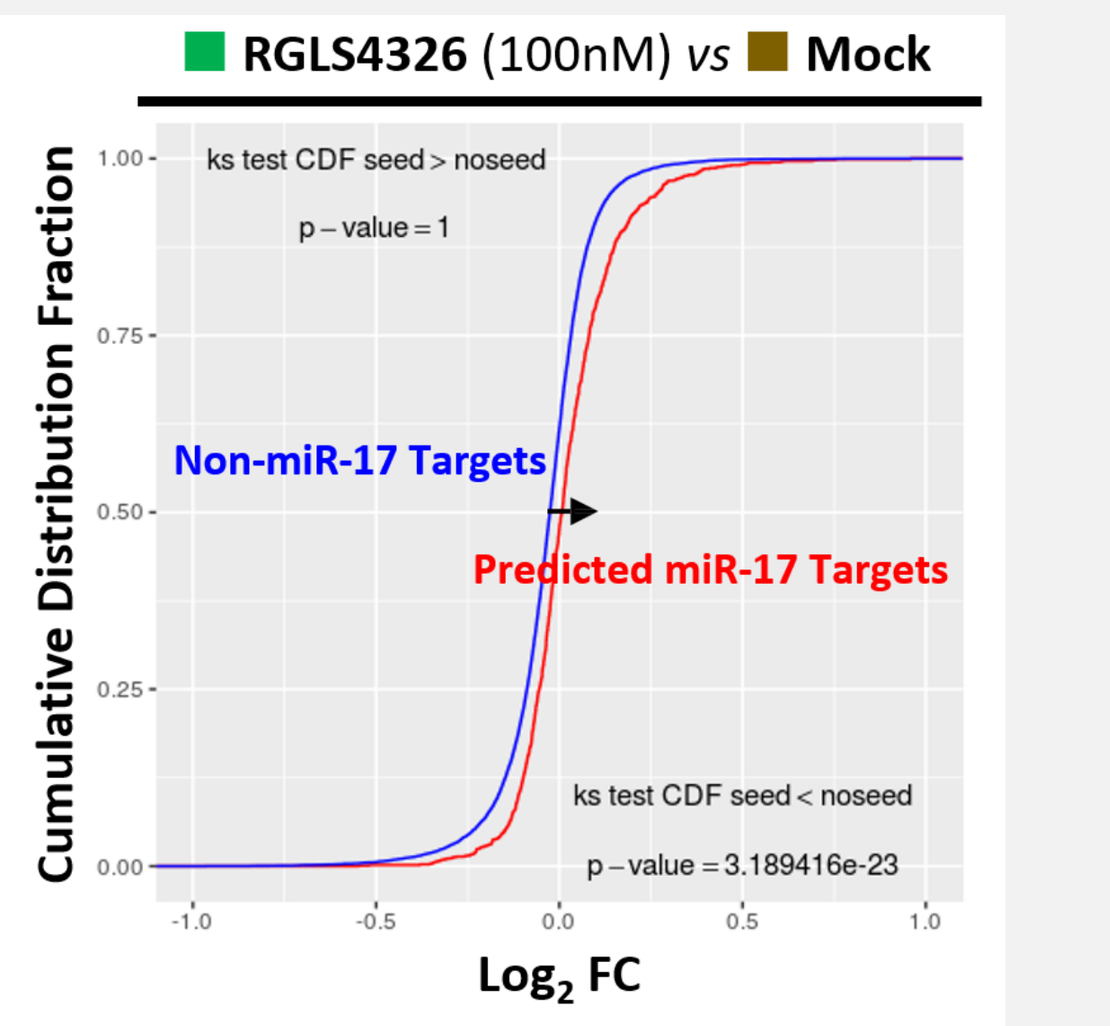
RGLS4326 (100nM) vs Mock
Cumulative Distribution Fraction
ks test CDF seed > noseed
p – value = 1
Non-miR-17 Targets
Predicted miR-17 Targets
ks test CDF seed < noseed
p – value = 3.189416e-23
Log2 FC

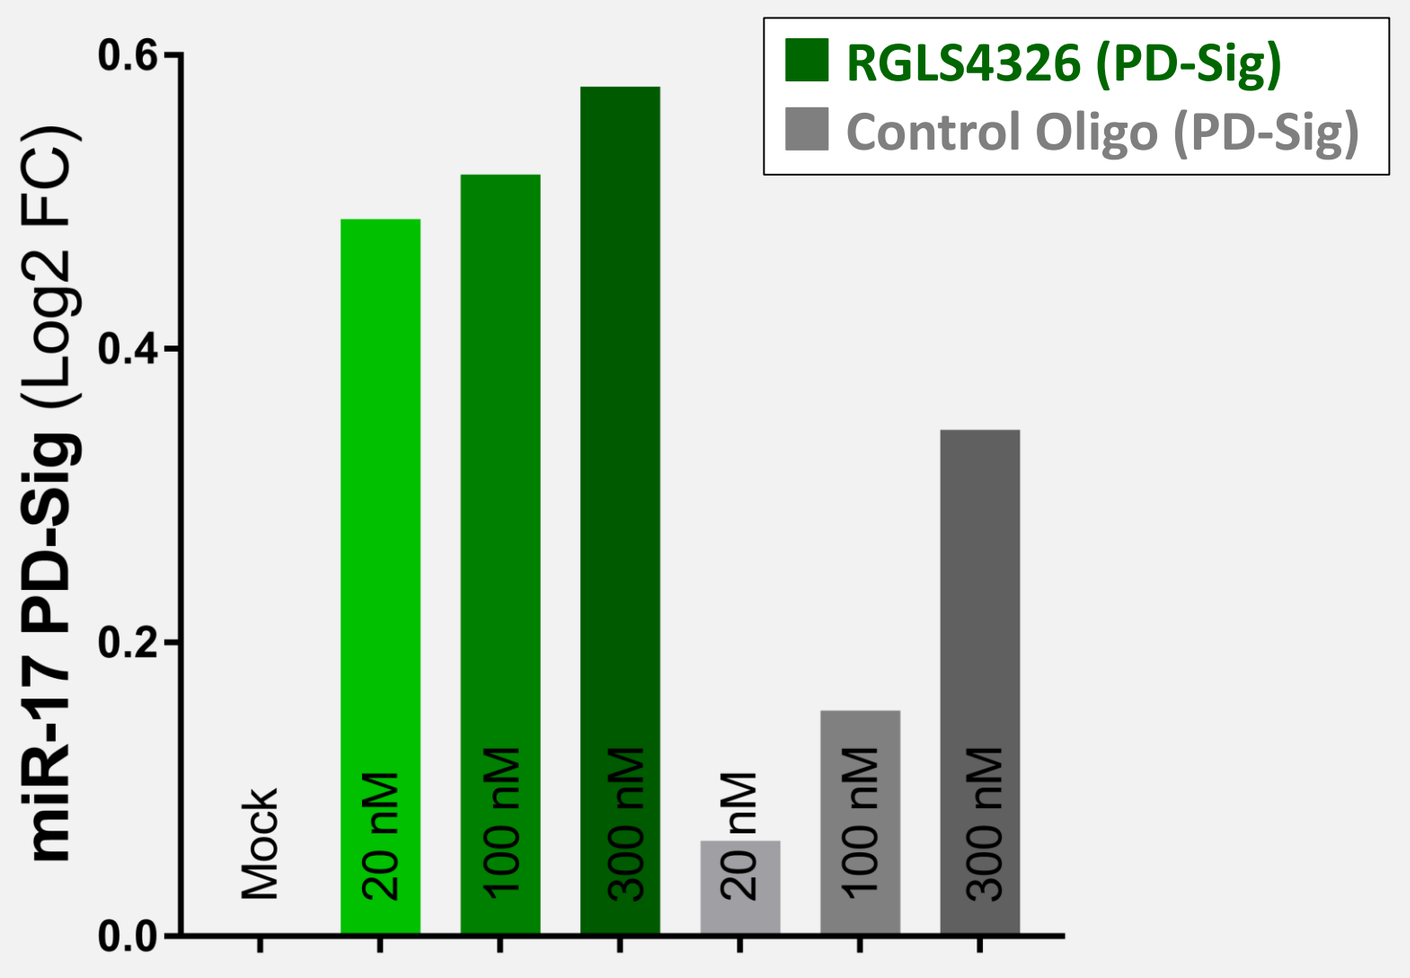
RGLS4326 (PD-Sig)
Control Oligo (PD-Sig)
miR-17 PD-Sig (Log2 FC)
Mock
20 nM
100 nM
300 nM
20 nM
100 nM
300 nM

- RNA-sequencing expression profiling was used to assess inhibition of miR17 in primary human ADPKD cyst cells following RGLS4326 treatment by transfection for 24h.
- Kolmogorov-Smironov test statistics comparing log2FC cumulative distribution indicated significant shift towards upregulation (i.e. de-repression) of predicted miR-17 target genes following RGLS4326 treatment (100nM) *in vitro*.
- For each subsequent experiment, functional inhibition of miR-17 was measured and shown by de-repression of a human miR-17 PD-Sig in representative cyst samples (N=1 for each treatment and dose) after 24h treatment and prior to further culturing in 3D cyst formation assay for 8 additional days.

RGLS4326 suppresses proliferation and cyst growth in primary ADPKD cyst derived from human donors *in vitro*

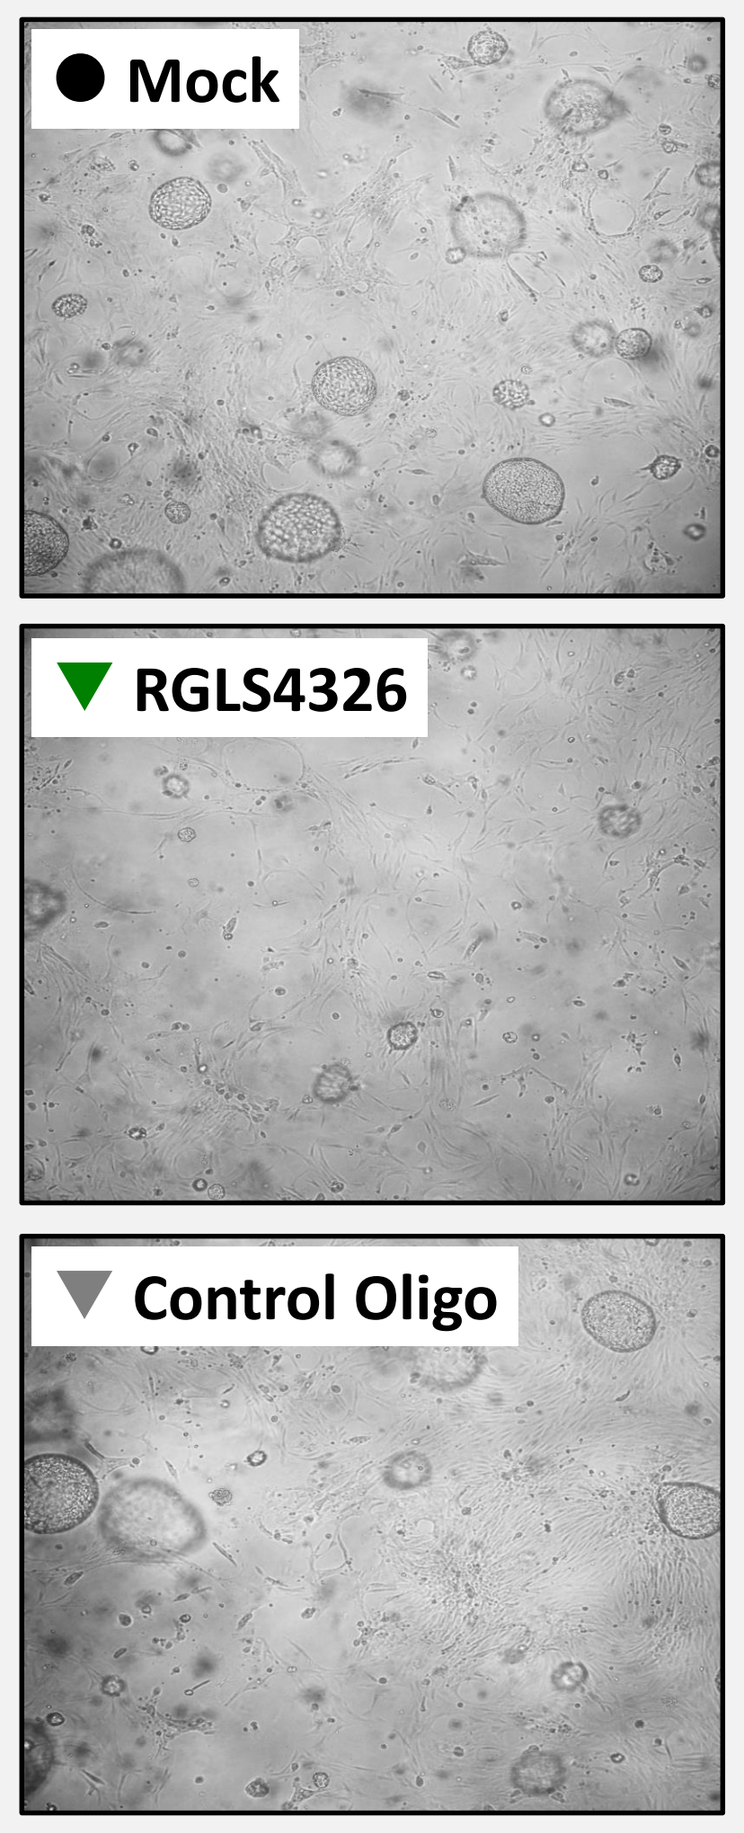
Mock
RGLS4326
Control Oligo

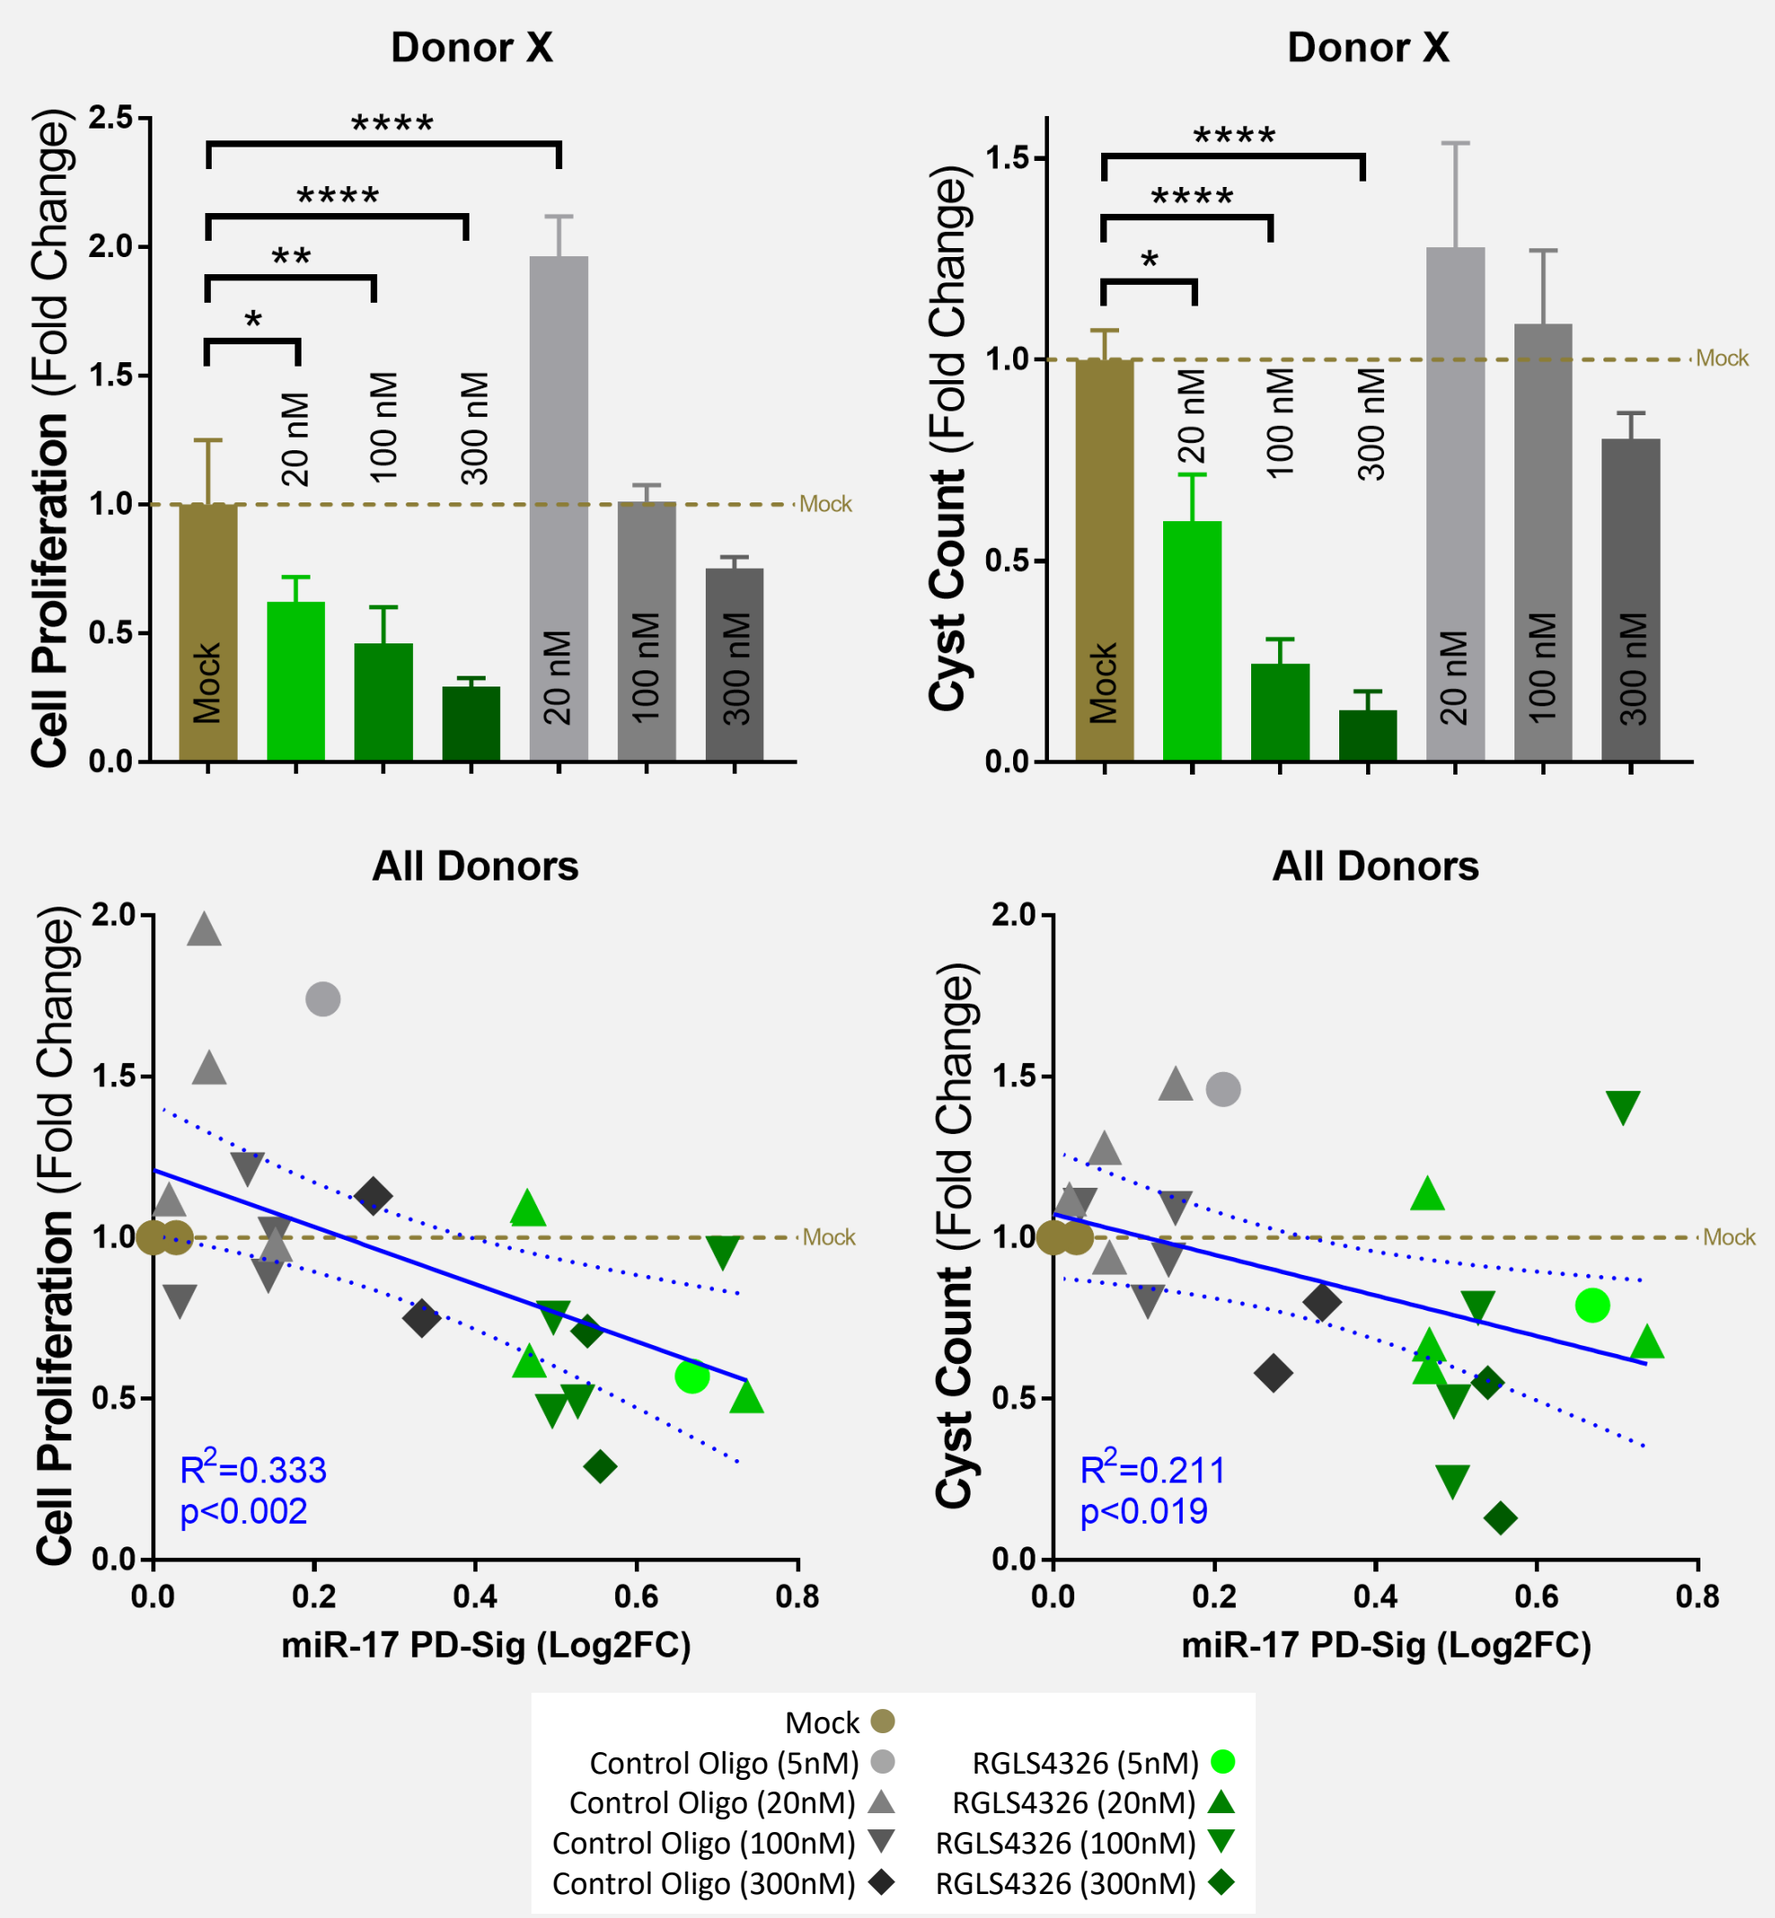
Donor X
Cell Proliferation (Fold Change)
Mock
20 nM
100 nM
300 nM
Donor X
Cyst Count (Fold Change)
All Donors
Cell Proliferation (Fold Change)
R^2=0.333
p<0.002
miR-17 PD-Sig (Log2FC)
All Donors
Cyst Count (Fold Change)
R^2=0.211
p<0.019
miR-17 PD-Sig (Log2FC)
Mock
Control Oligo (5nM)
Control Oligo (20nM)
Control Oligo (100nM)
Control Oligo (300nM)
RGLS4326 (5nM)
RGLS4326 (20nM)
RGLS4326 (100nM)
RGLS4326 (300nM)

- Representative images and quantification of 3D cyst formation showing reduction of proliferation (by CellTiterGlo) and cyst count (by imaging) 9 days following initial treatment (N=3 for each treatment and dose).
- Inhibition of miR-17 as measured by PD-Sig correlates with anti-proliferation and anti-cyst activity in primary human ADPKD cysts (total of N=4 studies among two donors). Control oligo at indicated concentrations were used as negative controls. Error bars represent standard deviations. *indicates p<0.05, **p<0.01, ***p<0.001, ****p<0.0001. One-way ANOVA, Dunnette's multiple comparison test.

Conclusion

- Our data from primary human ADPKD donors cysts demonstrated a correlation between miR-17 inhibition and cyst growth reduction across both RGLS4326 and control oligo treatment. Low magnitude of non-specific and idiosyncratic changes were observed from control oligo in the assay.
- Importantly, miR-17-inhibition-driven cyst growth reduction was specific to proliferating ADPKD cells, as no cytotoxic activity was observed in non-ADPKD collecting duct cells such as IMCD3 following RGLS4326 treatment *in vitro* or genetic knockdown of miR-17~92 cluster *in vivo* [Patel et al. (2013) PNAS. Jun 25;110(26):10765].
- Our preclinical data supports the clinical development of RGLS4326 for the treatment of human ADPKD.

American Society of Nephrology - Kidney Week, San Diego, CA (Oct 23 - 28, 2018)
Abstract Number: 3021589; Poster Board Number: TH-PO686